#6344 Engineering of ICAM-1 refractory antibodies for the development of therapeutic antibodies and antibody-drug conjugates (ADCs) in ICAM-1 over-expressing cancers.

Brad Kline, Luigi Grasso, and Nicholas C Nicolaides. Navrogen Inc. Cheyney, PA - nick@navrogen.com

navrogen

Leaders in Humoral Immuno-Oncology

Abstract

Human cancers employ a number of mechanisms to evade host immune responses against novel antigens generated from aberrant over-expression, mutations and/or epigenetic alterations. Humoral immunity utilizes antibodies and immune effector cells as well as molecular immune complexes involving the complement system to mediate the killing of dysregulated cancer cells. We refer to these anti-cancer mechanisms as Humoral Immuno-Oncology (HIO). HIO suppression is mediated by tumor-produced proteins called HIO factors. One such factor is CA125, which was previously shown to bind IgG-type antibodies and inhibit their immune-effector activities, including antibody-dependent (ADCC) and complement-dependent (CDC) cellular cytotoxicity. Using a combination of experimental screening and literature searches, a second protein produced by tumors and associated with a variety of cancer indications was discovered and found to be soluble ICAM-1 (sICAM-1). Through various functional studies we reported that sICAM-1 and membrane ICAM-1 (mICAM-1) are capable of binding IgG1-type antibodies that in turn inhibits their immune-effector activity. Through a combination of truncation and substitution mutagenesis, we identified a four amino acid motif within the CH3 domain of IgG1 essential for ICAM-1 binding, resulting in the inhibition of ADCC activity. While traditional glycine and alanine substitutions in this region abrogated ICAM-1 binding, these modifications caused tertiary structural changes in the Fc domain that resulted in loss of ADCC activity. Through a combinatorial amino acid substitution approach, we identified a four amino acid combination within the $_{407}$YSKL$_{410}$ to $_{407}$FARV$_{410}$ motif that resulted in antibodies refractory to sICAM-1 binding with robust ADCC activity. Additionally, isogenic wildtype and ICAM-1 knockdown target cell lines showed mICAM-1 inhibited target cell killing of a saporin antibody-drug conjugate (ADC) and this was the direct result of mICAM-1 reducing antibody internalization, a requisite for maximal ADC target cell killing. The inhibitory activity of mICAM-1 could be overcome by employing antibodies containing the $_{407}$FARV$_{410}$ substitution. These findings highlight yet another mechanism by which tumors can suppress the host's immune system for survival and offer new concepts for developing antibody-based therapies that can aid in the treatment of various cancer indications, especially those over-expressing ICAM-1. Moreover, the findings here offer diagnostic and therapeutic clinical design opportunities to improve upon existing approved immune-mediated therapies for which this factor is present.

Fig. 1- ICAM-1 directly binds IgG1 and inhibits ADCC activity

A) Biotinylated IgG1 binds immobilized ICAM-1. B) Biotinylated ICAM-1 binds immobilized IgG1. C) Dose-dependent inhibition of ADCC by ICAM-1. D) ICAM-1 inhibits ADCC of therapeutic Abs.

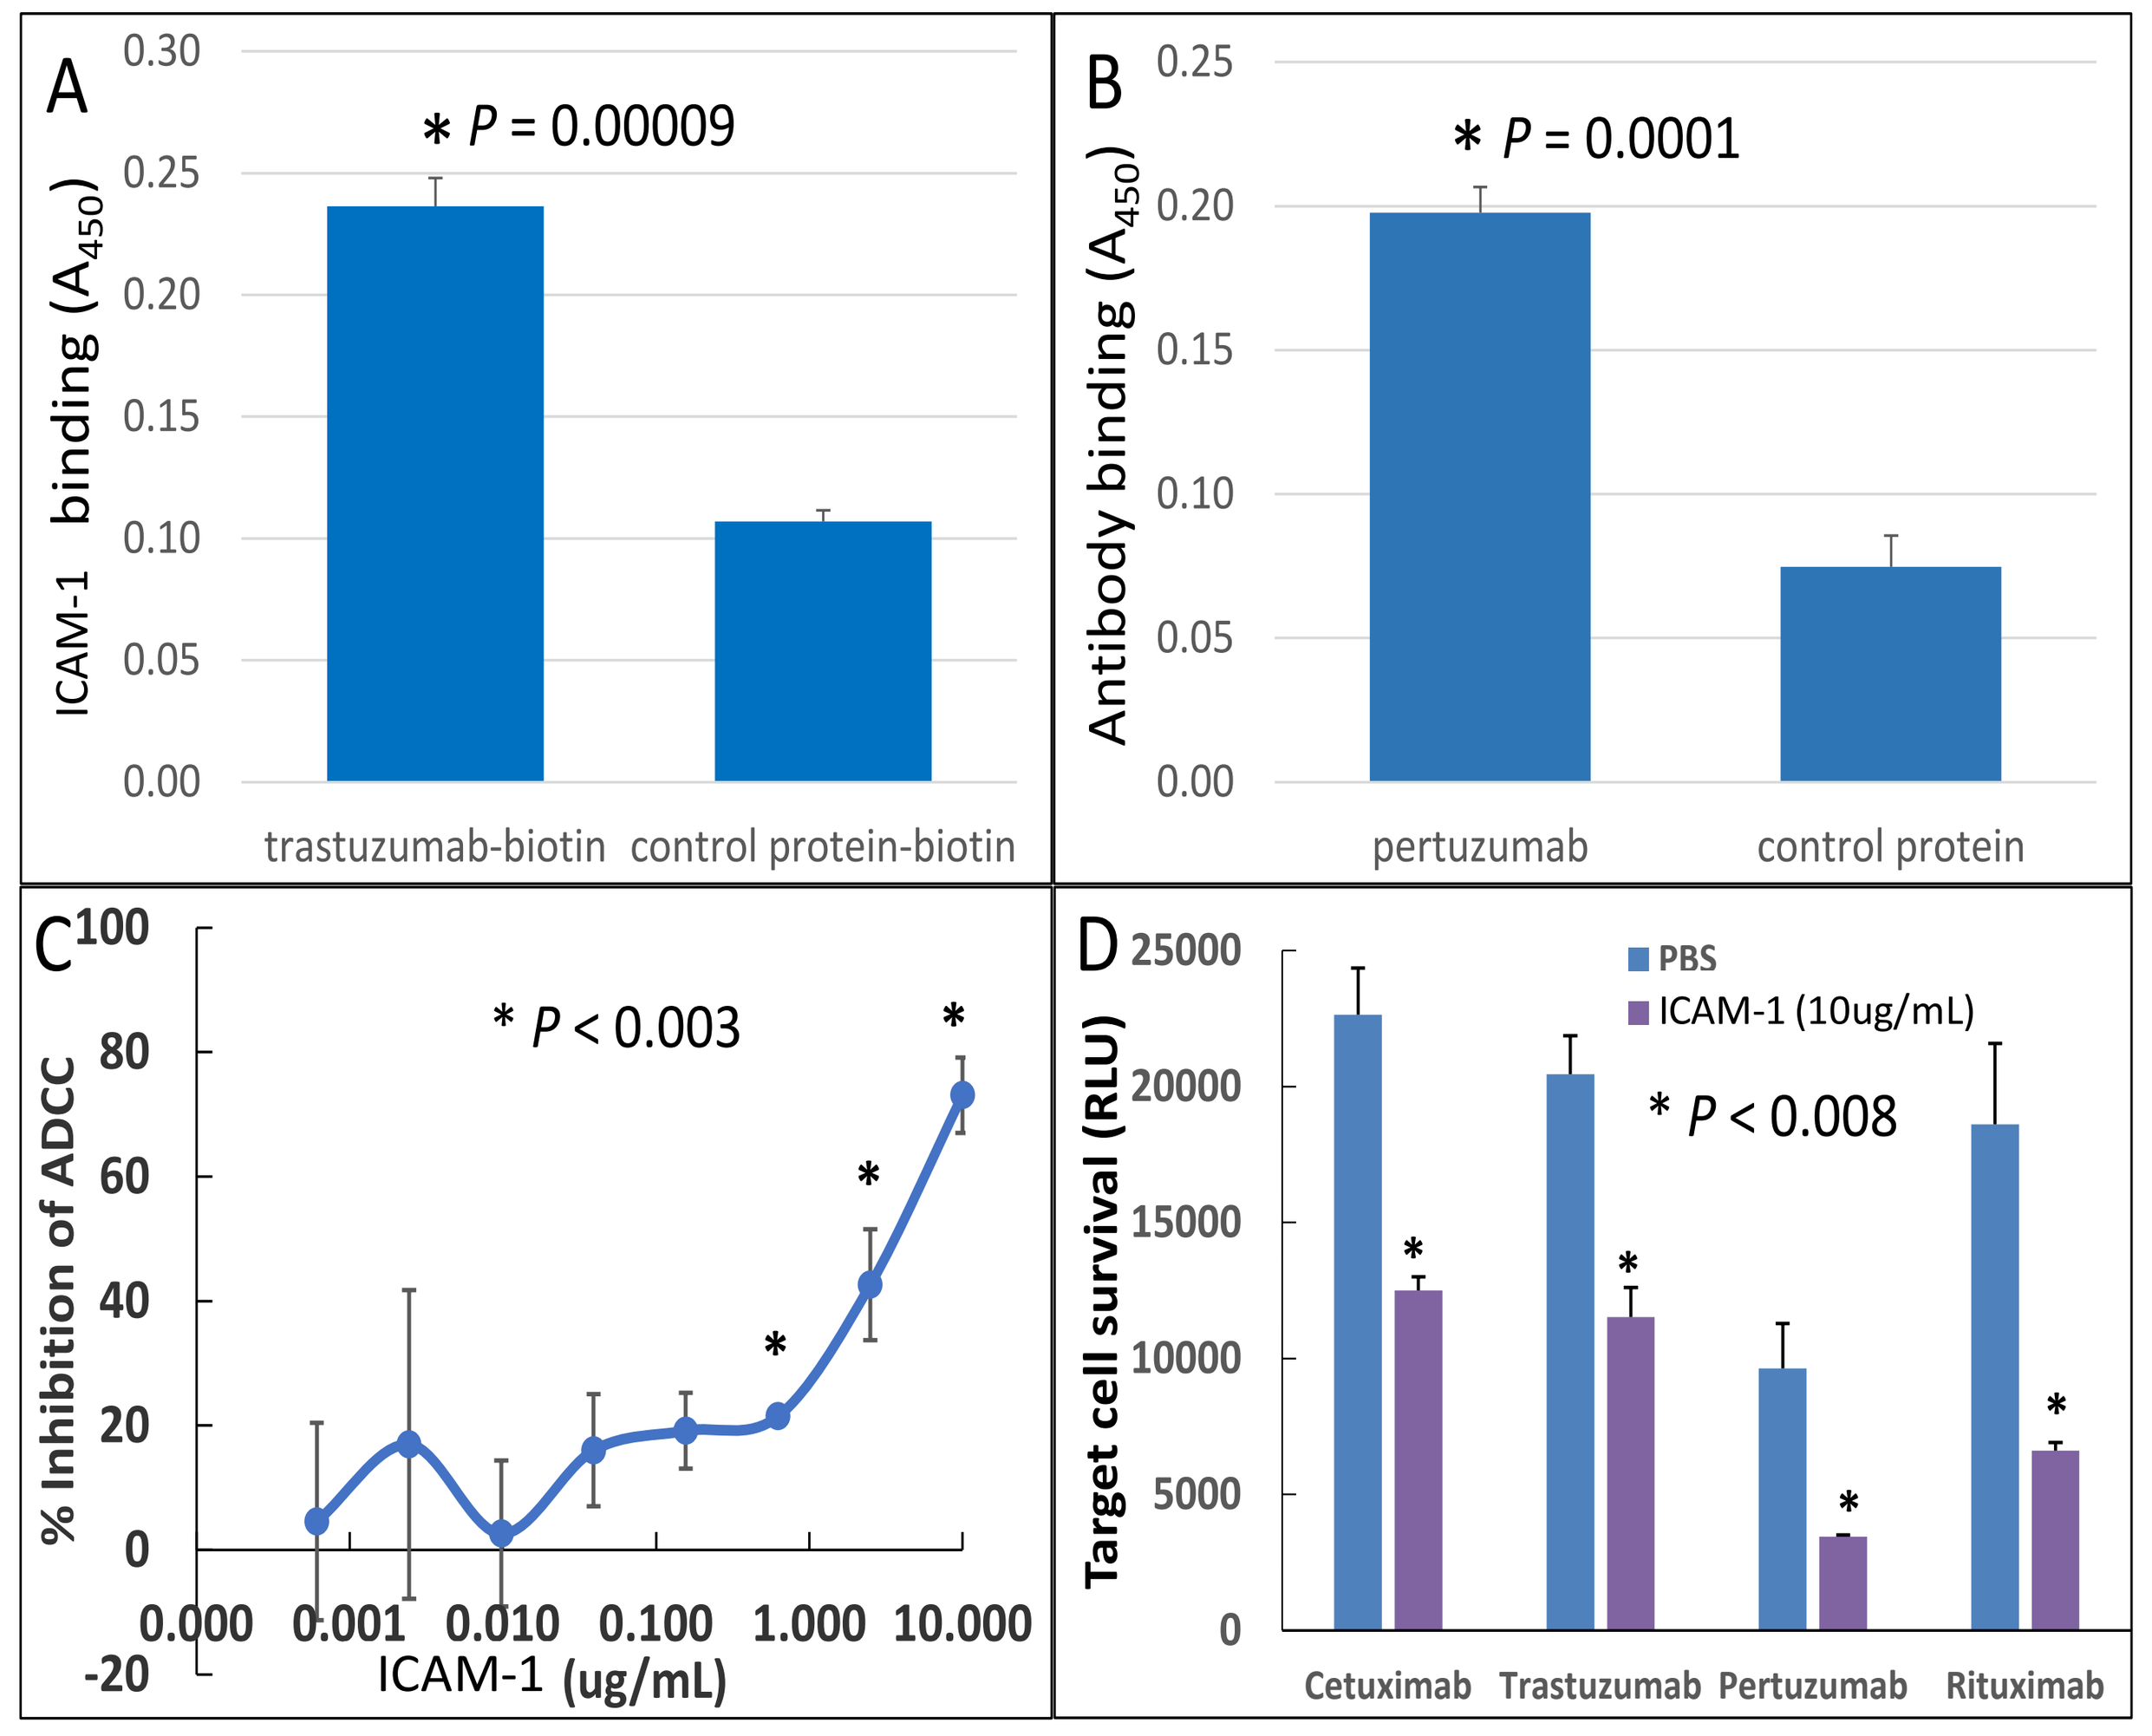
A
ICAM-1 binding (A_{450})
* P = 0.00009
trastuzumab-biotin
control protein-biotin
B
Antibody binding (A_{450})
* P = 0.0001
pertuzumab
control protein
C
% Inhibition of ADCC
* P < 0.003
ICAM-1 (ug/mL)
D
Target cell survival (RLU)
PBS
ICAM-1 (10ug/mL)
* P < 0.008
Cetuximab
Trastuzumab
Pertuzumab
Rituximab

Fig. 2- ICAM-1 blocks ADCC and CDC via suppressed CD16a Fc receptor and C1q protein engagement with IgG1 Fc domain

A) Biotinylated CD16a binds immobilized cetuximab and is inhibited by ICAM-1. B) Biotinylated C1q binds immobilized cetuximab and is inhibited by ICAM-1.

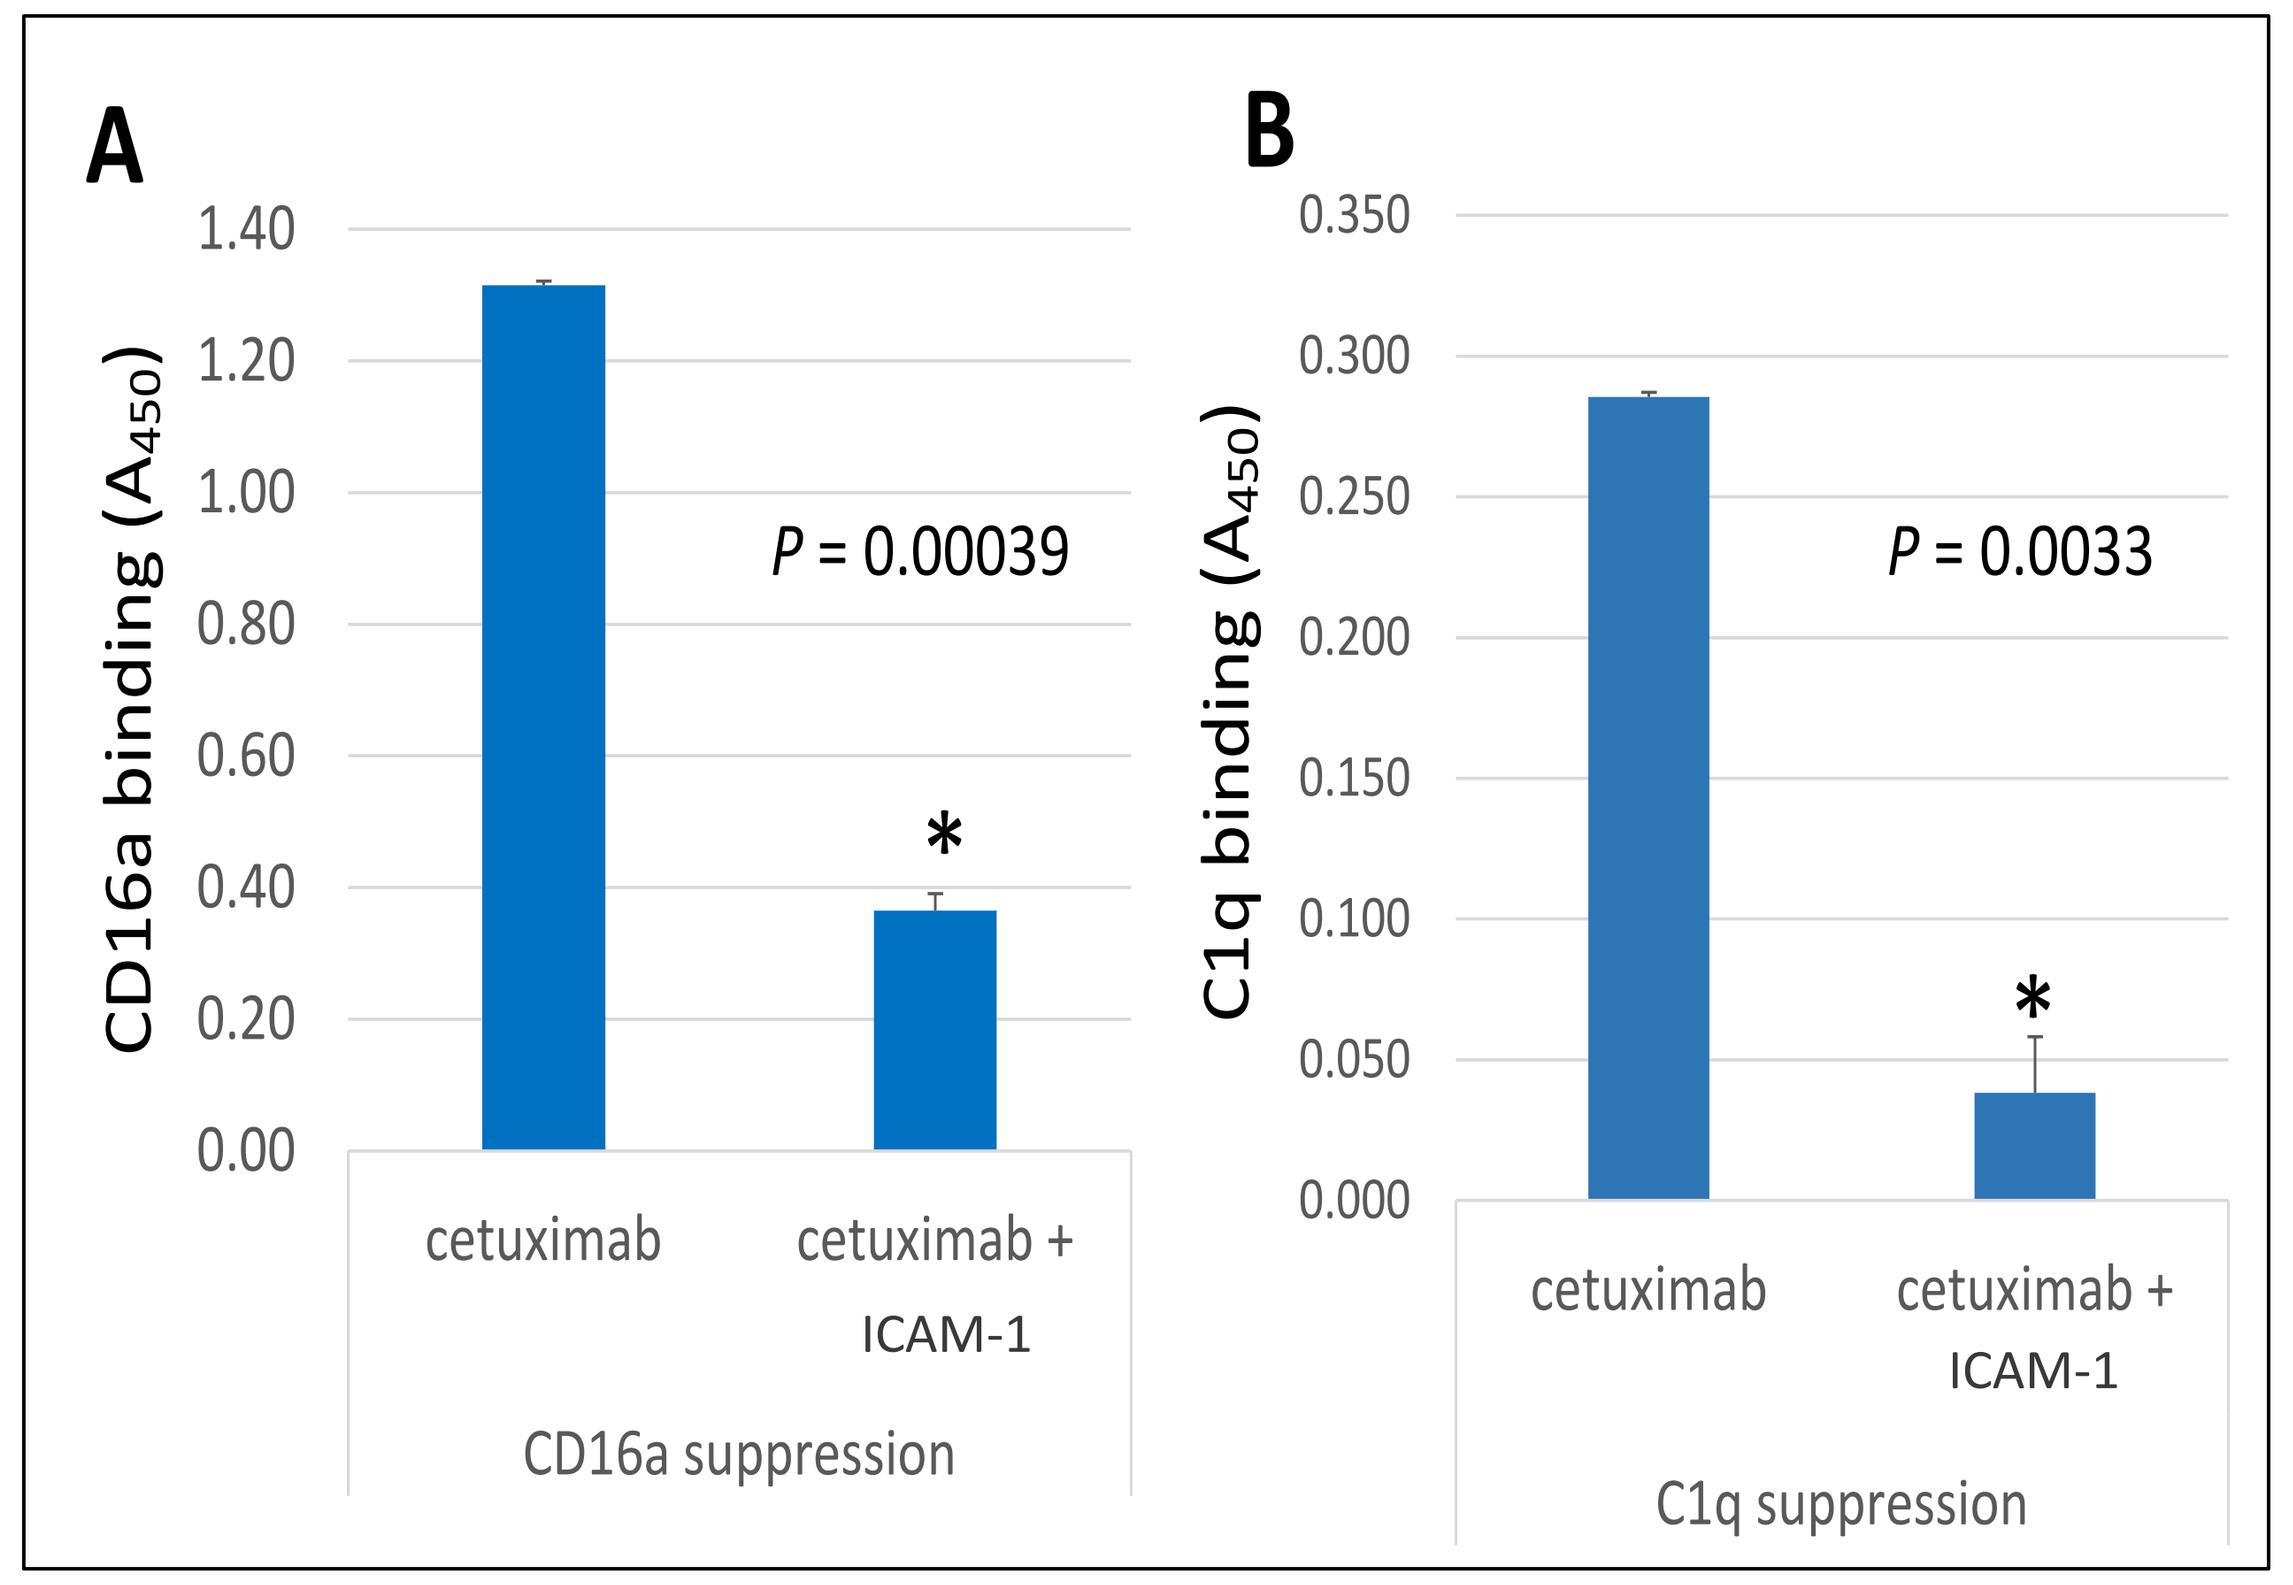
A
CD16a binding (A_{450})
P = 0.00039
cetuximab
cetuximab + ICAM-1
CD16a suppression
B
C1q binding (A_{450})
P = 0.0033
cetuximab
cetuximab + ICAM-1
C1q suppression

Fig. 3- ICAM-1 binds the Fc domain of IgG1 antibodies

A) Immobilized full-length (cetuximab) and IgG1 antibody fragments were probed with biotinylated ICAM-1 and binding was found to be localized to the Fc domain. B) Probing of immobilized dimeric (hu Fc) and monomeric Fc subdomains suggests ICAM-1 binding recognizes tertiary structures of dimerized IgG1 Fc heavy chains similar to full length rituximab (RTX).

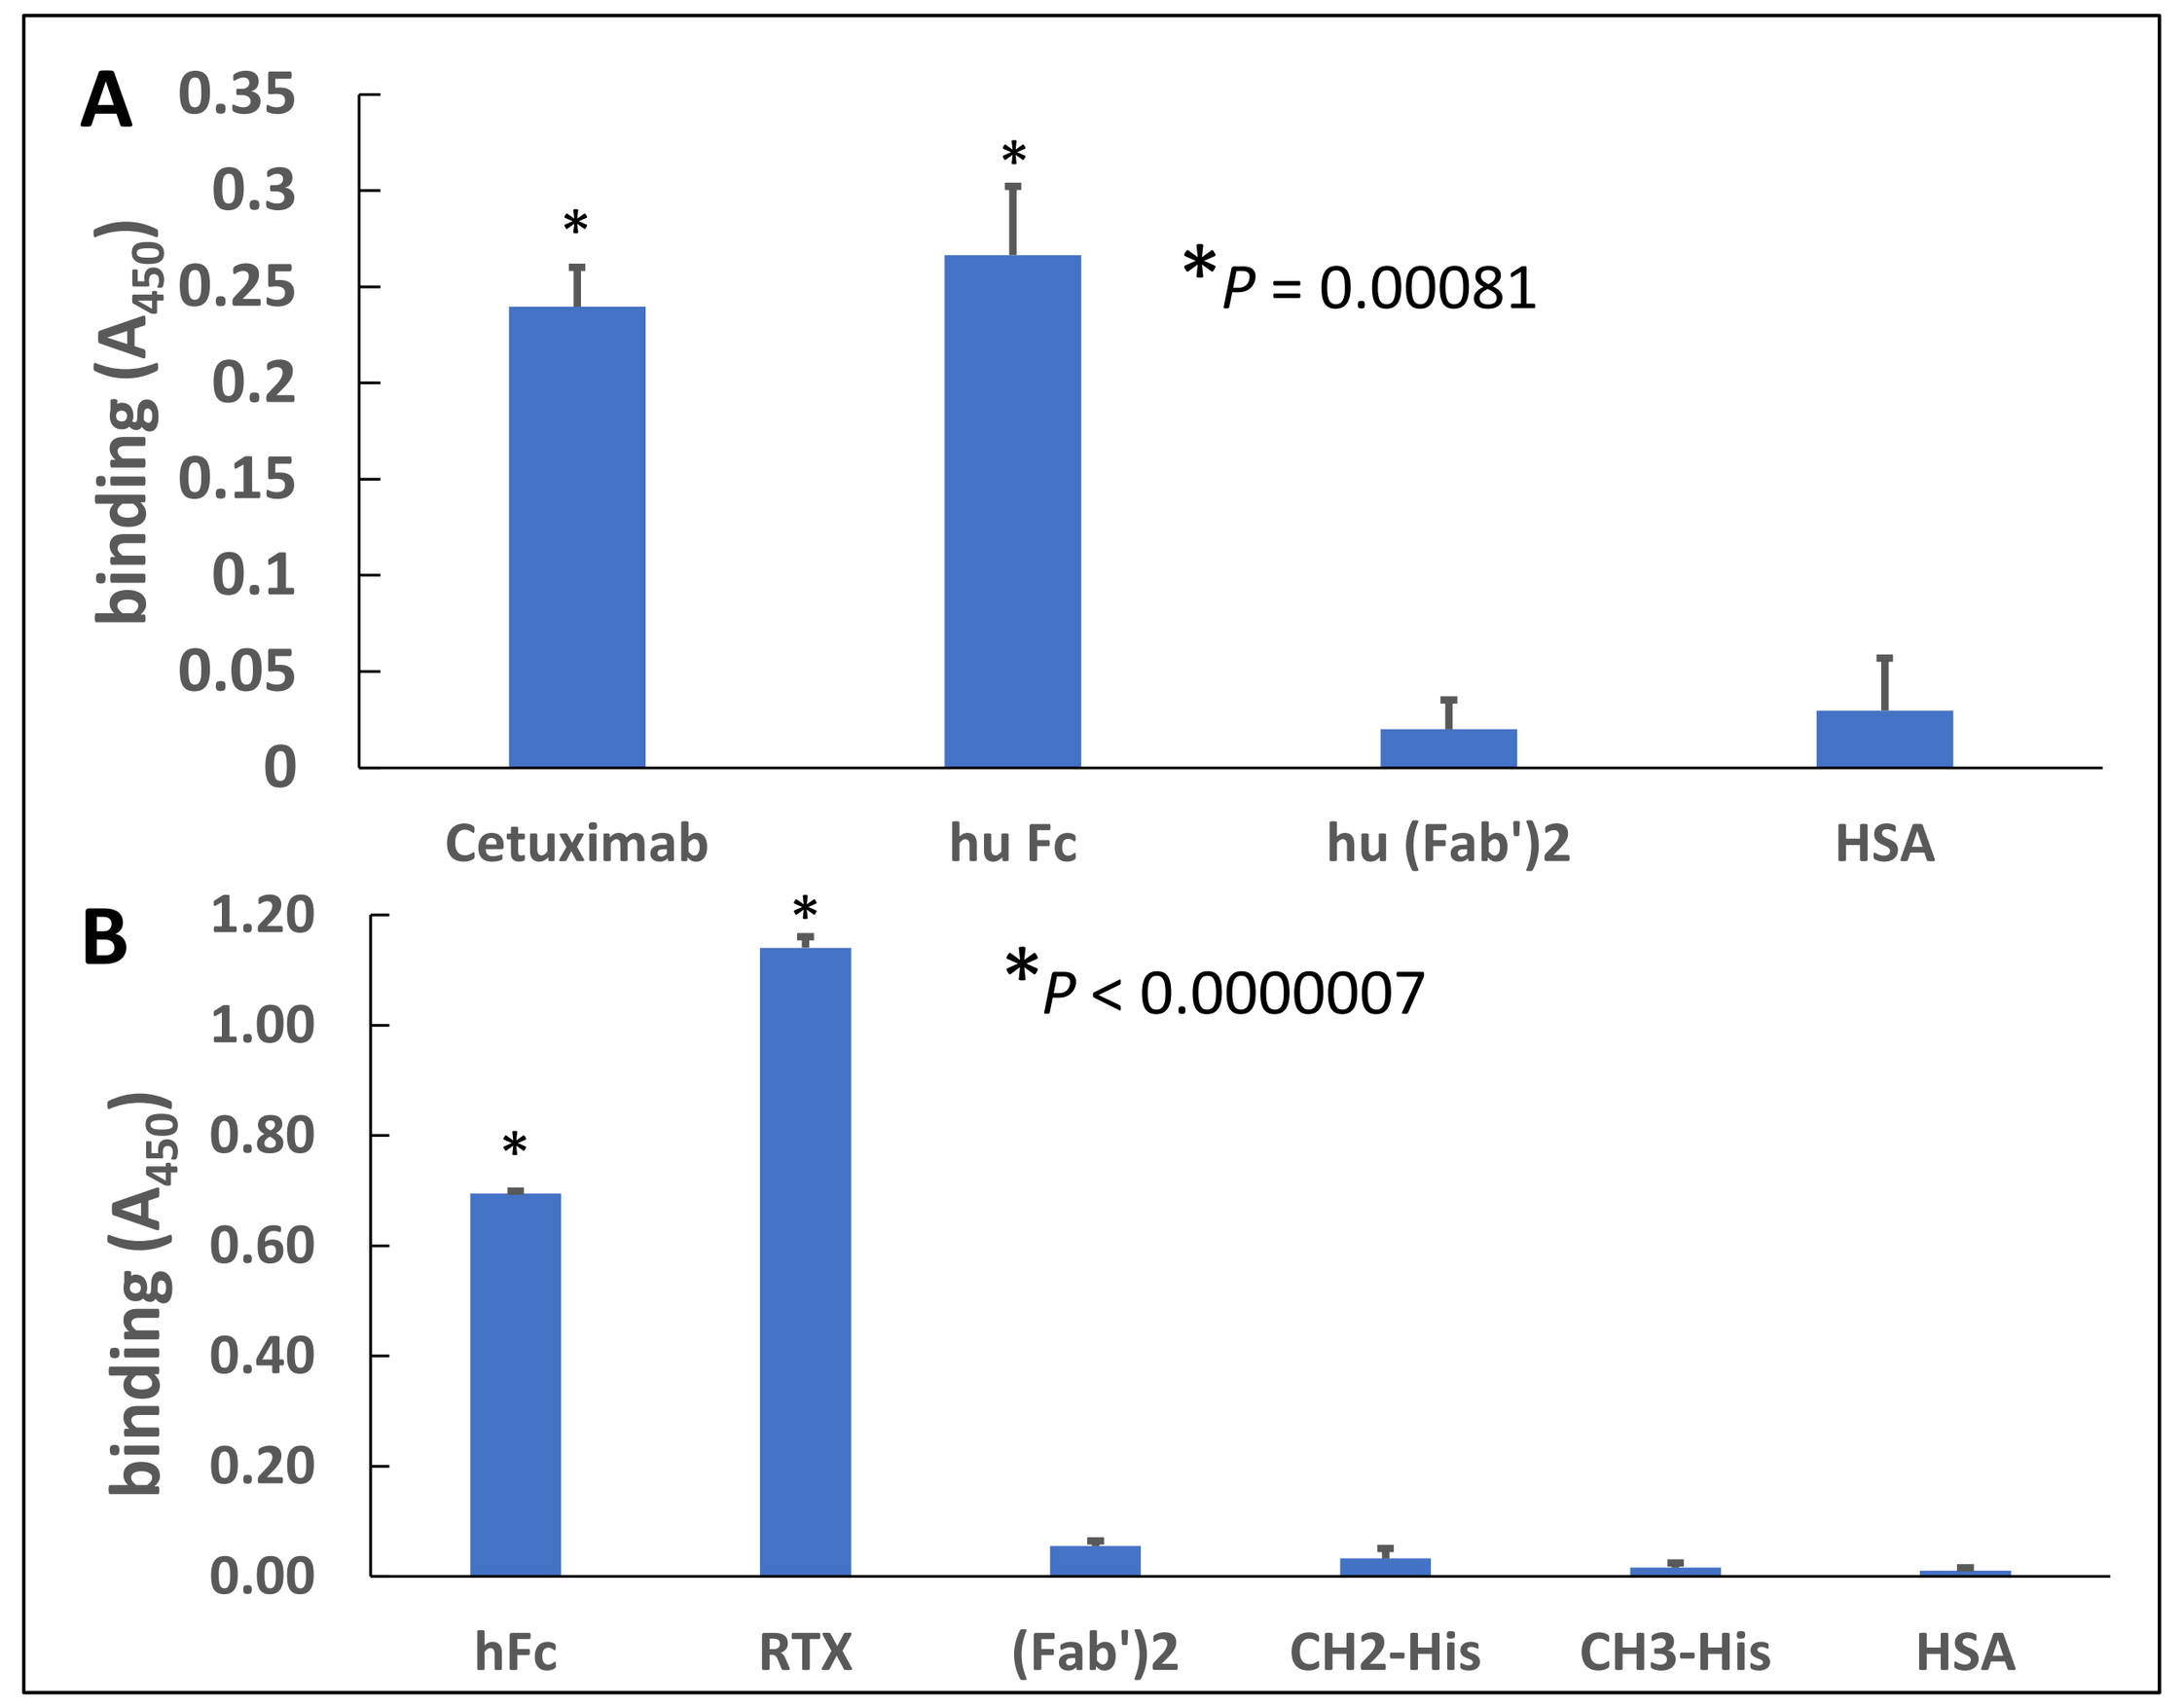
A
binding (A_{450})
*P = 0.00081
Cetuximab
hu Fc
hu (Fab')2
HSA
B
binding (A_{450})
*P < 0.0000007
hFc
RTX
(Fab')2
CH2-His
CH3-His
HSA

Fig. 4- IgG1 Fc domain mapping of the ICAM-1 binding domain

Amino acid sequence ID numbers (left) of IgG1 GST Fc deletion and truncation mutants to map the ICAM-1 binding domain. Bold sequences indicate substitution mutations. The right column summarizes Fc regions and residues required for ICAM-1 binding. Seq. 27 (FARV) indicates the substitution of 4 residues based on PAM250 mutation probability index.

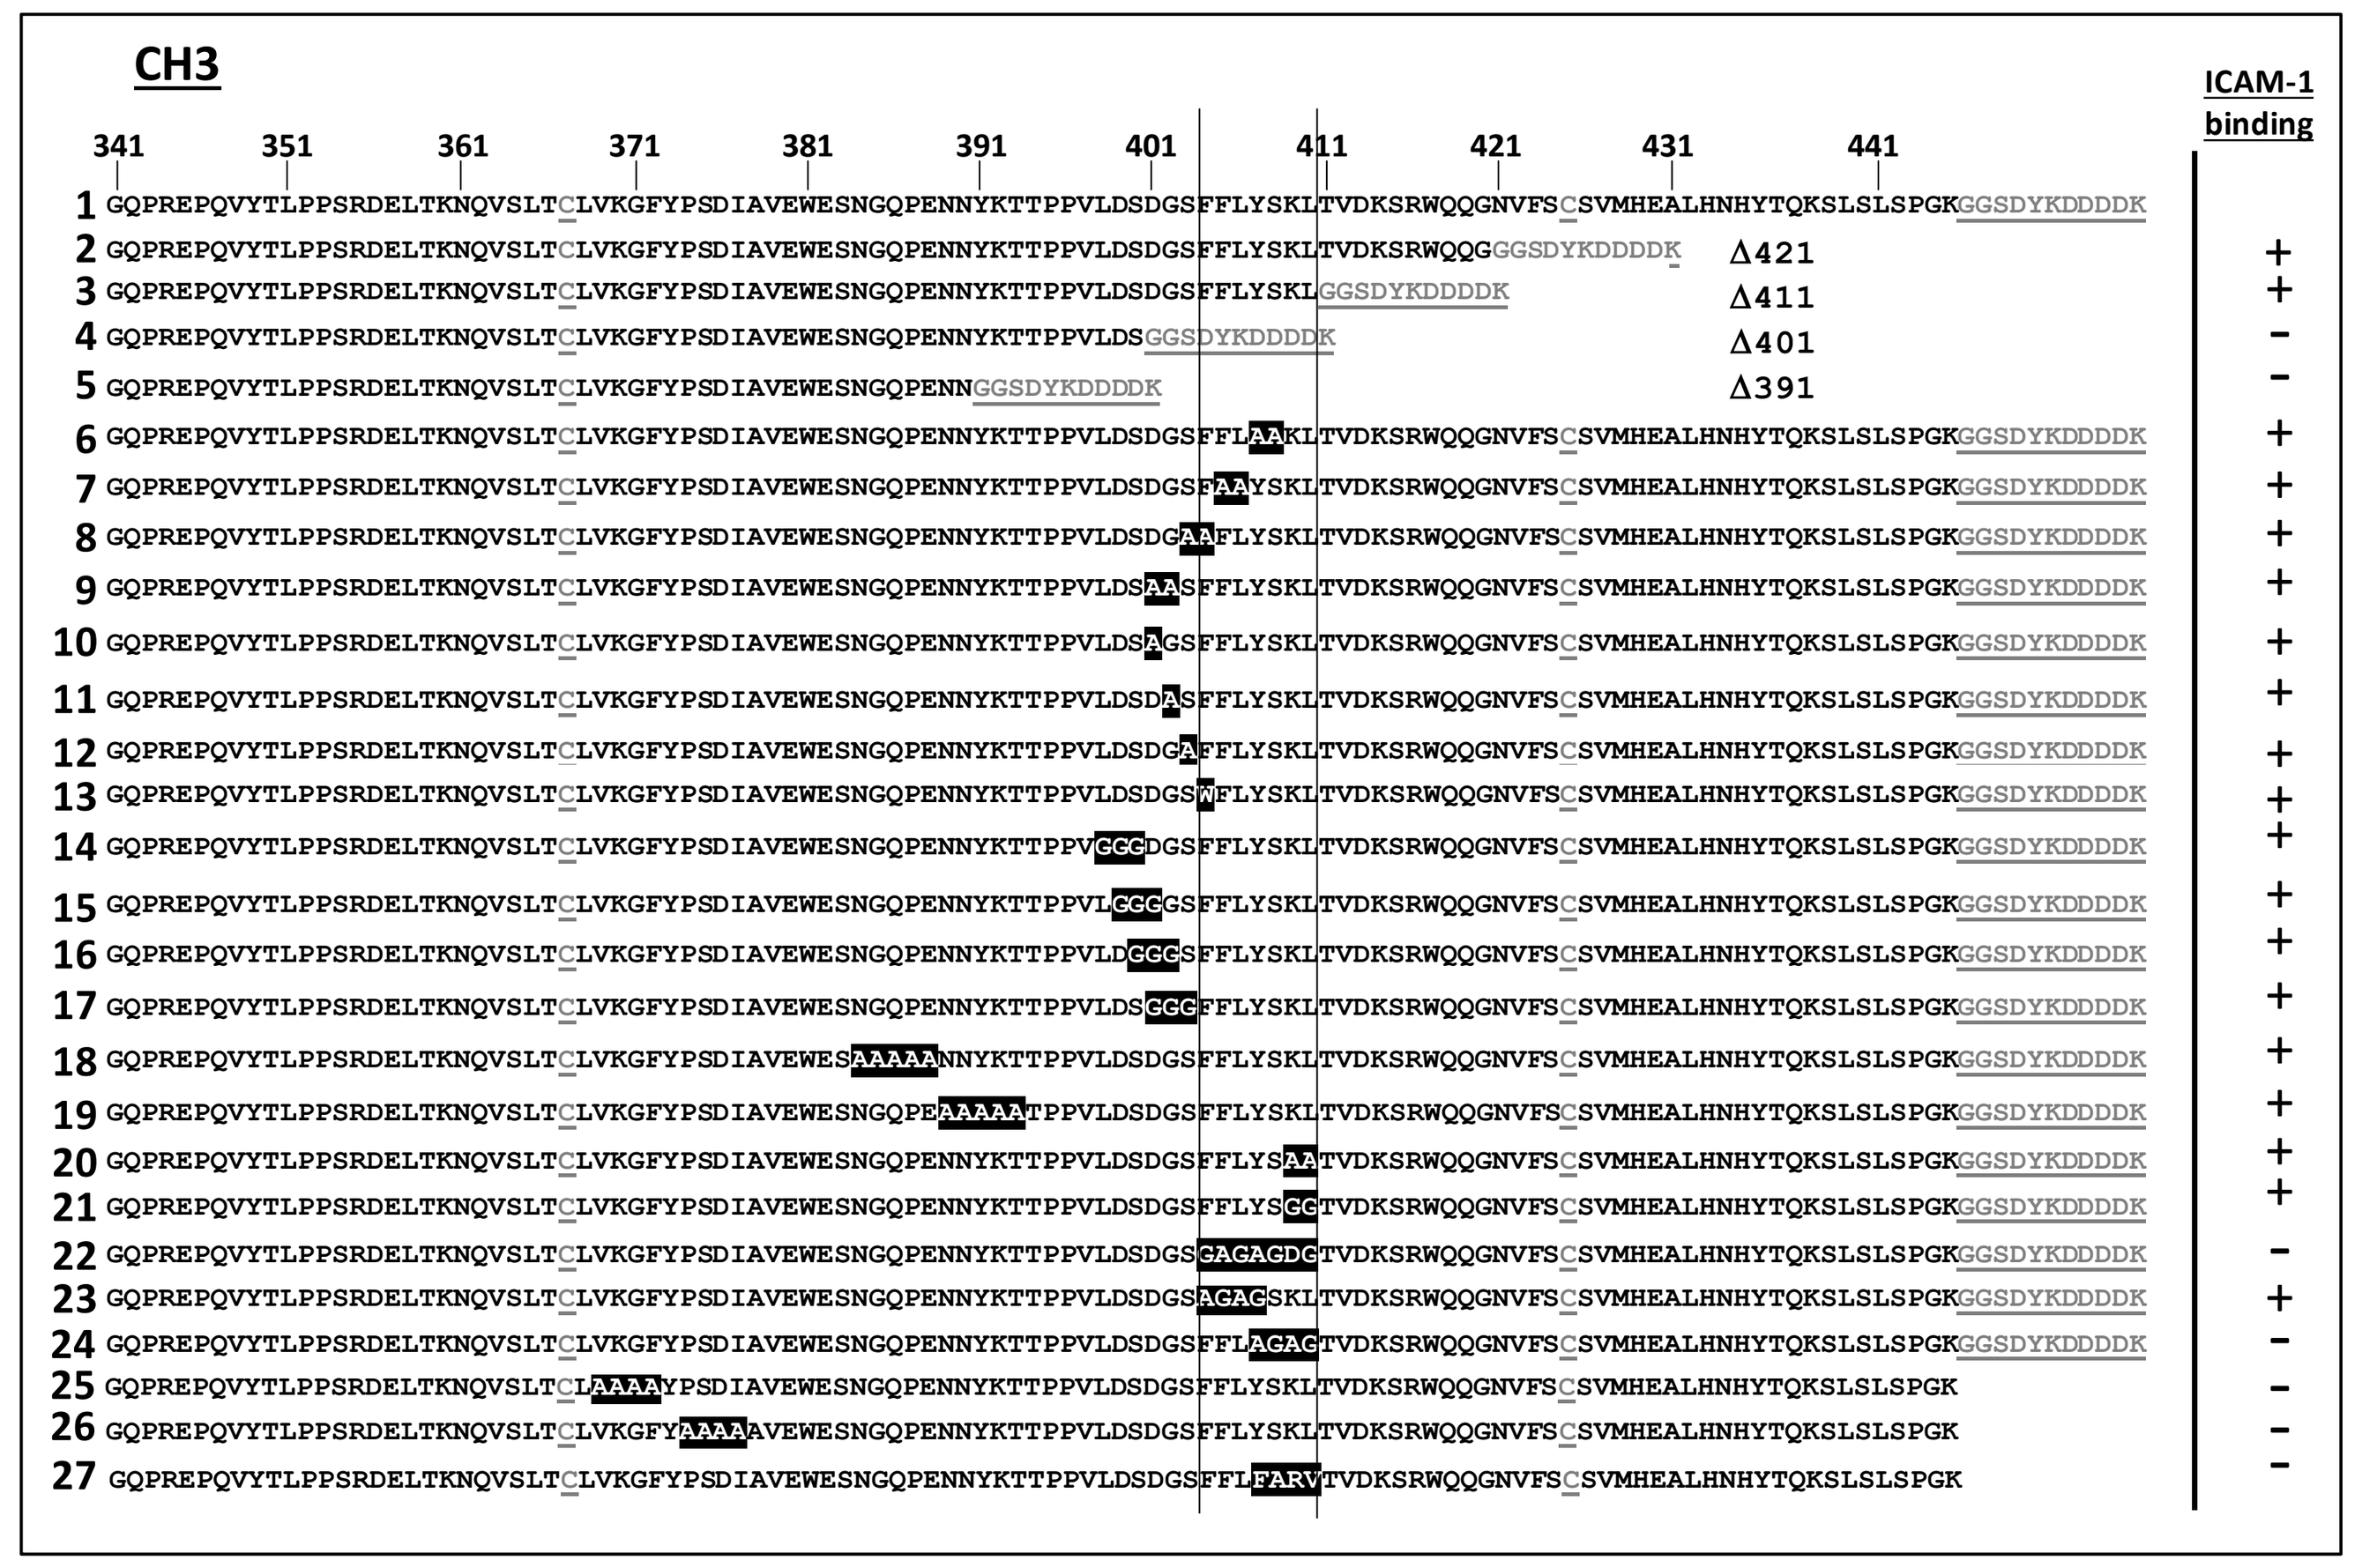
CH3
ICAM-1 binding
Δ421
Δ411
Δ401
Δ391

Fig. 5- ICAM-1 binding is localized to residues 407-410 in Fc domain

Rituximab (RTX) antibodies were engineered with amino acid substitutions within the ICAM-1 binding domain and tested for ICAM-1 binding via binding competition assays. Sequence #24 variant was found to be completely devoid of ICAM-1 binding, localizing binding to CH3.

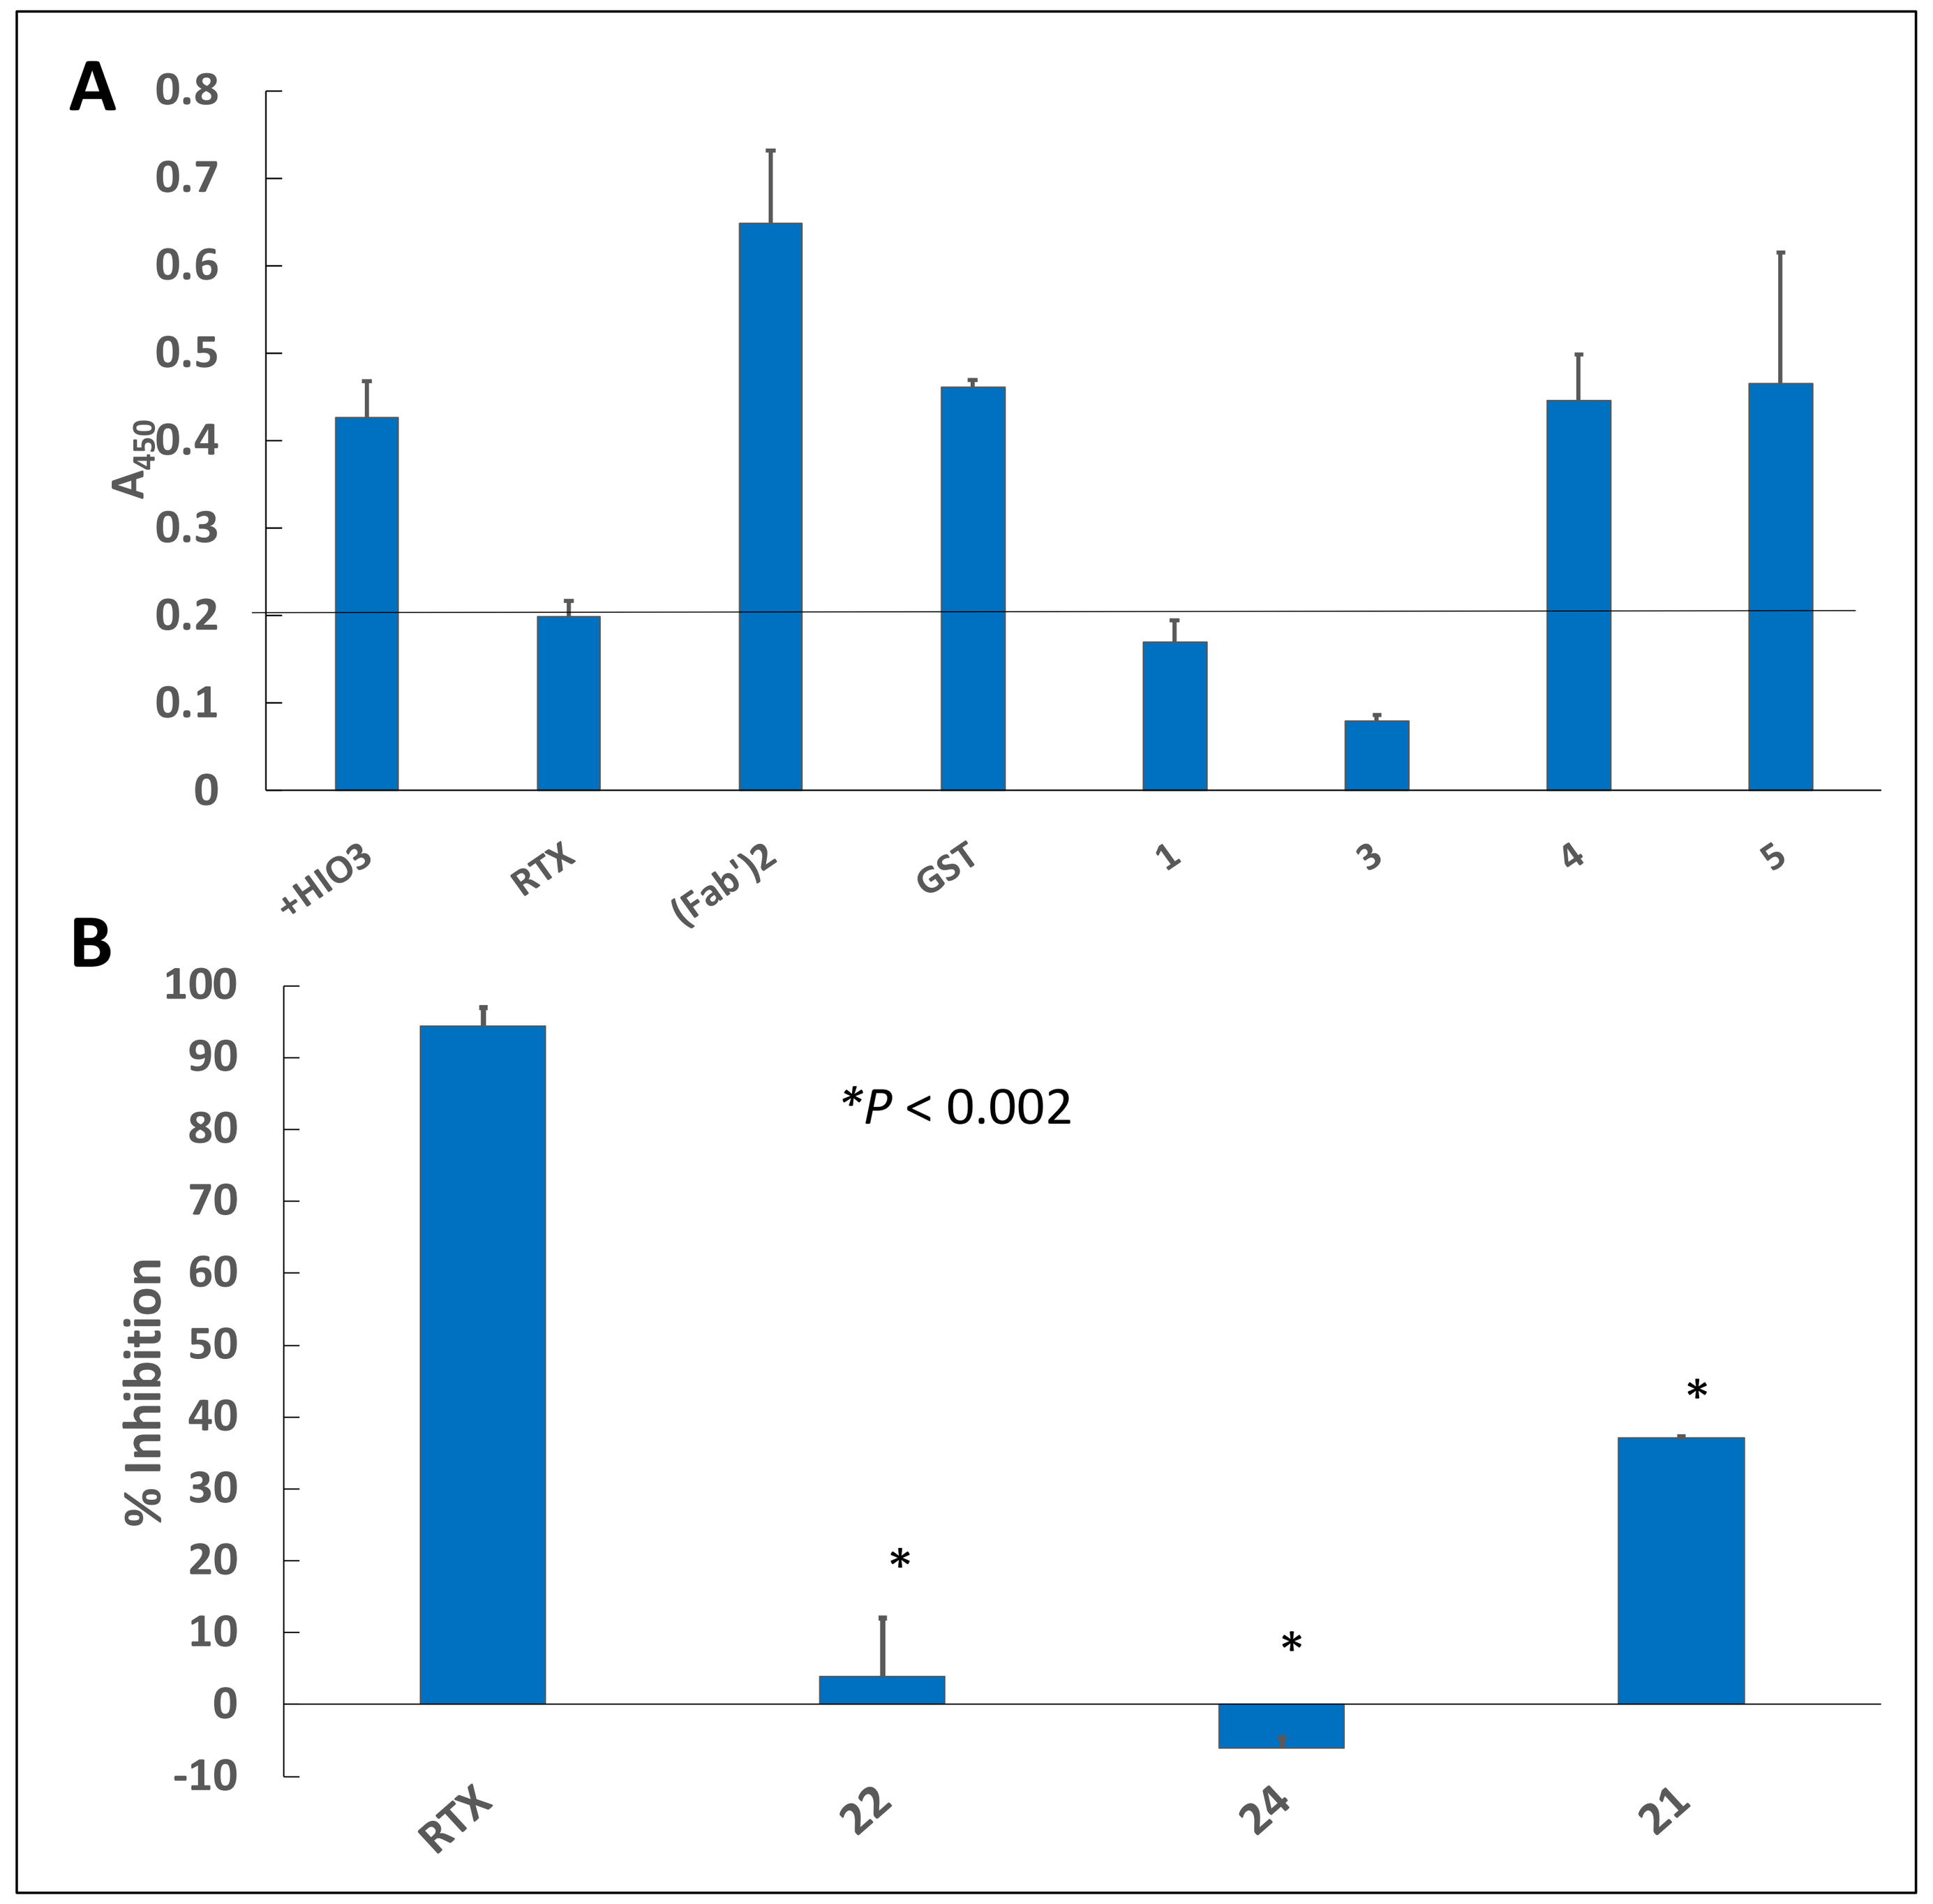
A
A_{450}
+HIO3
RTX
(Fab')2
GST
B
% Inhibition
*P < 0.002
RTX

Fig. 6- RTX-FARV (Seq. 27) has reduced ICAM-1 binding and enhanced ADC activity.

A) Soluble ICAM-1 has significantly reduced binding to RTX-FARV as compared to parental rituximab (RTX). RTX and RTX-FARV were used to immunoprecipitate His-tagged ICAM-1 or mesothelin (neg control) and analysed by Western blot via anti-His-HRP. RTX+ICAM-1 and RTX-FARV+ICAM-1 bands were quantitated using GelQuant.Net (Ver. 1.8.2, Biochemlabsolutions.com) and normalized to RTX band intensity. B) FARV substitution mutant (27) is resistant to ICAM-1 inhibition of ADCC activity using the Jurkat-CD16a ADCC reporter cell line. Daudi target cells were plated in triplicate with Jurkat-CD16-luc cells with 100 ng/mL parental RTX (RTX) or RTX-FARV in the presence of 5 µg/mL ICAM-1. P value indicates significantly less inhibition of FARV mutant by ICAM-1 as compared to WT RTX. C) Downregulation of ICAM-1 enhances antibody-drug conjugate target cell killing. HCT-116 clones with shRNA silenced ICAM-1 expression are susceptible to higher antibody-saporin conjugate toxicity. HCT-116 wt, scrambled shRNA control, and two independent shRNA ICAM-1 silenced clones were treated with trastuzumab-saporin or pertuzumab-saporin conjugates or saporin (ZAP) alone for 96 hours. Wells were washed and remaining viable cells stained with crystal violet to quantitate % cytotoxicity ((1-untreated cells/treated cells)*100%). Significant killing indicated by asterisks.

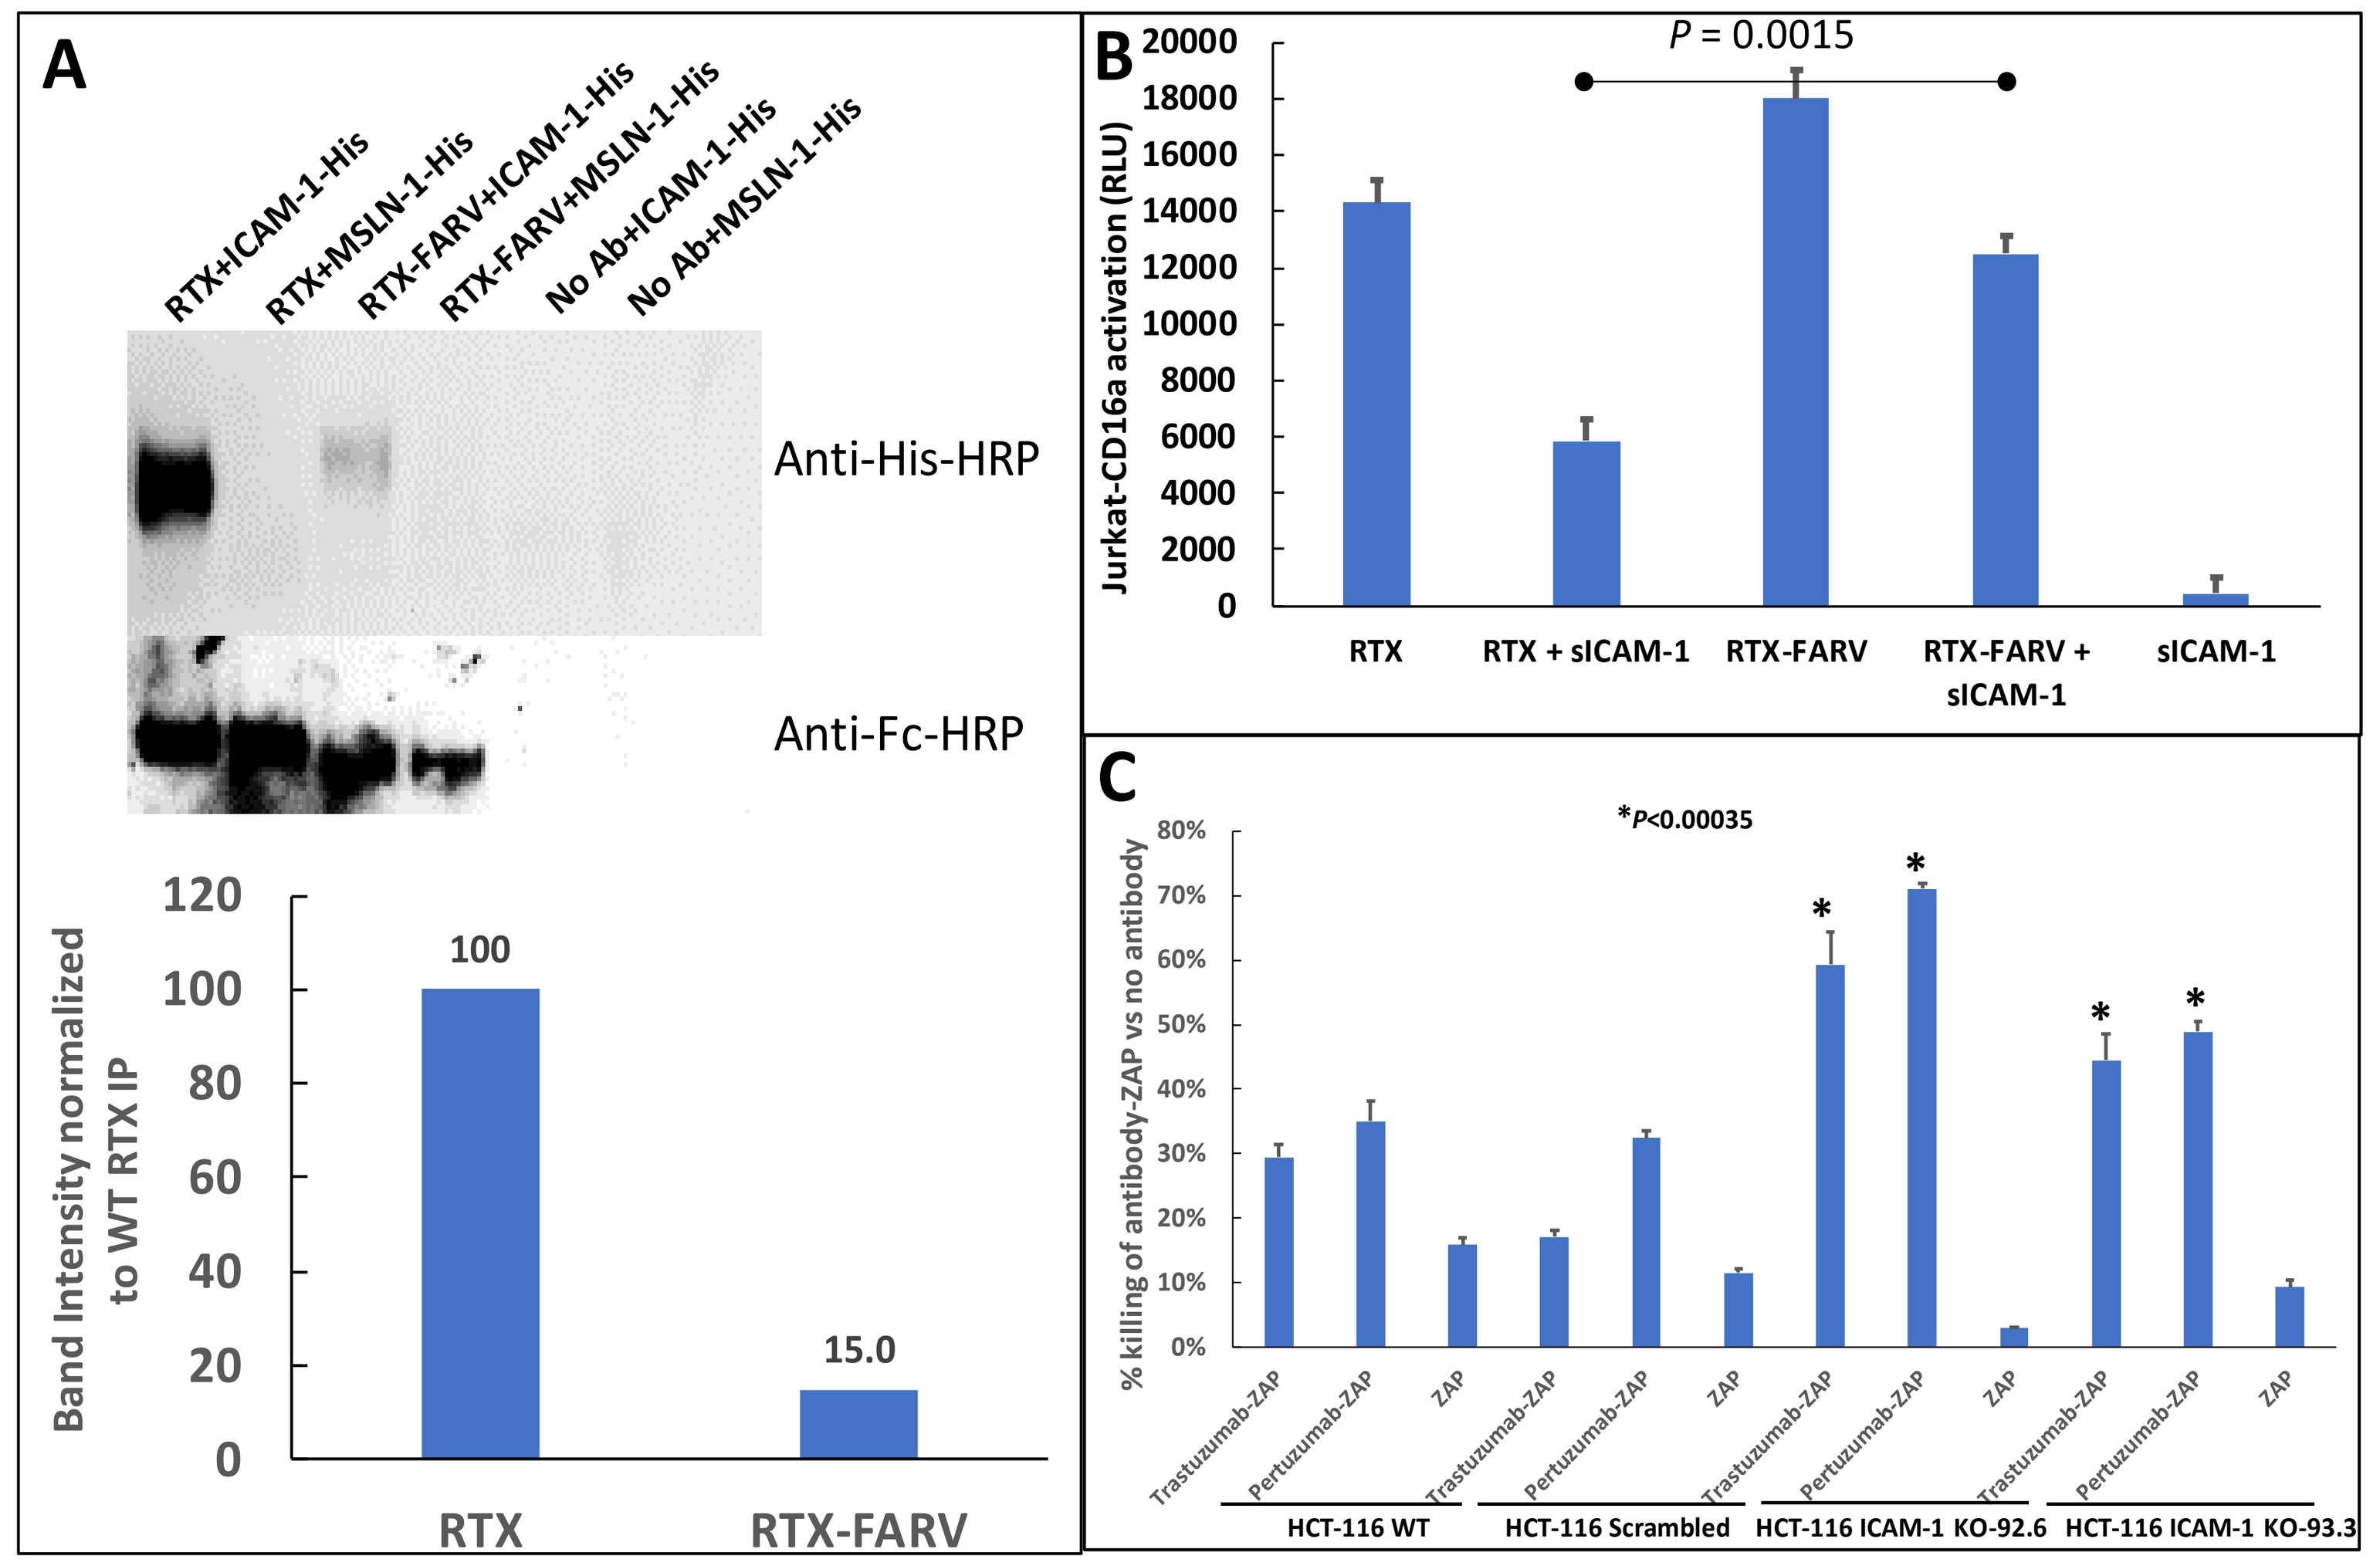
A
RTX+ICAM-1-His
RTX+MSLN-1-His
RTX-FARV+ICAM-1-His
RTX-FARV+MSLN-1-His
No Ab+ICAM-1-His
No Ab+MSLN-1-His
Anti-His-HRP
Anti-Fc-HRP
Band Intensity normalized to WT RTX IP
100
15.0
RTX
RTX-FARV
B
Jurkat-CD16a activation (RLU)
P = 0.0015
RTX
RTX + sICAM-1
RTX-FARV
RTX-FARV + sICAM-1
sICAM-1
C
% killing of antibody-ZAP vs no antibody
*P<0.00035
HCT-116 WT
HCT-116 Scrambled
HCT-116 ICAM-1 KO-92.6
HCT-116 ICAM-1 KO-93.3

Fig. 7- FARV IgG mutants (Seq. 27) are resistant to ICAM-1 binding at intra- and intercellular pHs, as well as ICAM-1 mediated suppressed ADC cytotoxicity as compared to wildtype IgG1 parental antibodies due to their reduced internalization.

A) IgG1-FARV/ICAM-1 binding is unaffected by pH. ELISA plates coated with antibodies were exposed to b-ICAM-1 at low and neutral pH and monitored for ICAM-1 binding. B) % killing of 1 ug/mL (10 nM) TSTZ-ZAP and TSTZ-FARV-ZAP on HCT116 and HCT116-ICAM1-KO cells +/- 10 ug/ml ICAM-1 . As shown, membrane bound and soluble ICAM-1 suppressed wild type IgG1-ADC C) Enhanced FARV mutant killing is a direct result of enhanced antibody internalization. IgG1 mutants containing the conserved 407FARV410 substitution have enhanced internalization rates. HCT116 cells were tested for antibody internalization of parental trastuzumab (TSTZ) and TSTZ-FARV. Each antibody was converted into pHrodo probes and used to measure internalization from incubation time 0 (T0) up to 1,020 hours. As shown, TSTZ-FARV had a significantly more robust internalization than parental TSTZ (P < 0.002). Percent internalization was determined measuring relative fluorescence units (RFU) at various incubation time points vs T0. All data represent triplicate values.

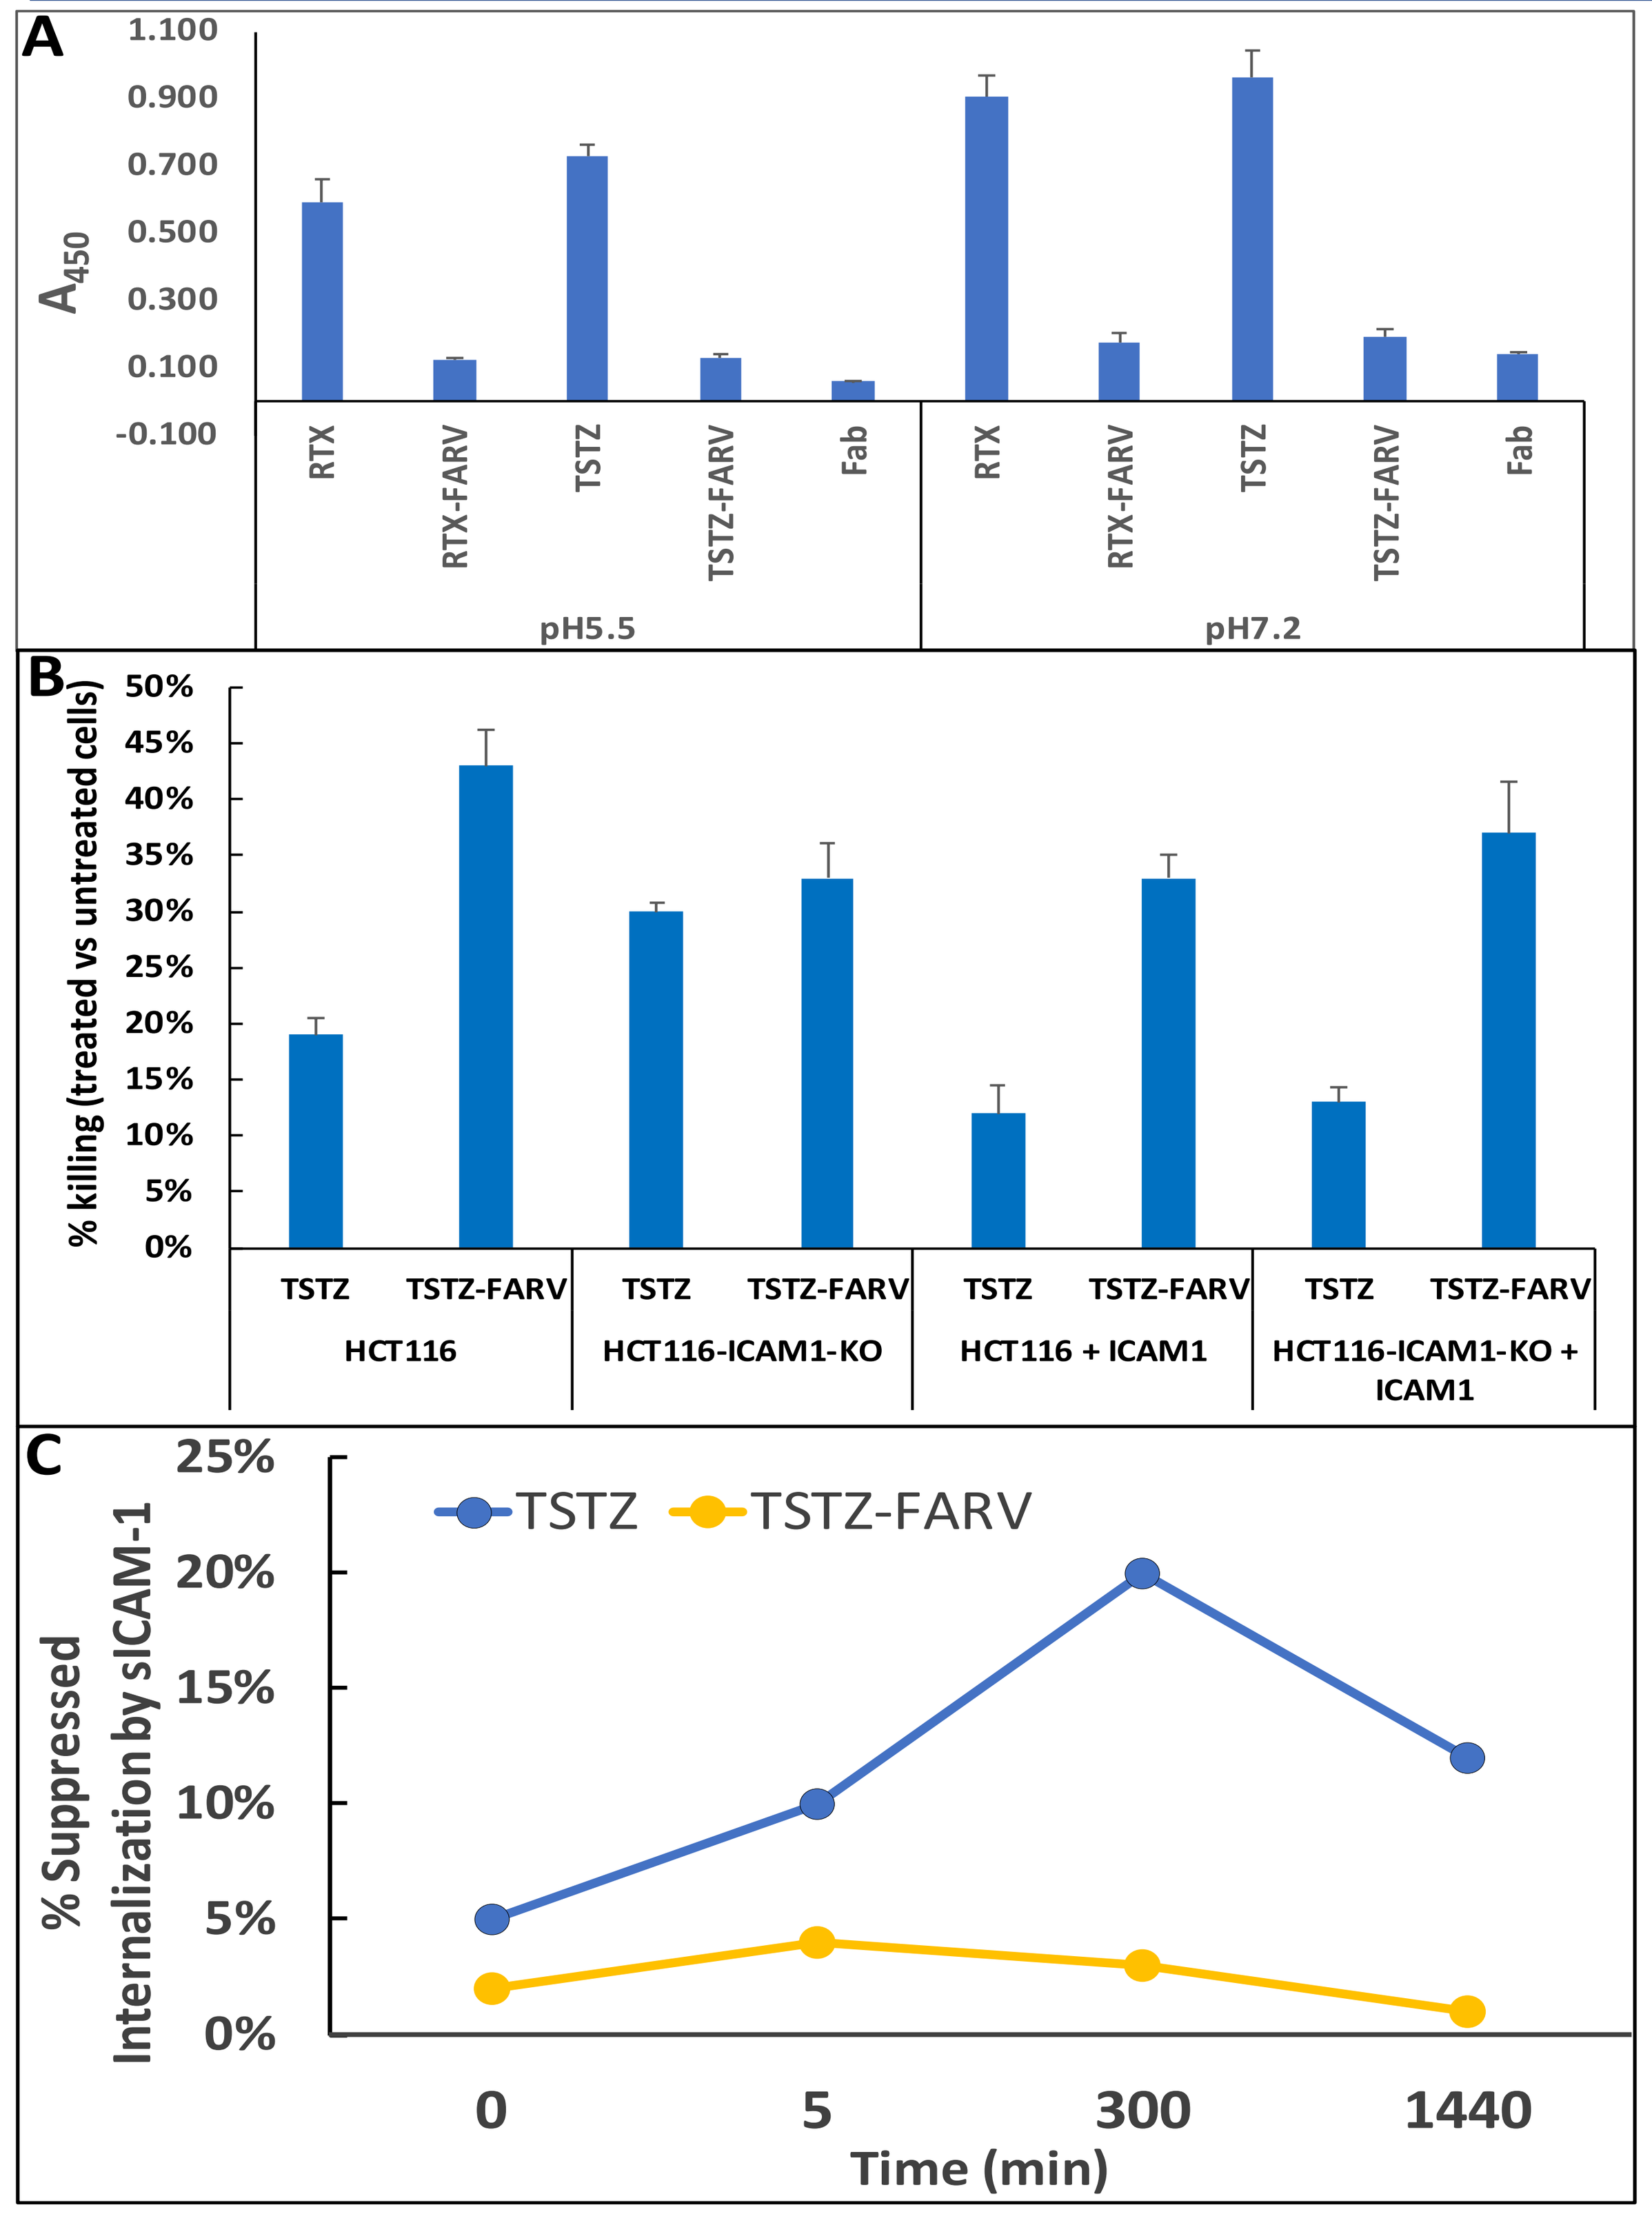
A
A_{450}
RTX
RTX-FARV
TSTZ
TSTZ-FARV
Fab
pH5.5
pH7.2
B
% killing (treated vs untreated cells)
HCT116
HCT116-ICAM1-KO
HCT116 + ICAM1
HCT116-ICAM1-KO + ICAM1
C
% Suppressed Internalization by sICAM-1
TSTZ
TSTZ-FARV
Time (min)

Conclusions & Future Directions

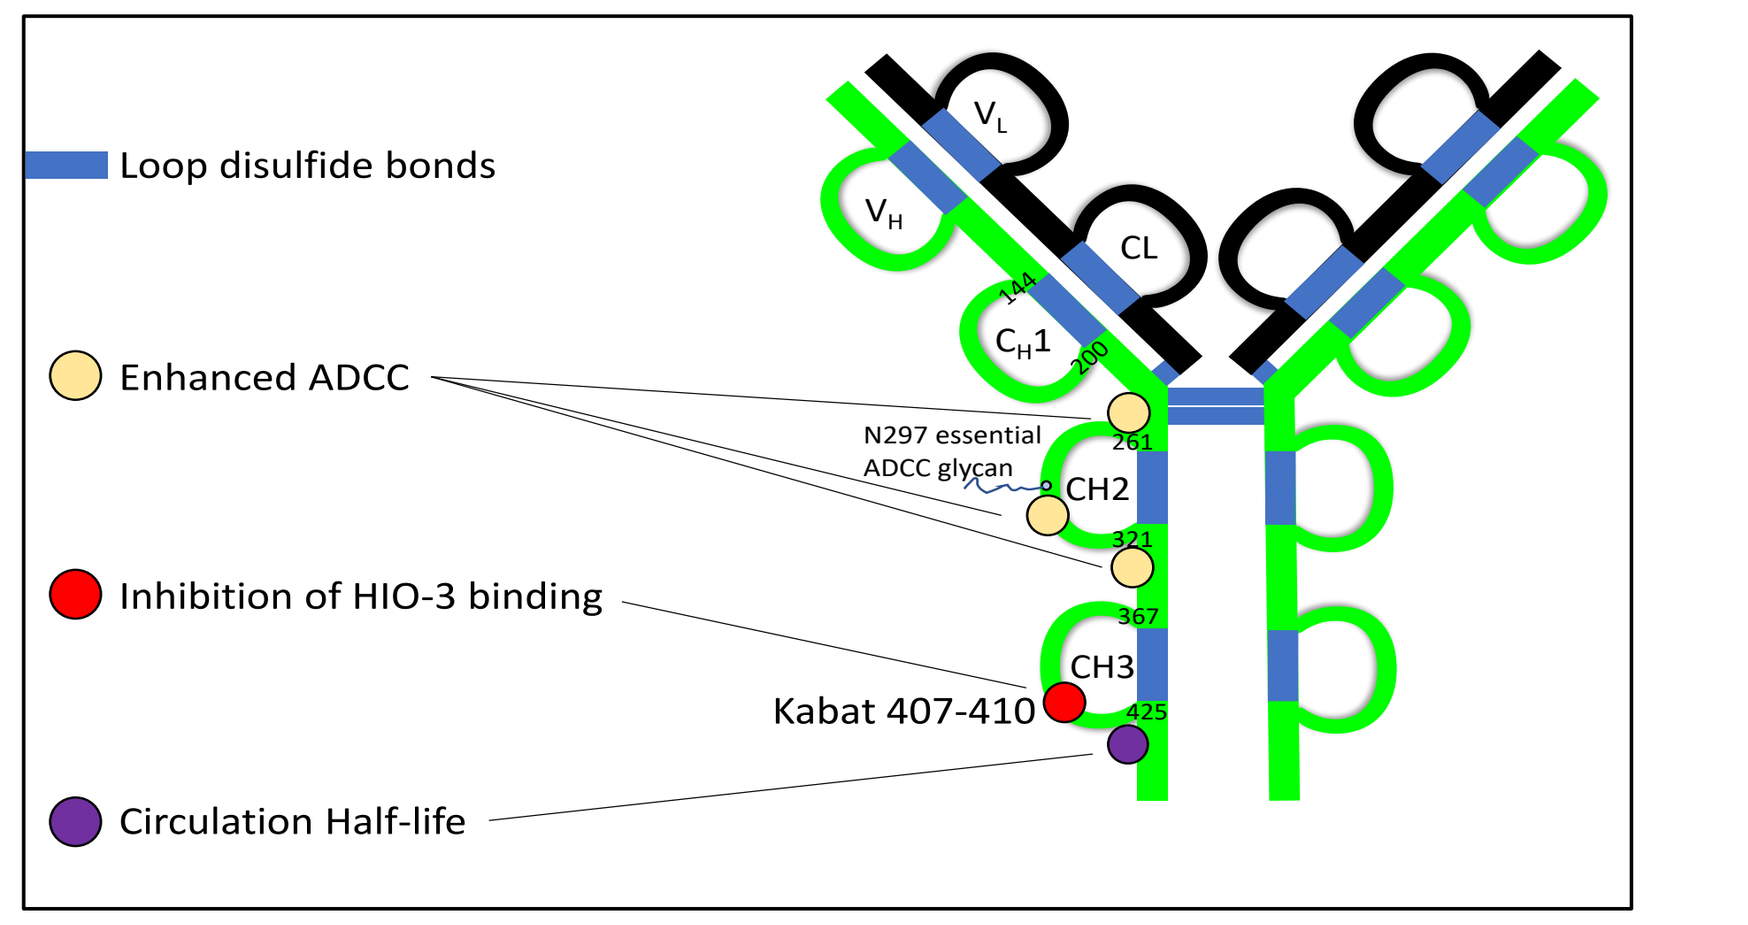
Loop disulfide bonds
Enhanced ADCC
Inhibition of HIO-3 binding
Circulation Half-life
N297 essential ADCC glycan
CH2
CH3
CL
Kabat 407-410

- ICAM-1 is a secreted and membrane-bound tumor protein that inhibits IgG1 ADCC and CDC by blockade of CD16a and C1q engagement, respectively
- Fc substitution mutations localized ICAM-1 binding to the CH3 residues 407-410 and does not overlap with any domains previously known to alter ADCC or CDC functions nor IgG1 circulating half-life (FcRn)
- Most likely spontaneous mutation substitutions in this region (PAM250 matrix, YSKL->FARV) lowered ICAM-1 interactions, resulting in enhanced surrogate ADC killing and uptake
- ICAM-1 refractory antibodies (e.g., cetuximab, daratumumab, rituximab, trastuzumab) are being tested in ICAM-1 expressing cancer models as next gen antibody therapies