Abstract 1622 - Block-Removed Immunoglobulin Technology (BRITE) to enhance rituximab effector function by counteracting CA125-mediated immunosuppression

Luigi Grasso, J. Bradford Kline, and Nicholas C. Nicolaides - Navrogen Inc. Cheyney, PA - luigi@navrogen.com

navrogen
Leaders in Humoral Immuno-Oncology

ABSTRACT - Rituximab is a CD20-targeting antibody that is the standard-of-care for patients with Non-Hodgkin Lymphoma (NHL) cases. Rituximab's mechanism of action includes complement-dependent cytotoxicity (CDC) and antibody-dependent cellular cytotoxicity (ADCC). Recent clinical evidence suggests that high serum levels of the tumor-produced MUC16/CA125 protein has a negative impact on the effectiveness of rituximab clinical activity on up to 40% of follicular lymphoma patients. In this study, we demonstrate that CA125 binds to rituximab and reduces its tumor cell killing activity. Moreover, we describe the generation of a rituximab variant, named NAV-006 (RTX-N109D), using a proprietary technology called Block-Removed Immunoglobulin Technology (BRITE) that employs randomized amino substitution and high-throughput screening to identify CA125-refractory rituximab variants.

CONCLUSION - We demonstrate that NAV-006 (RTX-N109D) is more refractory to the immunosuppressive effects mediated by CA125 as shown by its reduced CA125 interaction and increased ADCC and CDC activity when compared to parent rituximab. These data warrant further investigation of NAV-006 as a next generation anti-CD20 antibody that could improve upon the efficacy of the parent rituximab in NHL patients with high levels of CA125.

Fig. 1 - CA125 inhibits rituximab activity

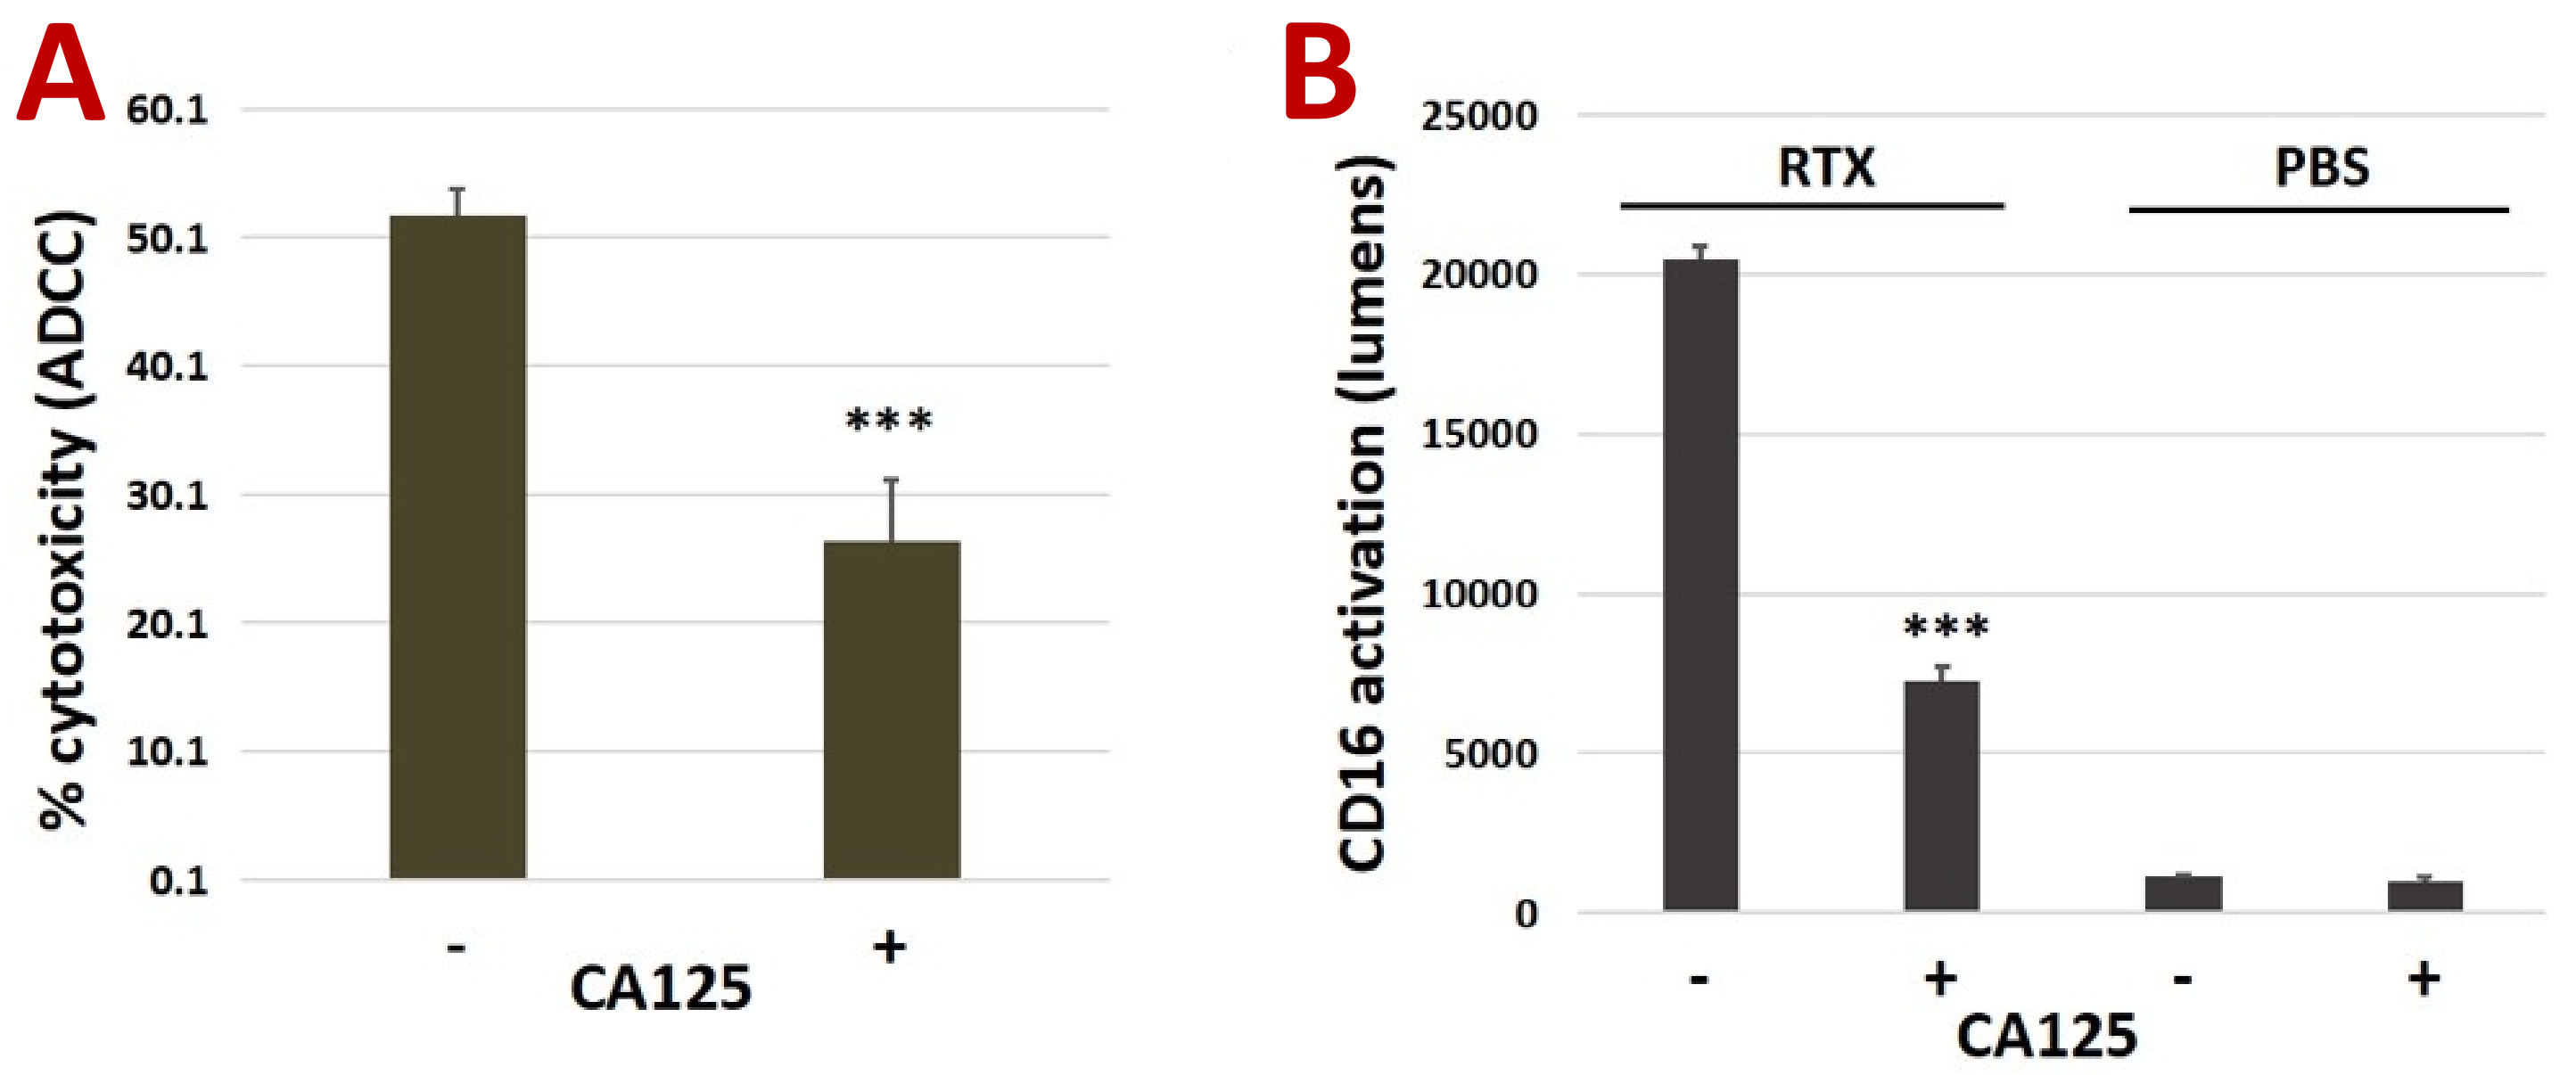
A
% cytotoxicity (ADCC)

CA125
-
+
B
CD16 activation (lumens)
RTX
PBS

CA125

CA125 inhibits RTX-mediated ADCC and Fc receptor/CD16a activation. A) CD20-positive Daudi cells were targeted with RTX (30 µg/mL) and human PBMCs (effector/target ratio of 10:1). CA125 (50,000 U/mL) was adde in some reactions and showed to be immunosuppressive against RTX-mediated ADCC compared to reactions without CA125. B) CD16a activation mediated by RTX was significantly inhibited by CA125.

Fig. 2 - CA125 binds less to RTX N109D

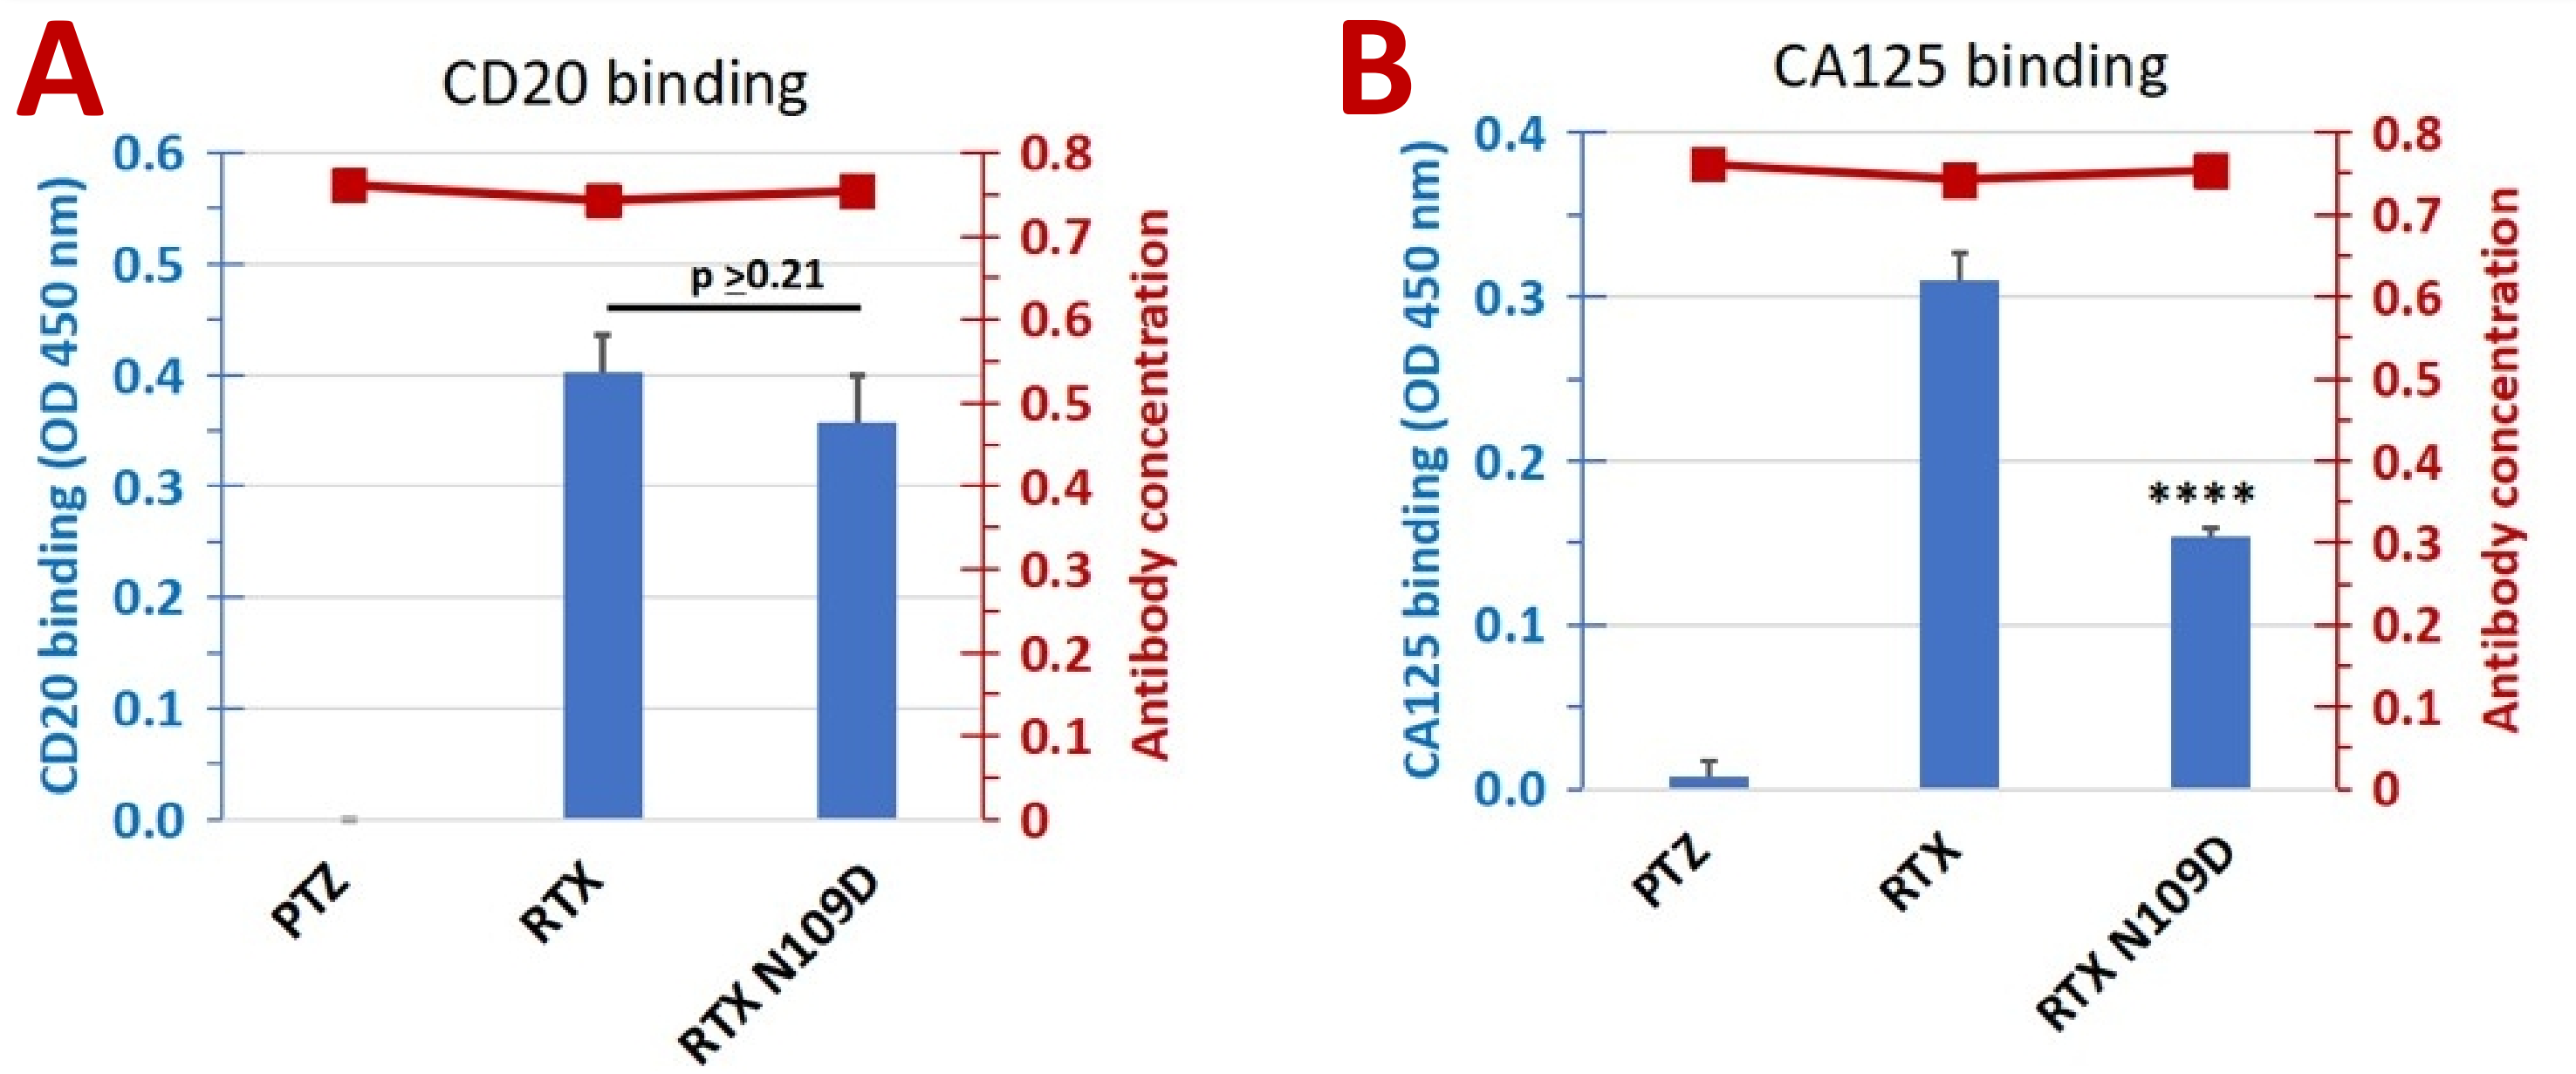
A
CD20 binding
CD20 binding (OD 450 nm)
Antibody concentration
p ≥0.21
PTZ
RTX
RTX N109D
B
CA125 binding
CA125 binding (OD 450 nm)
Antibody concentration

PTZ
RTX
RTX N109D

A) Antibody binding to CD20 was measured by using an ELISA. RTX N109D CD20 binding was comparable to RTX and the difference was not statistically significant (p>0.21). B) Antibody binding to CA125 was measured by using an ELISA. Binding of RTX N109D to CA125 was found to be reduced by 50% compared to RTX and the difference was statistically significant (p<0.00009).

Fig. 3 - Parent RTX vs. RTX N109D affinity

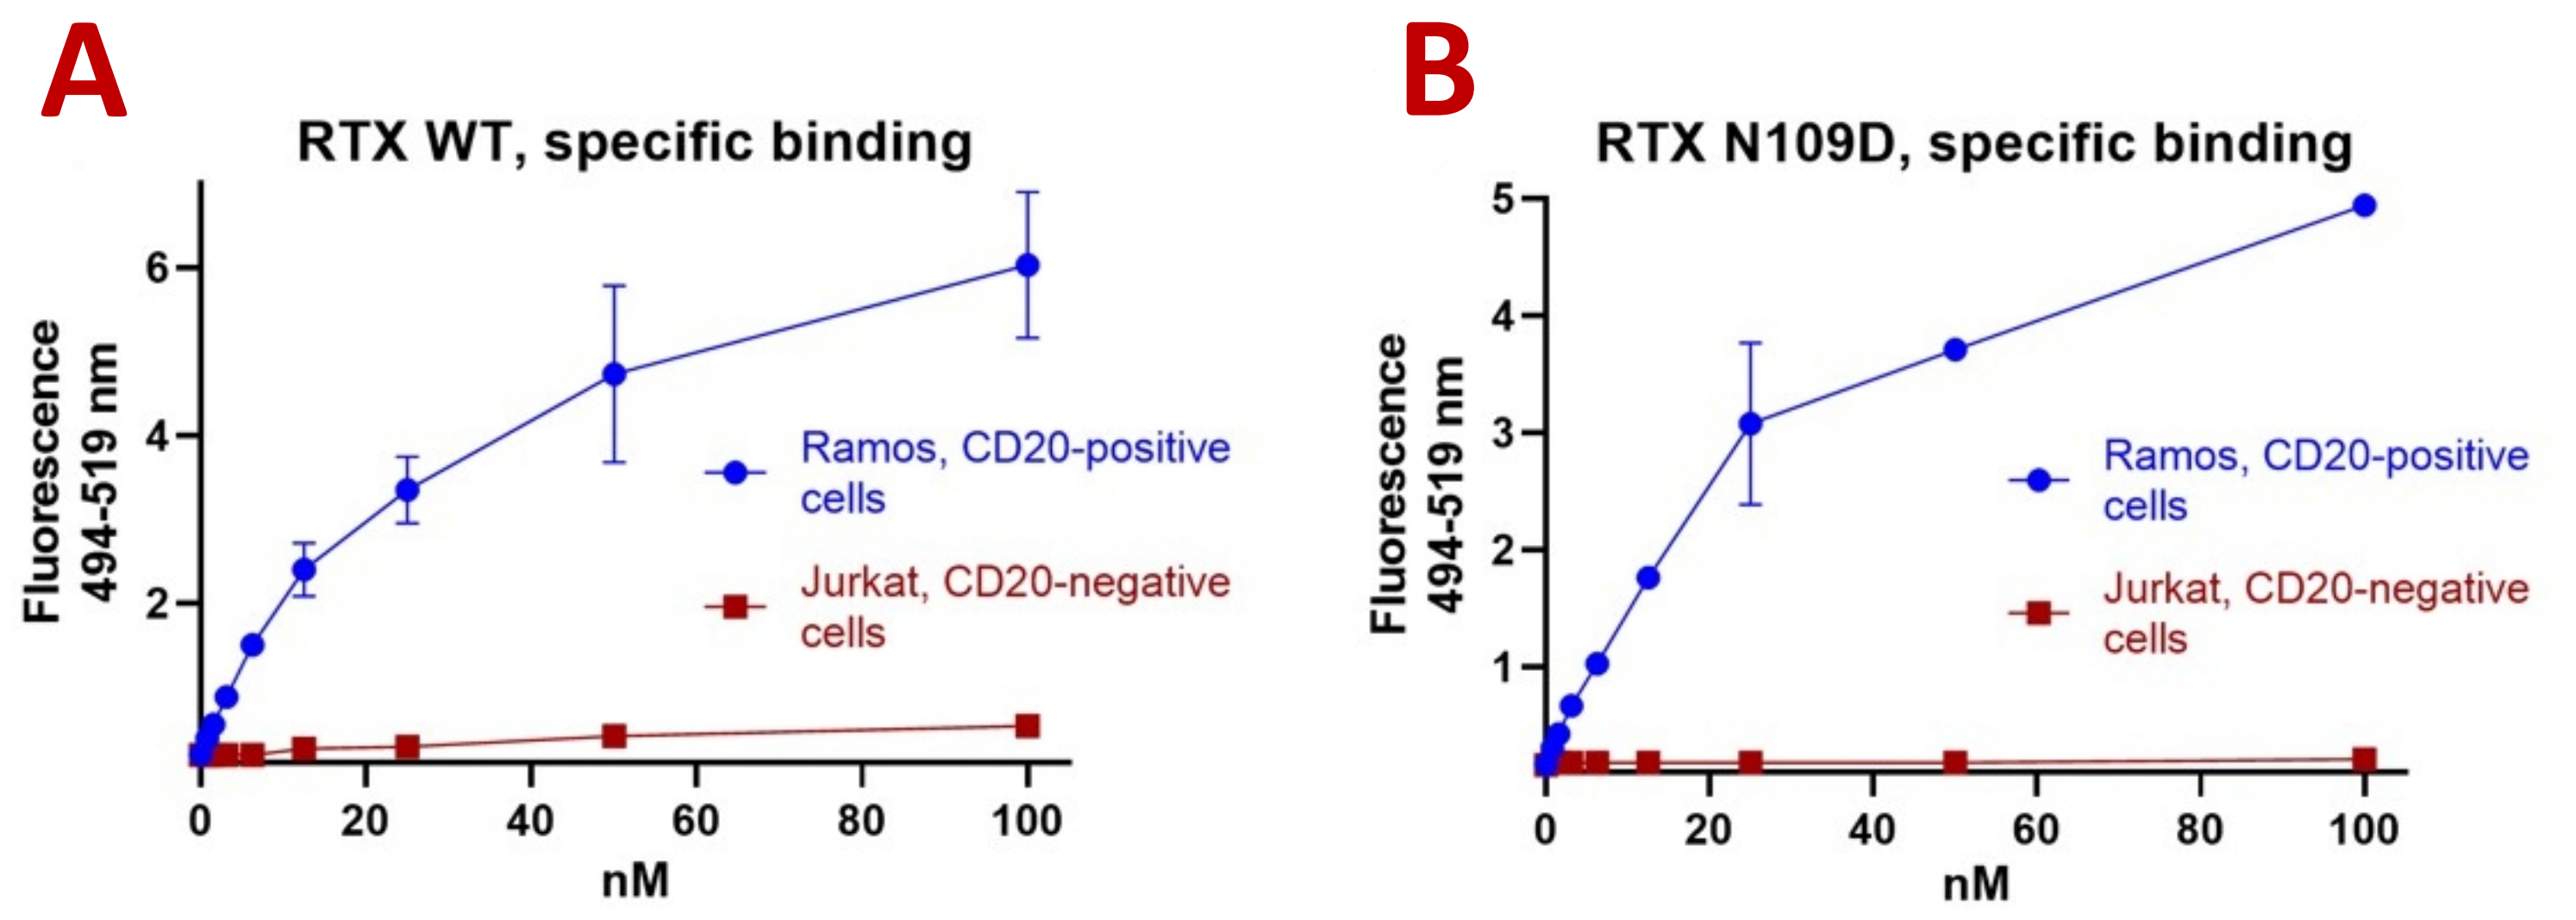
A
RTX WT, specific binding
Fluorescence 494-519 nm
Ramos, CD20-positive cells
Jurkat, CD20-negative cells
nM
B
RTX N109D, specific binding
Fluorescence 494-519 nm
Ramos, CD20-positive cells
Jurkat, CD20-negative cells
nM

Parent (WT) RTX (panel A) and RTX N109D (panel B) were directly conjugated to AlexaFluor 488 dye and used to stain live cells at concentration ranging 0.78 to 100 nM. Specific binding to CD20-positive and negative cells were measured. Affinity/Kd for RTX and RTX N109D were 20.6 and 30.5 nM, respectively. Overall RTX N109D showed CD20-specific binding and affinity comparable to parent RTX.

Fig. 4 - RTX N109D has enhanced CDC

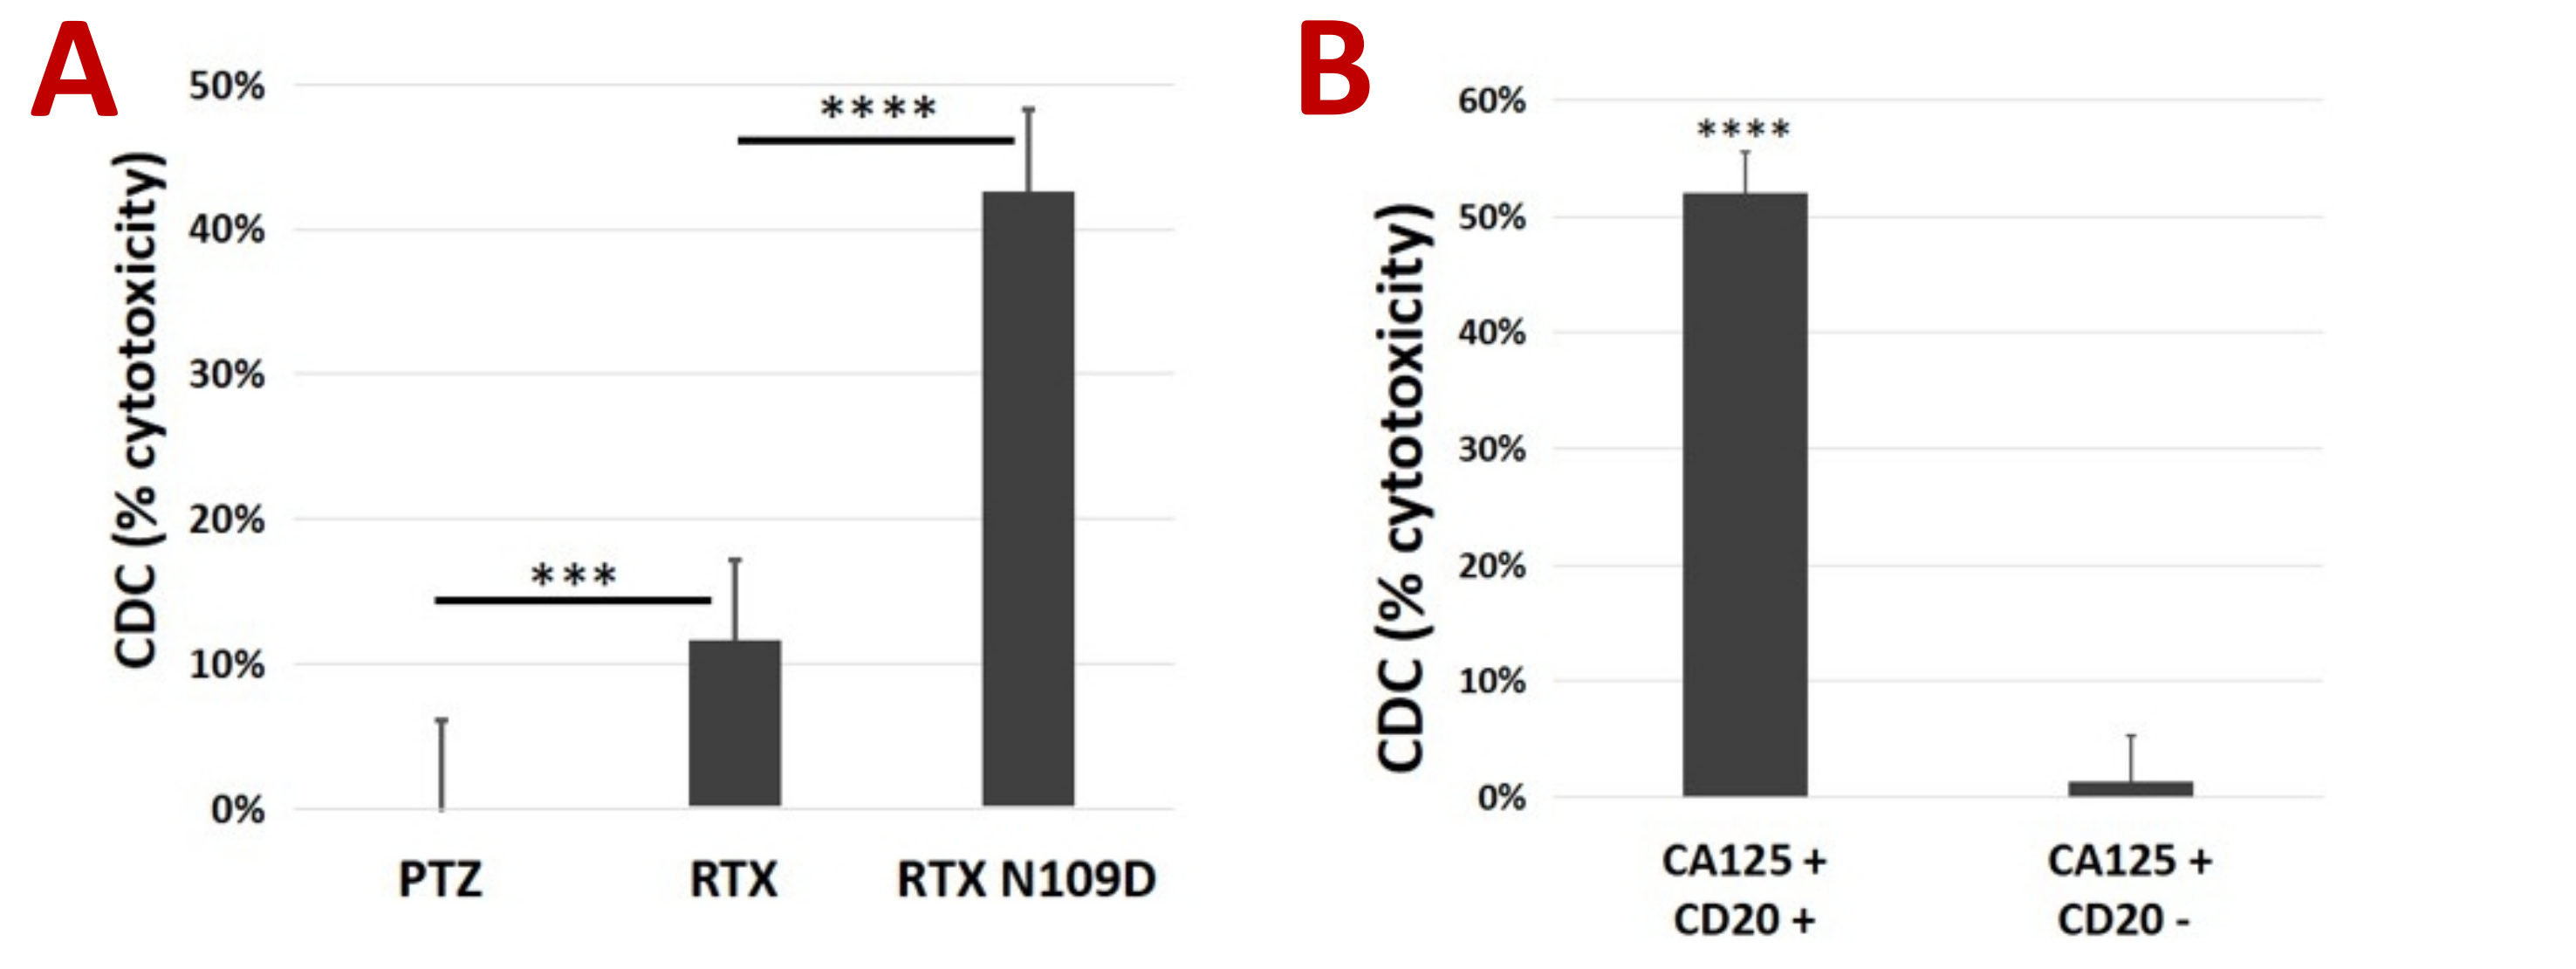
A
CDC (% cytotoxicity)

PTZ
RTX
RTX N109D
B
CDC (% cytotoxicity)

CA125 + CD20 +
CA125 + CD20 -

A) CD20-positive cancer cells expressing CA125 were targeted with antibodies in the presence of complement. RTX N109D shows superior CDC activity against CA125-positive cells compared to parent RTX. B) CD20-positive and CD20-negative cancer cells expressing CA125 were targeted with RTX N109D in the presence of complement. RTX N109D specifically killed only CD20-postive cells

Fig. 5 - RTX N109D has enhanced ADCC

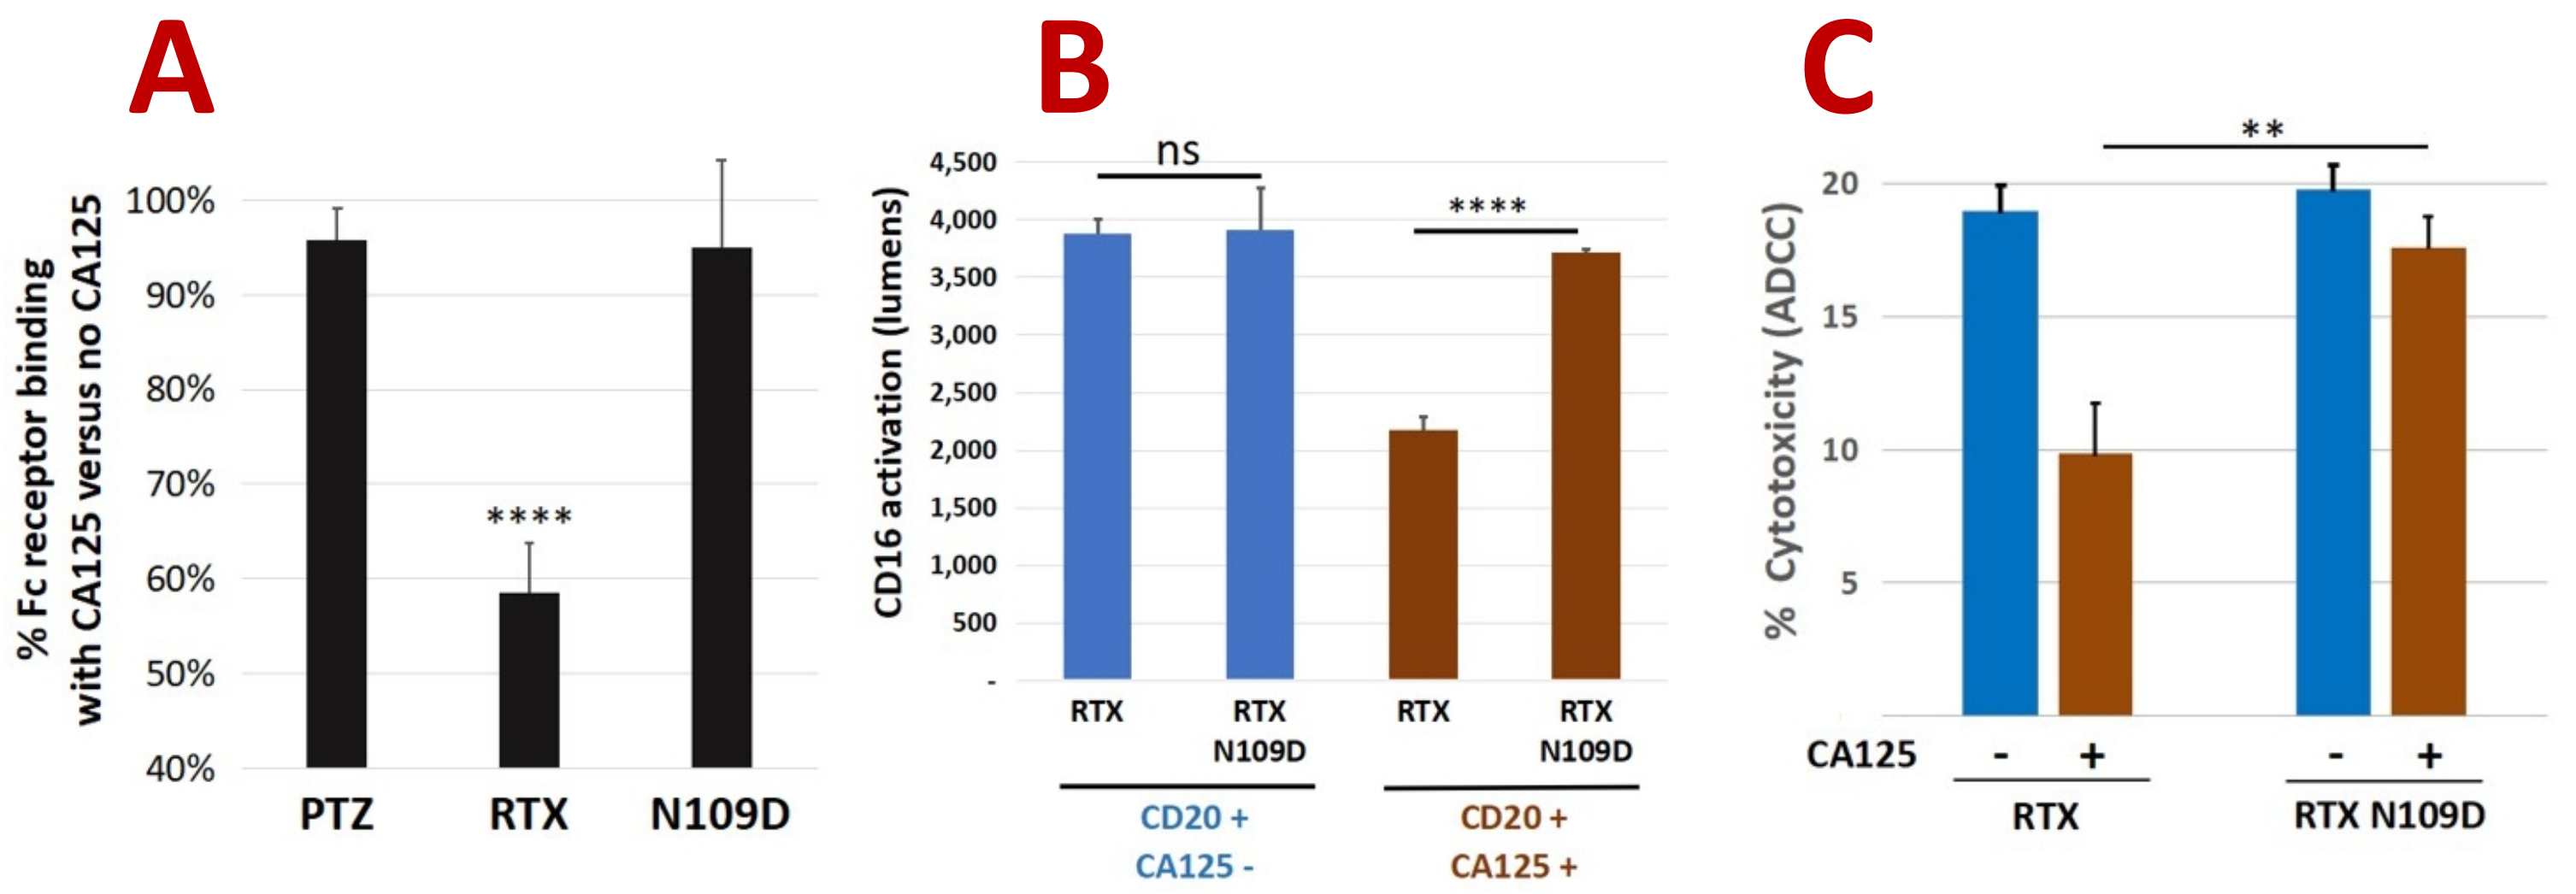
A
% Fc receptor binding with CA125 versus no CA125

PTZ
RTX
N109D
B
CD16 activation (lumens)
ns

RTX
RTX N109D
RTX
RTX N109D
CD20 + CA125 -
CD20 + CA125 +
C
% Cytotoxicity (ADCC)
**
CA125
-
+
RTX
RTX N109D

A) RTX N109D retains >90% Fc receptor binding in the presence of CA125 compared to <60% for RTX. B) CD16a receptor activation mediated by RTX N109D in the presence of CA125 is significantly higher than parent RTX (p <0.0001). C) Human PBMCs-mediated killing of Daudi target cells (effector:target cells ratio 5:1). ADCC mediated by RTX N109D in the presence of CA125 is significantly higher than parent RTX (p <0.01).