Abstract 1703 Development of NAV-005, a humoral immuno-suppressor antagonist for the treatment of CA125-expressing cancers

J. Bradford Kline, Luigi Grasso, and Nicholas C. Nicolaides - Navrogen Inc. Cheyney, PA email: nick@navrogen.com

navrogen

ABSTRACT **- Tumors employ a variety of mechanisms to avoid host cellular and humoral immune responses. Recent findings have found that the tumor-produced MUC16/CA125 protein elicits humoral immunosuppressive effects on antibody-mediated tumor cell killing. This effect is facilitated via direct binding to SIGLEC regulatory receptors on natural killer (NK) cells as well as through direct binding to therapeutic antibodies that blocks Fc-γ-receptor and C1q antibody engagement, leading to suppressed antibody dependent cellular cytotoxicity (ADCC) and complement dependent cytotoxicity (CDC) activities, respectively. We have engineered a human IgG1 Fc fusion protein, called NAV-005, that binds to CA125 with high affinity and blocks its binding to SIGLEC receptors and affected antibodies, thus enabling efficient humoral immune mediated tumor cell killing. NAV-005 is being developed as a novel therapeutic agent to treat humoral immuno-suppressed, CA125 expressing cancers.**

CONCLUSIONS

- **The novel dipeptide-Fc fusion NAV-005 binds with high affinity to CA125**
- **NAV-005 blocks the immuno-effector (IE) inhibition of CA125**
- **NAV-005 restores ADCC and CDC mediated killing of CA125 expressing tumors**
- **NAV-005 blocks the binding of CA125 to both cancer-targeting antibodies as well as SIGLEC-9 inhibitory receptor on NK effector cells**

Figure 1. Follicular Lymphoma (FL) patients with elevated CA125 have reduced response to rituximab (RTX)

Rituximab (RTX) is a commercially approved antibody (Ab) based drug for the treatment of FL. It's mode of function has been shown to involve ADCC and CDC, as well as Ab-induced apoptosis. CA125 binds RTX and suppresses its ADCC and CDC activity and its over-expression has been associated with the reduced clinical response in patients treated with RTX in the front-line setting, suggesting that it may have an inhibitory role in clinical response in patients with CA125-postive FL.

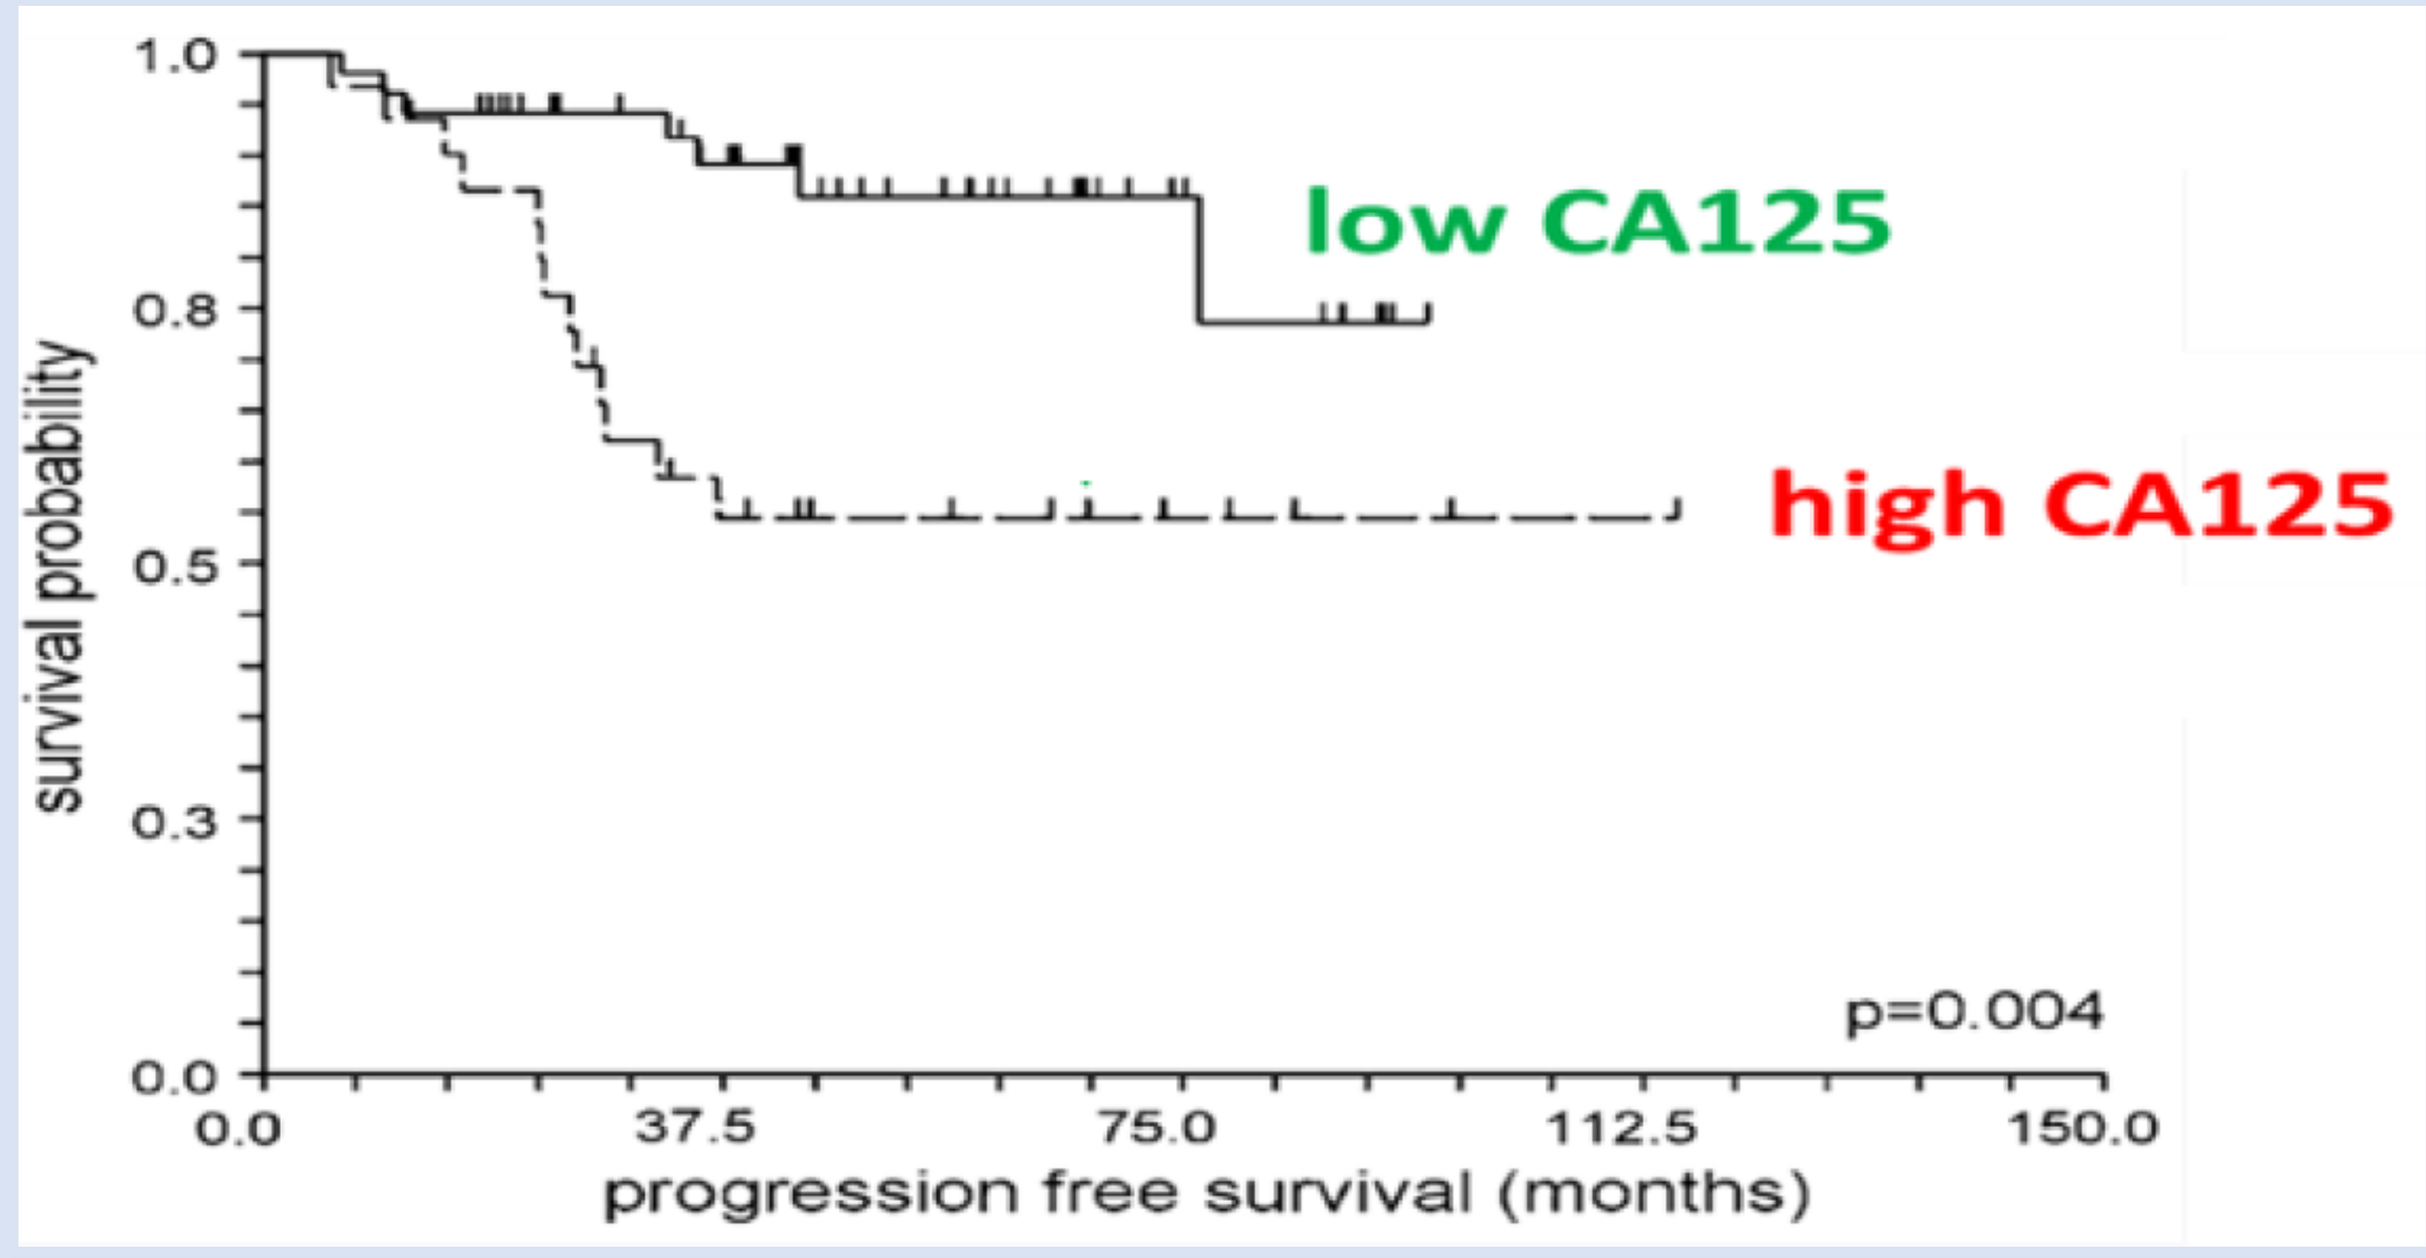

survival probability
1.0
0.8
0.5
0.3
0.0
0.0
37.5
75.0
112.5
150.0
progression free survival (months)
low CA125
high CA125
p=0.004

Fig.1. Follicular lymphoma patients with elevated CA125 have reduced response to RTX therapy

Figure 2. Soluble CA125 inhibits RTX ADCC activity in vitro

Several studies have recently described mechanisms by which CA125 impacts Ab immune-effector (IE) functions. Kline et al. reported that CA125 was capable of binding to a subset of IgG1-type Abs via the Fab domain that in turn perturbed their Fc interaction with the CD16a and CD32a Fc-γ-activating receptors on NK and myeloid cells as well as with the C1q complement initiating protein (1,2). In Figure 2, we targeted Daudi (CD20+) lymphoma cells with RTX for ADCC using human PBMCs as a source of effector cells +/- soluble CA125. In the presence of soluble CA125, significant inhibition of RTX ADCC activity was observed.

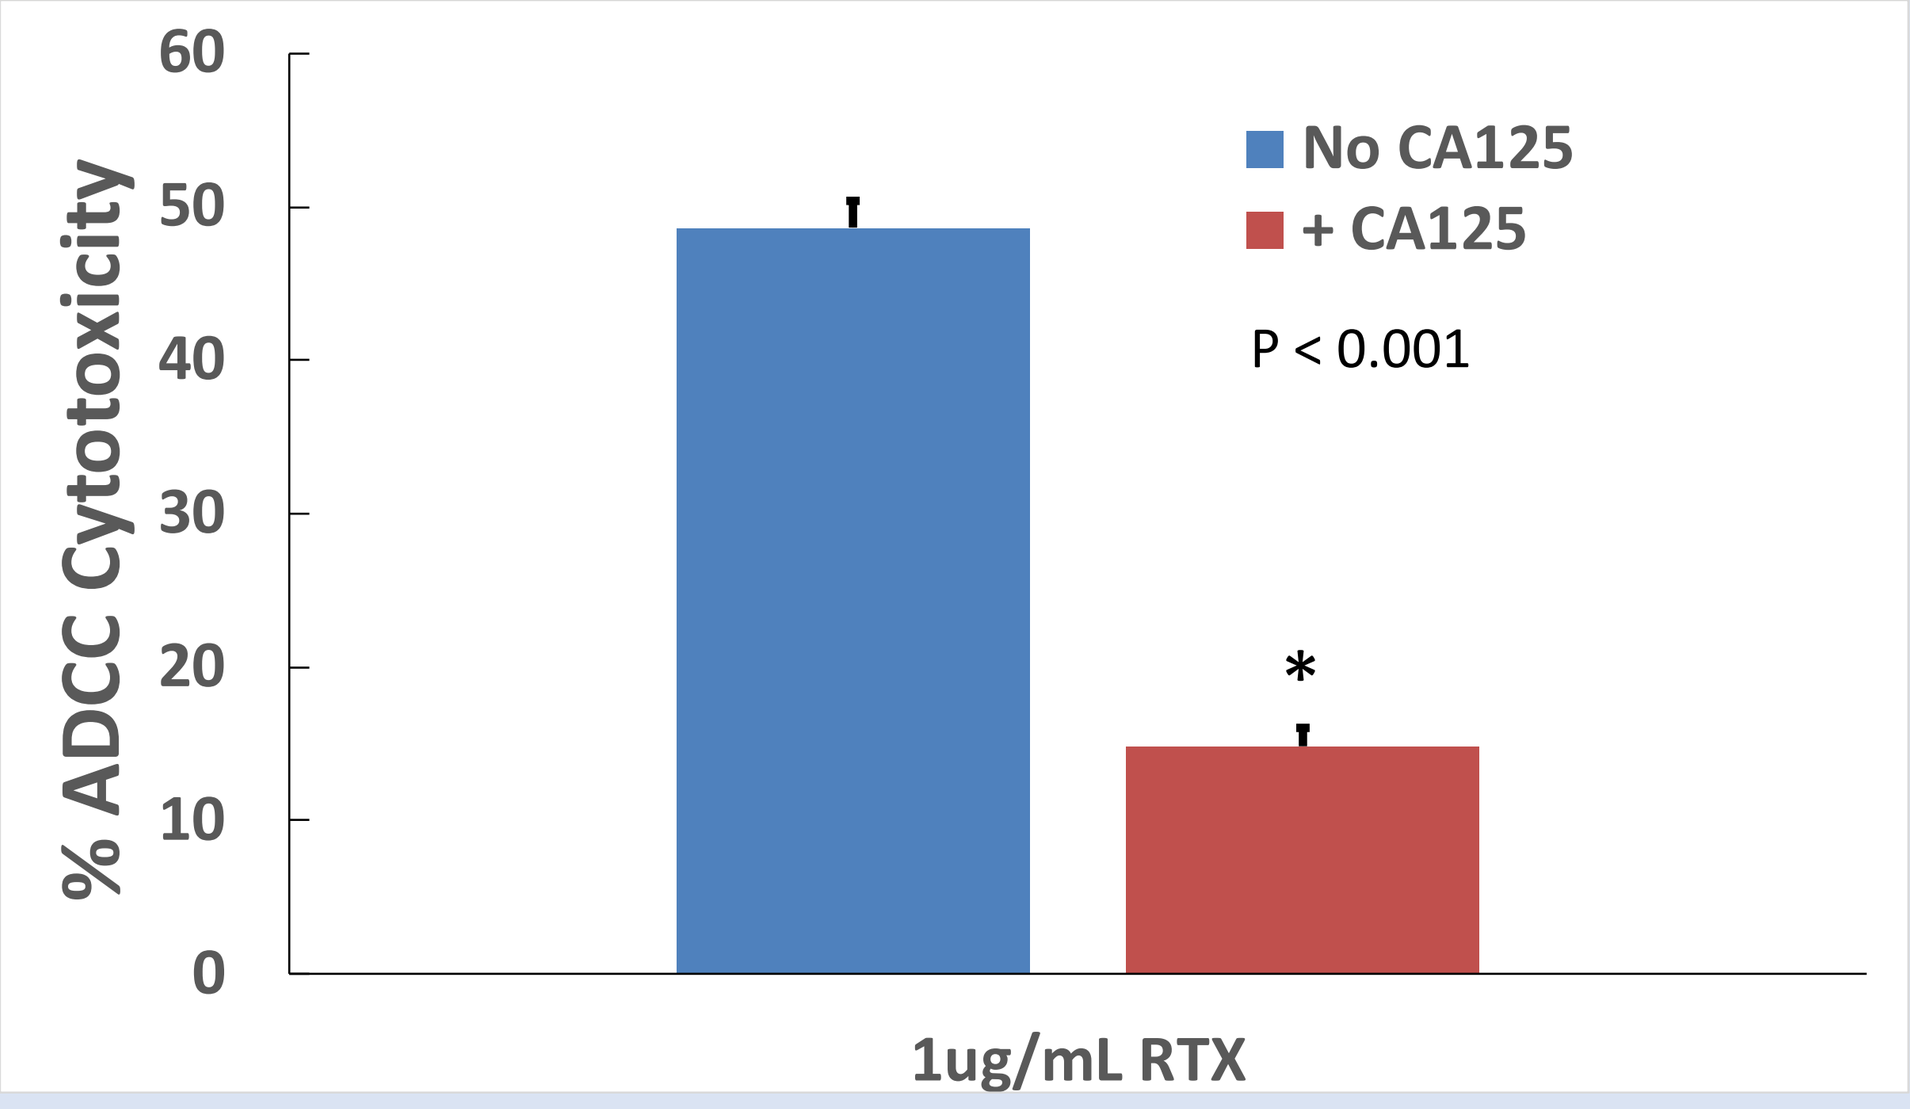

% ADCC Cytotoxicity
No CA125
+ CA125
P < 0.001
1ug/mL RTX

Fig.2. RTX ADCC activity is inhibited by soluble CA125. $2x10^5$ Daudi cells were placed in culture with $2.75x10^5$ PBMCs (Effector:Target cell ratio 14:1) for 6 hrs at 37°C at 5% CO_2. % ADCC was determined using LDH release as follows:

(% cytotoxicity = RTX treated cells – effector cells without RTX / triton X lysed cells – effector cells only)

Figures 3 & 4. Discovery of a novel tandem di-peptide that overcomes the immune-effector (IE) inhibitory activity of CA125

In order to identify agents that can potentially block CA125 binding to an affected Ab, we employed our Block Removed Immunoglobulin Technology (*BRITE*) platform to screen chemical and peptide libraries for potential inhibitors that could specifically block the CA125-RTX interaction. As shown in Fig. 3A, a peptide (referred to as PEP5) appeared to suppress CA125 binding to RTX as compared to controls. This peptide was further evaluated in various assays for its ability to overcome the suppressed IE activity of RTX in the presence of CA125. As CA125 inhibits ADCC via the suppression of CD16a or CD32a binding to antibody Fc, PEP5 was tested for the ability to overcome CA125 suppressed CD16a binding to RTX via ELISA. RTX coated plates were probed with biotinylated human CD16a probe +/- varying amounts of PEP5 or a nonspecific peptide (PEP1). As shown in Fig. 3B, PEP5 was able to antagonize CA125's suppressed CD16a binding to RTX at levels similar to CD16a-RTX binding alone. A similar effect was observed using biotinylated C1q protein to probe immobilized RTX +/- CA125 plus PEP5 (Fig. 3C). Moreover, an ELISA using biotinylated PEP5 showed that it elicits its effect via direct binding to CA125 (Fig. 3D).

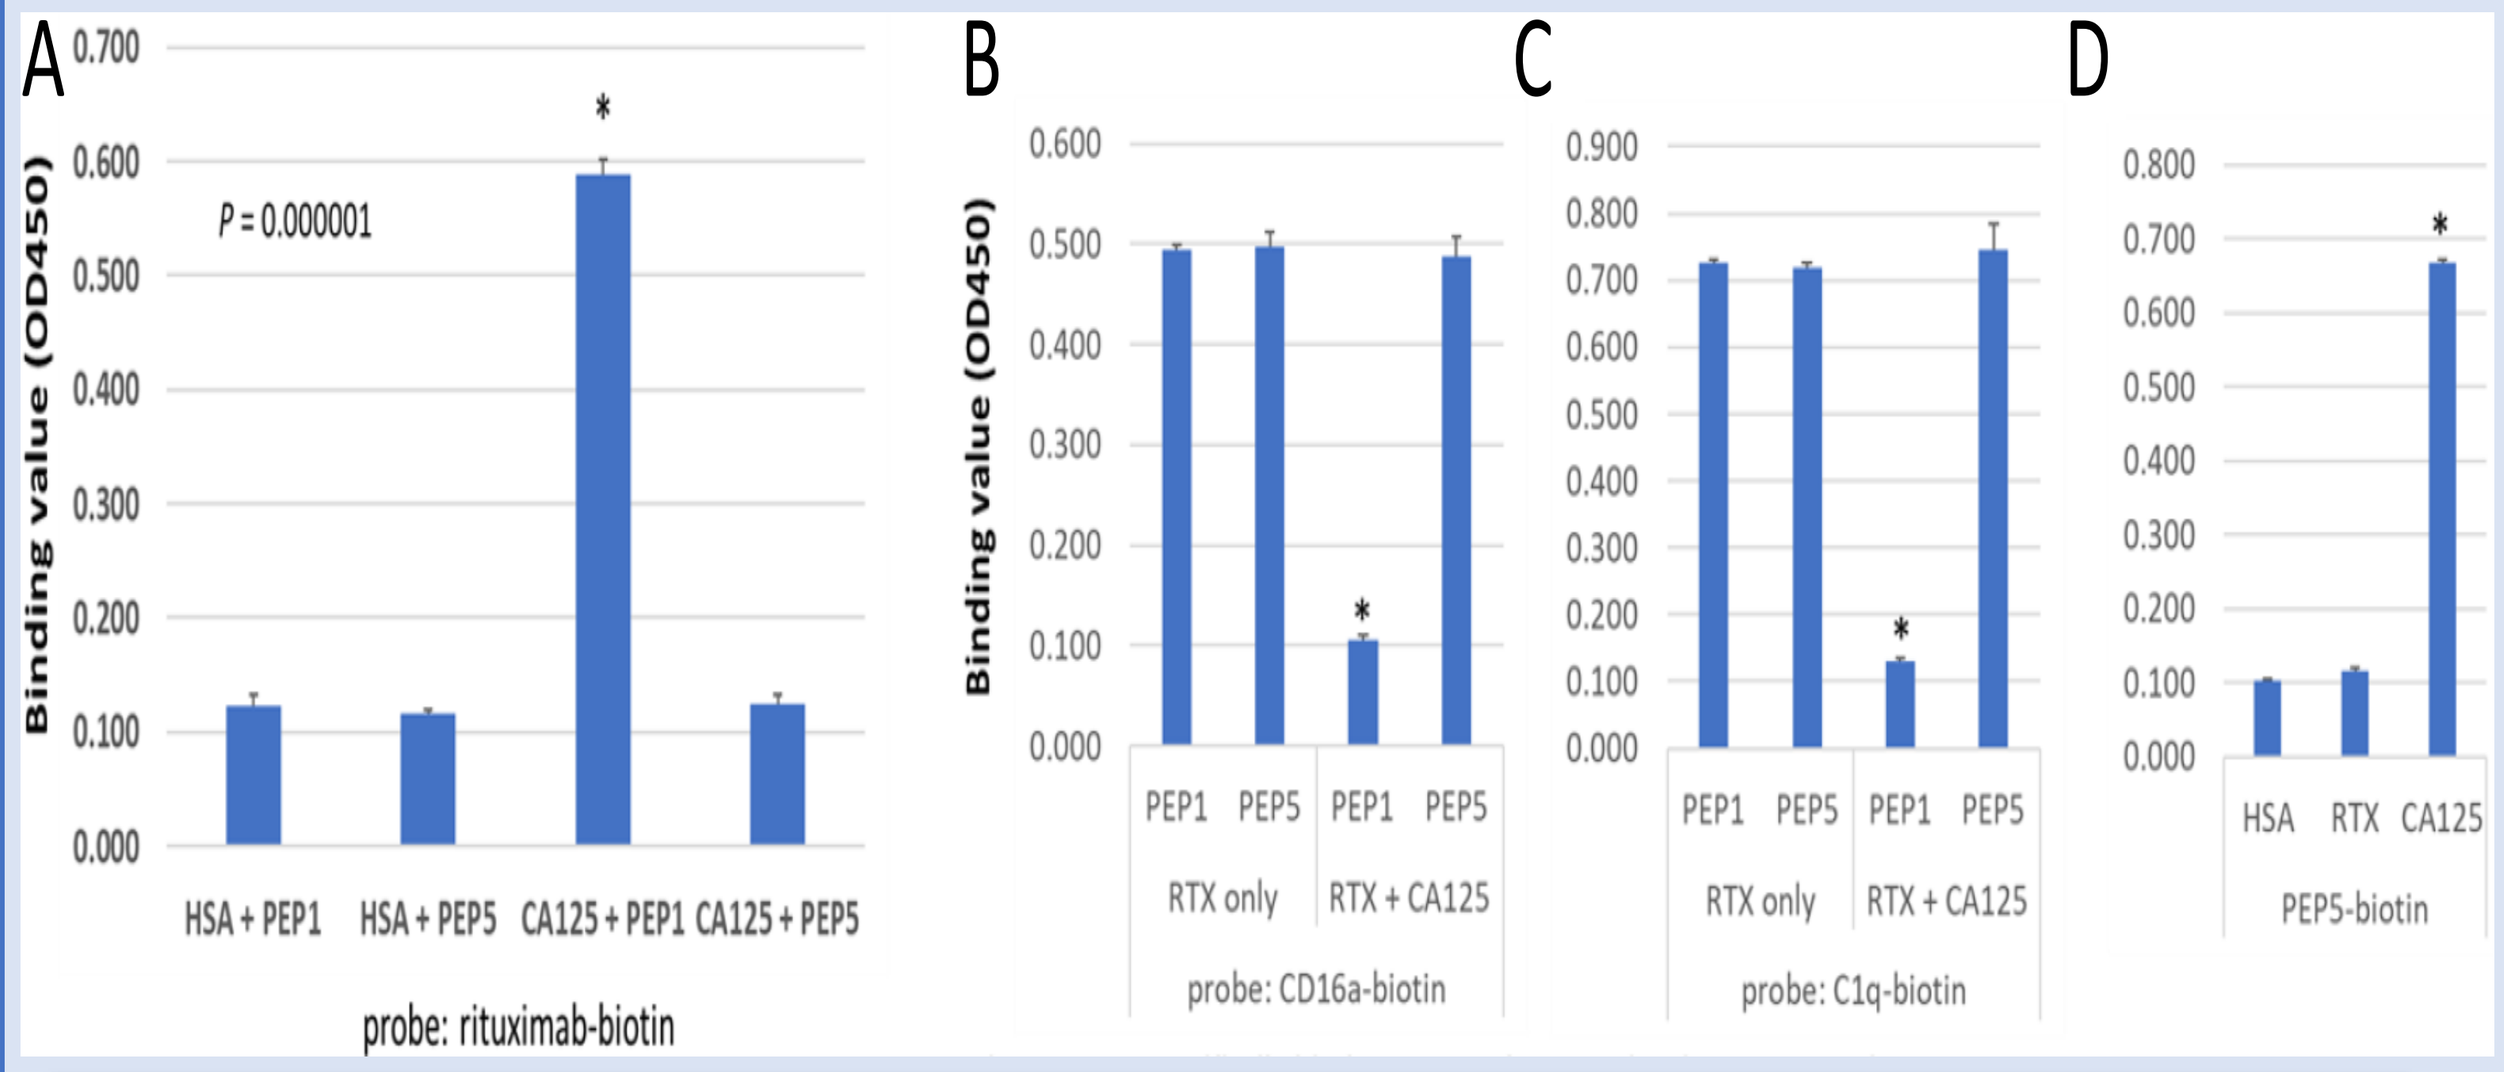

A
Binding value (OD450)
P = 0.000001
HSA + PEP1
HSA + PEP5
CA125 + PEP1
CA125 + PEP5
probe: rituximab-biotin
B
Binding value (OD450)
PEP1
PEP5
RTX only
RTX + CA125
probe: CD16a-biotin
C
RTX only
RTX + CA125
probe: C1q-biotin
D
HSA
RTX
CA125
PEP5-biotin

Fig.3. Discovery and characterization of NAV-005 peptide (PEP5). A, PEP5 can block CA125 binding to RTX. 96 well ELISA plates coated with human serum albumin (HSA) or CA125 were probed with 1 μg/mL biotin-RTX +/- 1 μg/mL PEP5 or an irrelevant peptide (PEP1) in triplicate. Panels B-D, PEP5 specifically binds CA125 and antagonizes its IE suppression on RTX. ELISA plates were coated with RTX and probed with biotin-CD16a (B), or biotin-C1q (C) +/- 15 KU/mL CA125 and 1 μg/mL PEP1 or PEP5. D, PEP5 directly binds to CA125. ELISA plates were coated with HSA, RTX, or 15 KU/mL CA125 and probed with biotin-PEP5. P < 0.001 in panels B-D using T-test.

Similar effects were observed in cell-based assays using Chinese hamster ovary cells stably expressing human CD20 (CHO-CD20), whereby CA125 significantly suppressed RTX CDC activity on CHO-CD20 cells and was reversed in a dose-dependent manner by PEP5 (Fig. 4).

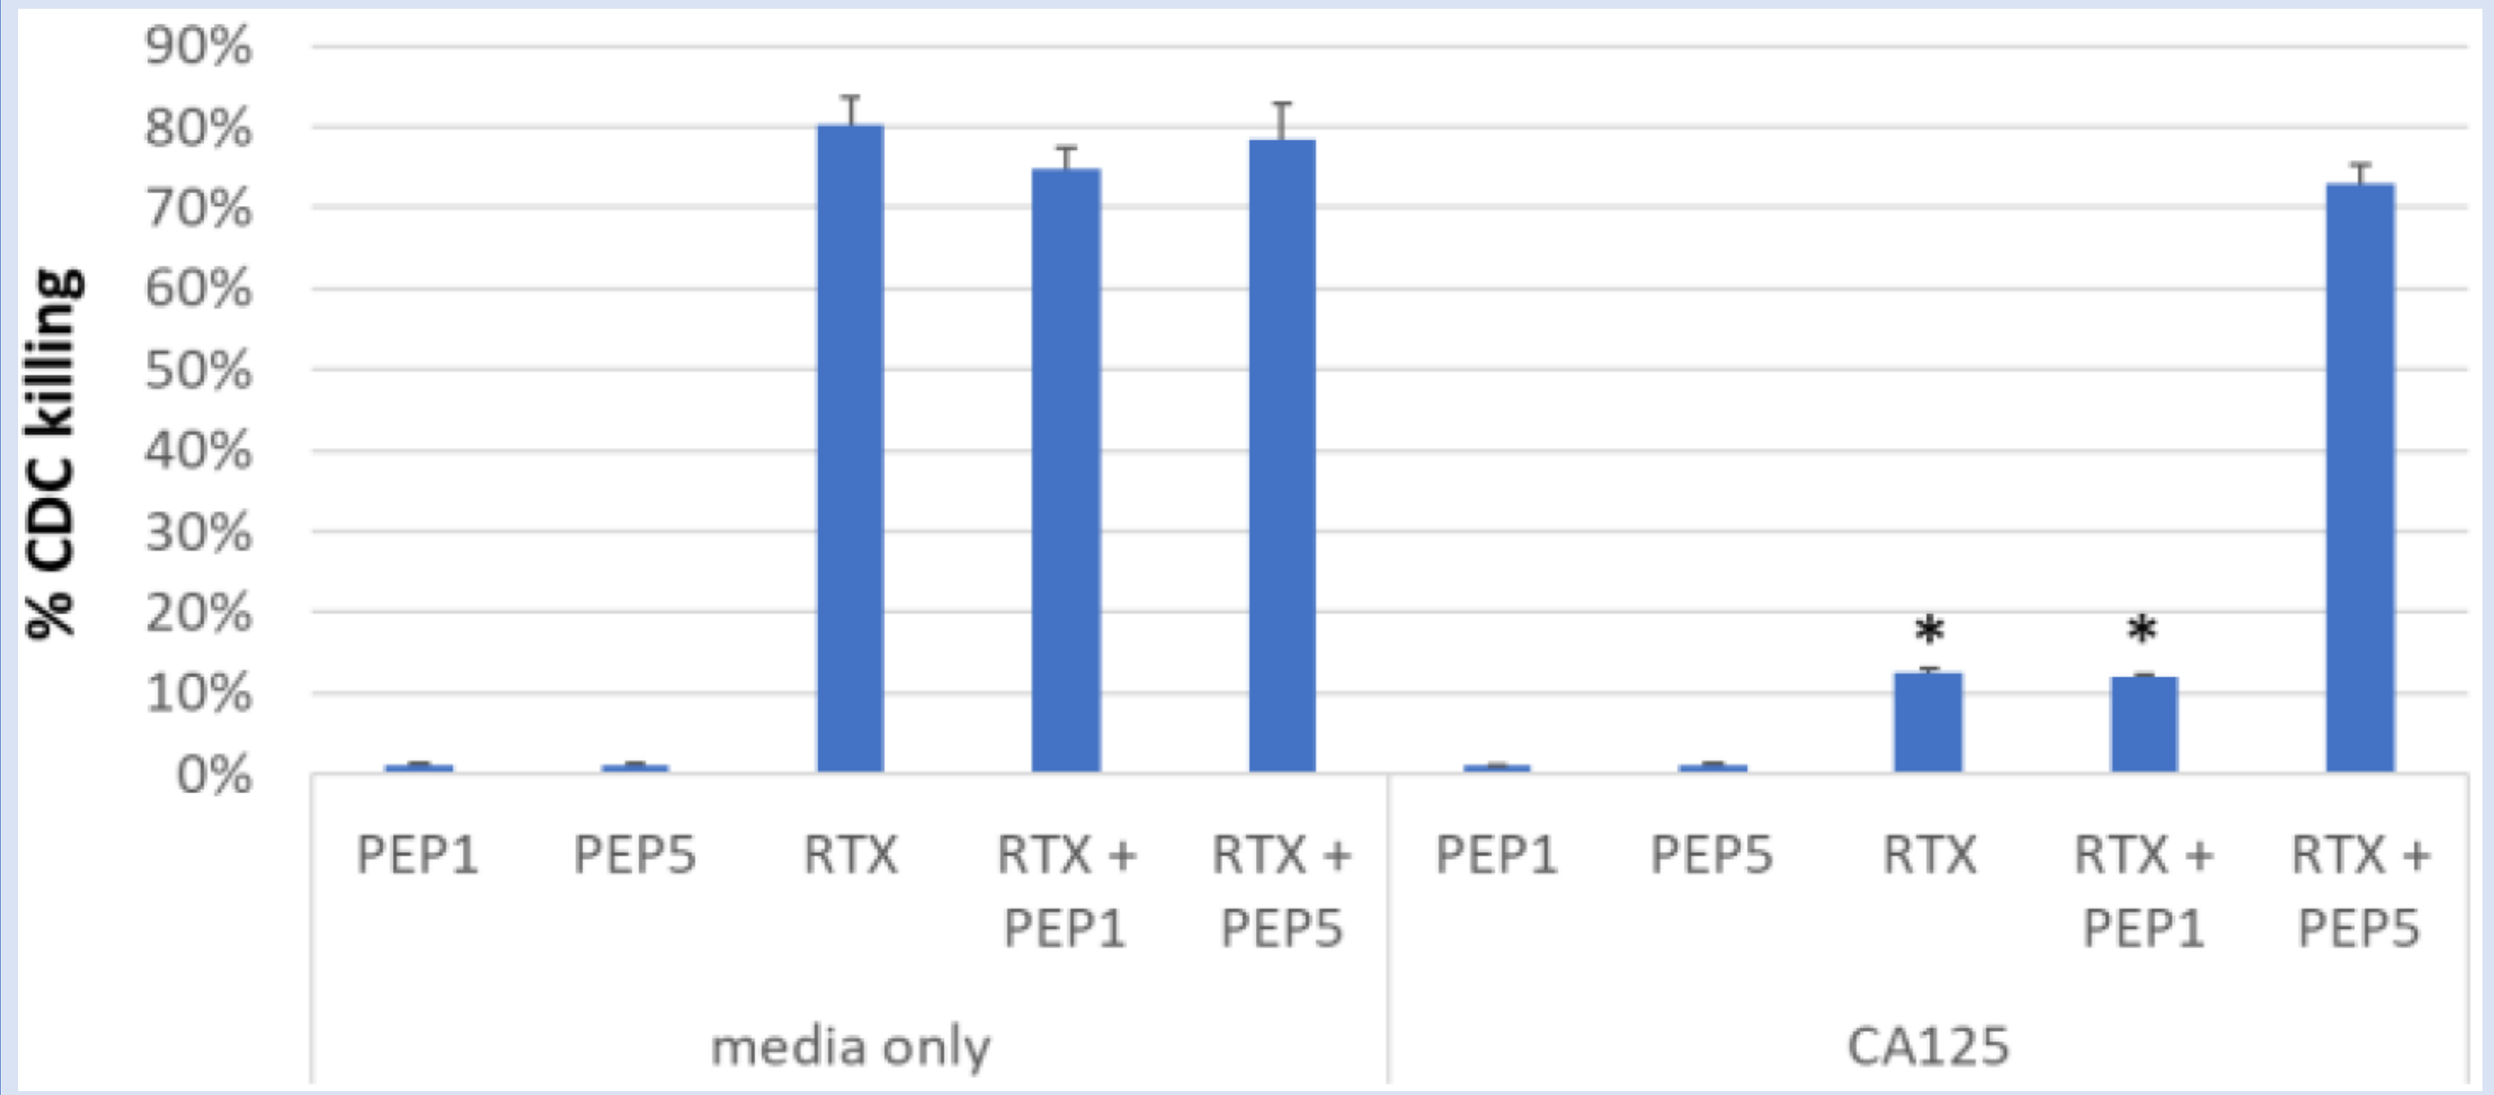

% CDC killing
PEP1
PEP5
RTX
RTX + PEP1
RTX + PEP5
media only
CA125

Fig.4. PEP5 can block CA125 CDC immune-effector suppression on rituximab (RTX). CHO-CD20 cells were plated in triplicate in 96 well plates in complement-containing growth media +/- 15 KU/mL CA125 along with PEP1, PEP5, RTX alone or in combination After 1.5 hours, cell viability was analyzed as a readout of CDC killing using Cell Titer-GLO (Promega). As shown, CA125 suppression of RTX CDC was blocked by addition of PEP5. P < 0.001 using T-test.

Figure 5. Generation and characterization of PEP5-Fc fusion proteins

Historically, the use of peptides in medicine have been challenging because of their short steady-state serum levels due to degradation and renal secretion. Fusion to the human IgG1 Fc domain has frequently been employed to generate larger, more stable polypeptides for medicinal use. We hypothesized that PEP5 could be formatted as an IgG1 Fc fusion protein with multiple repeating PEP5 domains and could be more effective at antagonizing CA125 IE suppression. We engineered PEP5-human IgG1 Fc fusions containing 1 - 3 PEP5 domains linked via a 5 amino acid spacer into a high-titer mammalian expression vector containing the zeocin selection marker. Constructs were stably transfected into human 293 cells, selected and screened for secreted PEP5-Fc fusion proteins using mouse anti-human-Fc-HRP as a probe via ELISA. Fc fusions containing 1, 2 and 3 PEP5 domains on the N-terminus were successfully generated (Fig. 5A), purified and tested for CA125 binding.

Results showed that Fc fusions containing two or more PEP5 domains (FC-2 and FC-3) bound CA125 stronger than the fusion containing a single PEP5 domain (FC-1). Further analysis found that constructs with two PEP5 domains were similar to those with three or more so the FC-2 construct was pursued. We mapped the binding of PEP5 to the C-terminal extracellular region of CA125 and used this fragment to measure the binding affinity of FC-1 and FC-2 to CA125 via surface plasmon resonance (SPR) by tethering the FC-1 or FC-2 to a biosensor chip and using the CA125 fragment as analyte. FC-2 was found to have a binding affinity of 15 pM while FC-1 has a binding affinity of 65 pM (Fig. 5B). Based on these data, we pursued development of FC-2 (renamed NAV-005).

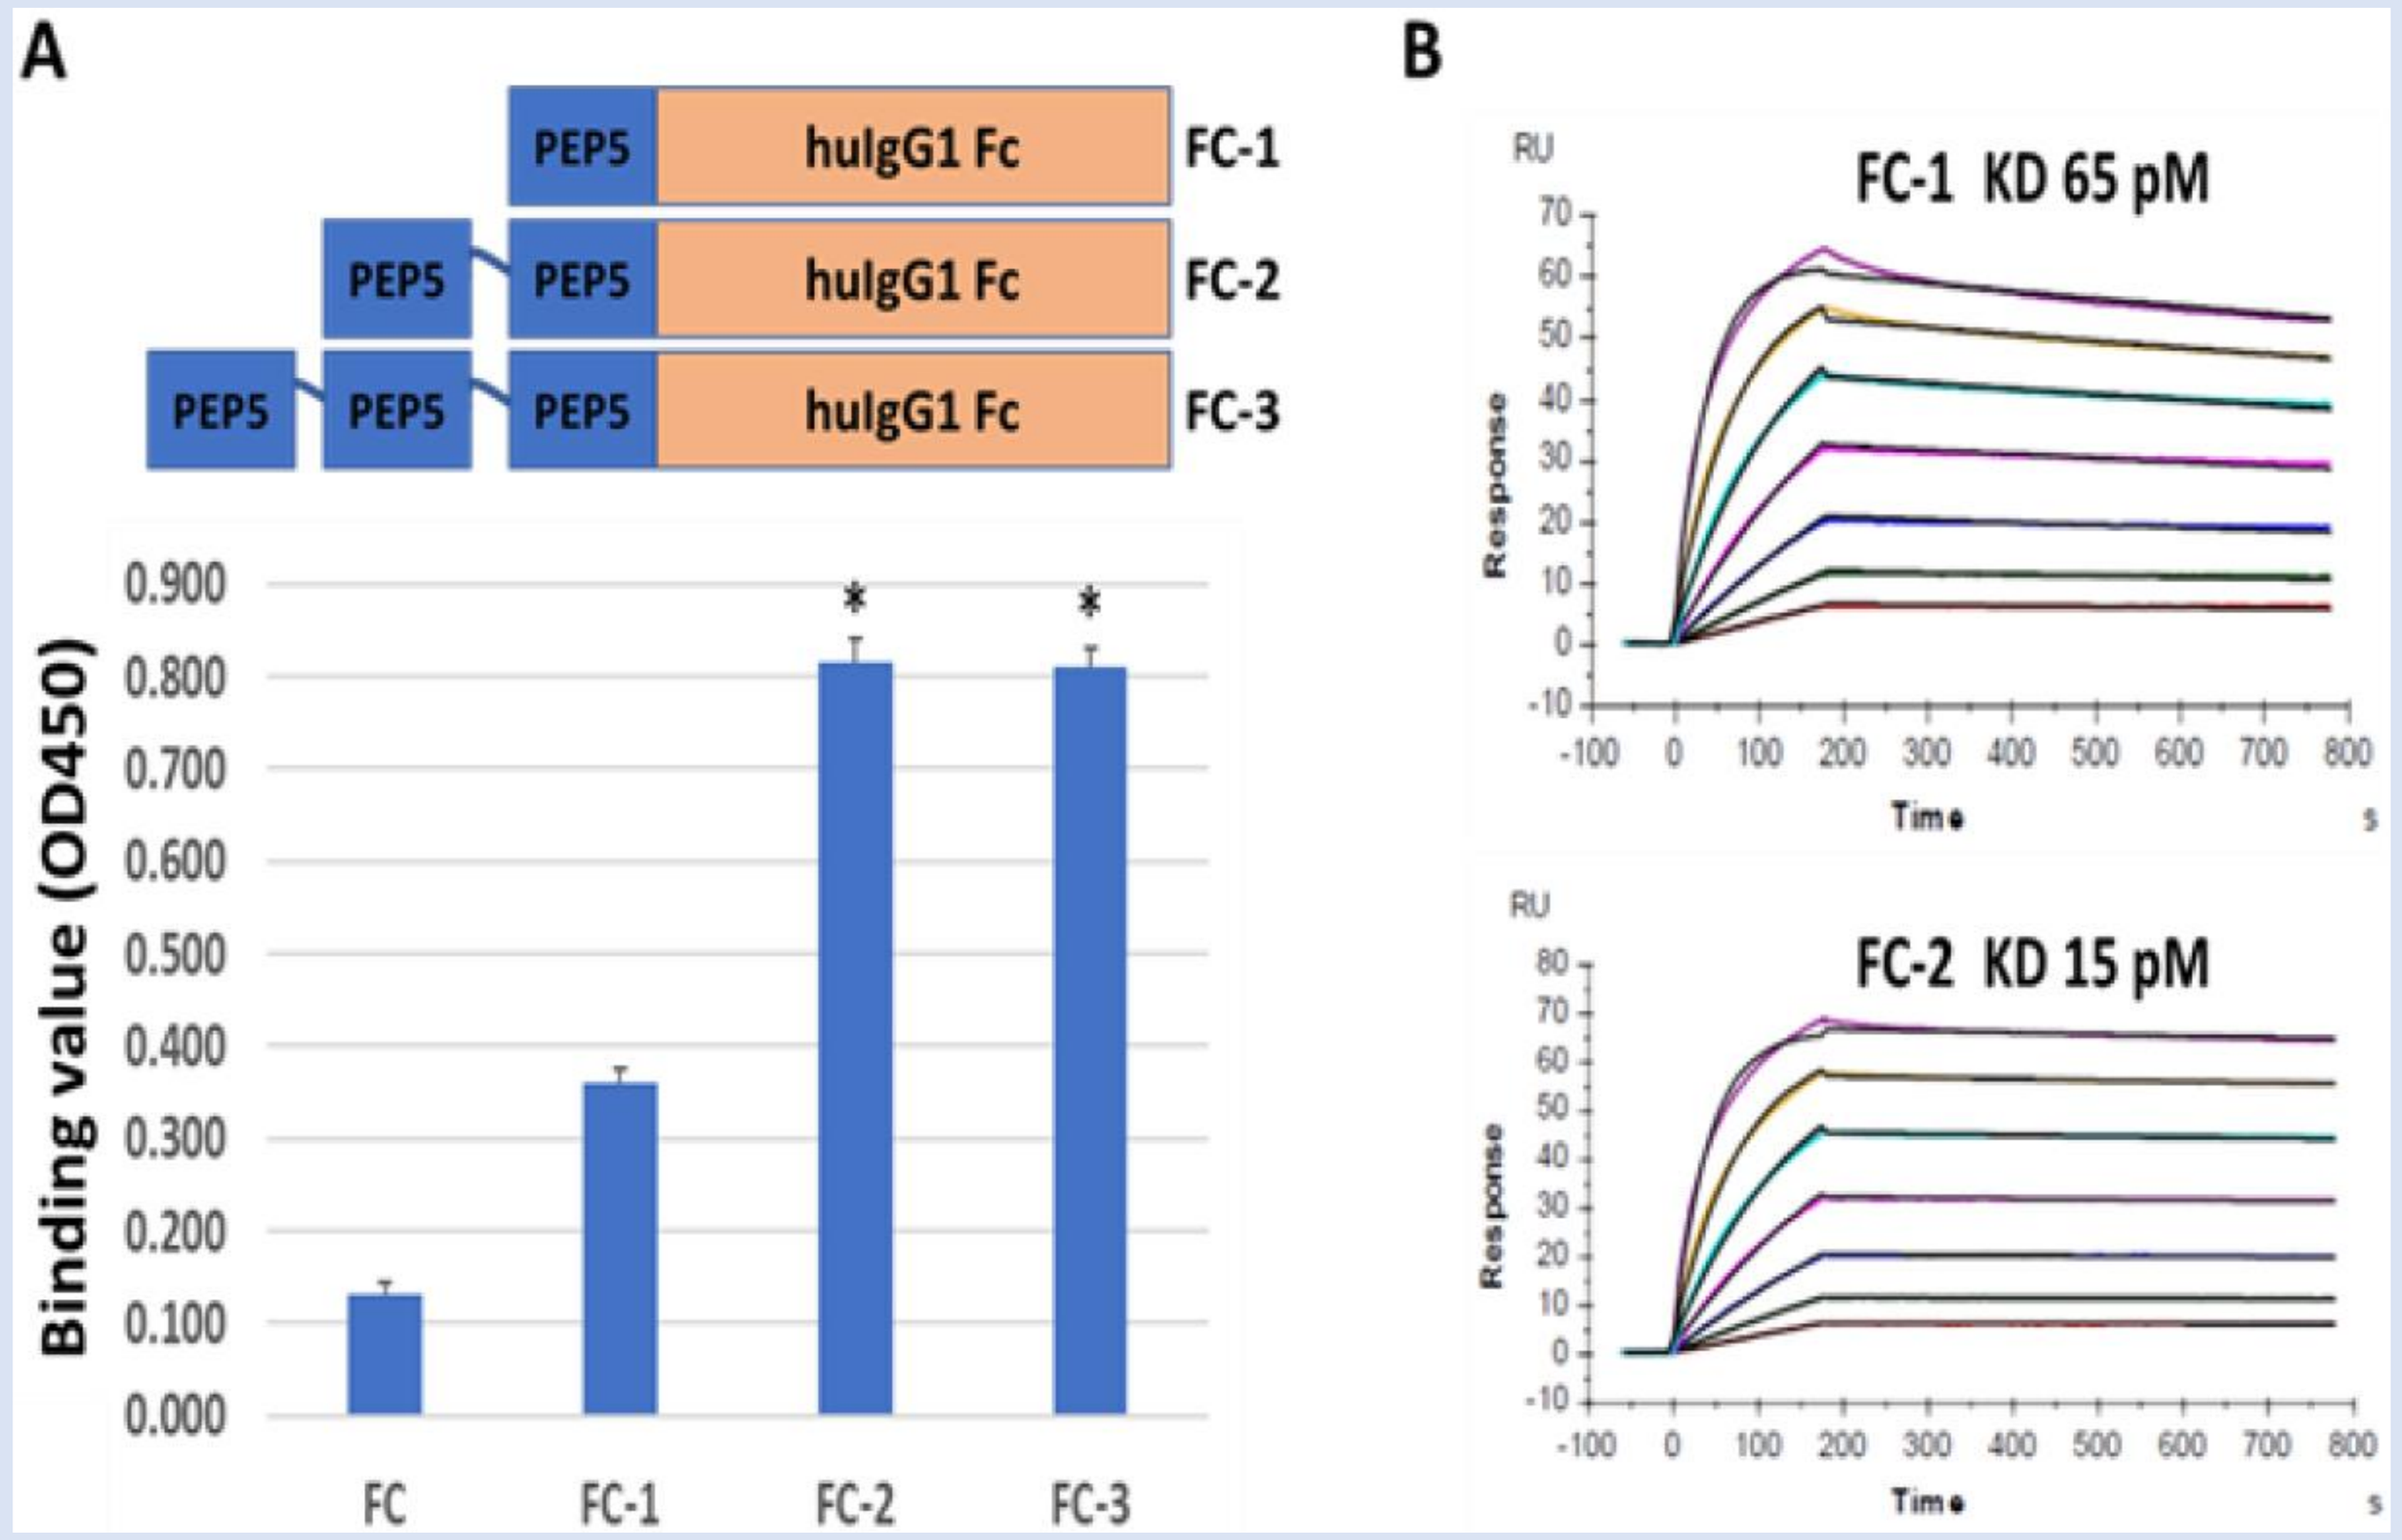

A
PEP5
huIgG1 Fc
FC-1
FC-2
FC-3
Binding value (OD450)
FC
FC-1
FC-2
FC-3
B
FC-1 KD 65 pM
FC-2 KD 15 pM
RU
Response
Time

Fig.5. PEP5-Fc fusion proteins retain high affinity CA125 binding. A, ELISA plates were coated with CA125 and probed with biotin-FC, FC-1, FC-2, or FC-3. Dimeric (FC-2) and trimeric (FC-3) fusions exhibited greatest binding. B, SPR quantitation of CA125-FC interactions. FC proteins were immobilized on biosensor chips and CA125 fragment was used as analyte.

Figure 6. NAV-005 fusion protein overcomes IE inhibition of CA125 on RTX target cell killing

We next examined if NAV-005's (FC-2) ability to bind CA125 could restore immuno-effector functions of RTX in the presence of CA125. As shown in Fig. 6A, NAV-005 could effectively restore CD16a, CD32a and C1q binding to RTX in the presence of CA125. These effects were also observed in cellular assays measuring the effect of CA125 on ADCC using human primary peripheral blood mononuclear cells (PBMCs) as well as CDC using human complement and CHO-CD20 target cells (Fig. 6B). Based on these findings and the potential need for novel agents to help combat Ab-based therapies in CA125-expressing cancers, including RTX in FL, further development of NAV-005 is being pursued for *in vivo* validation and optimal dosing.

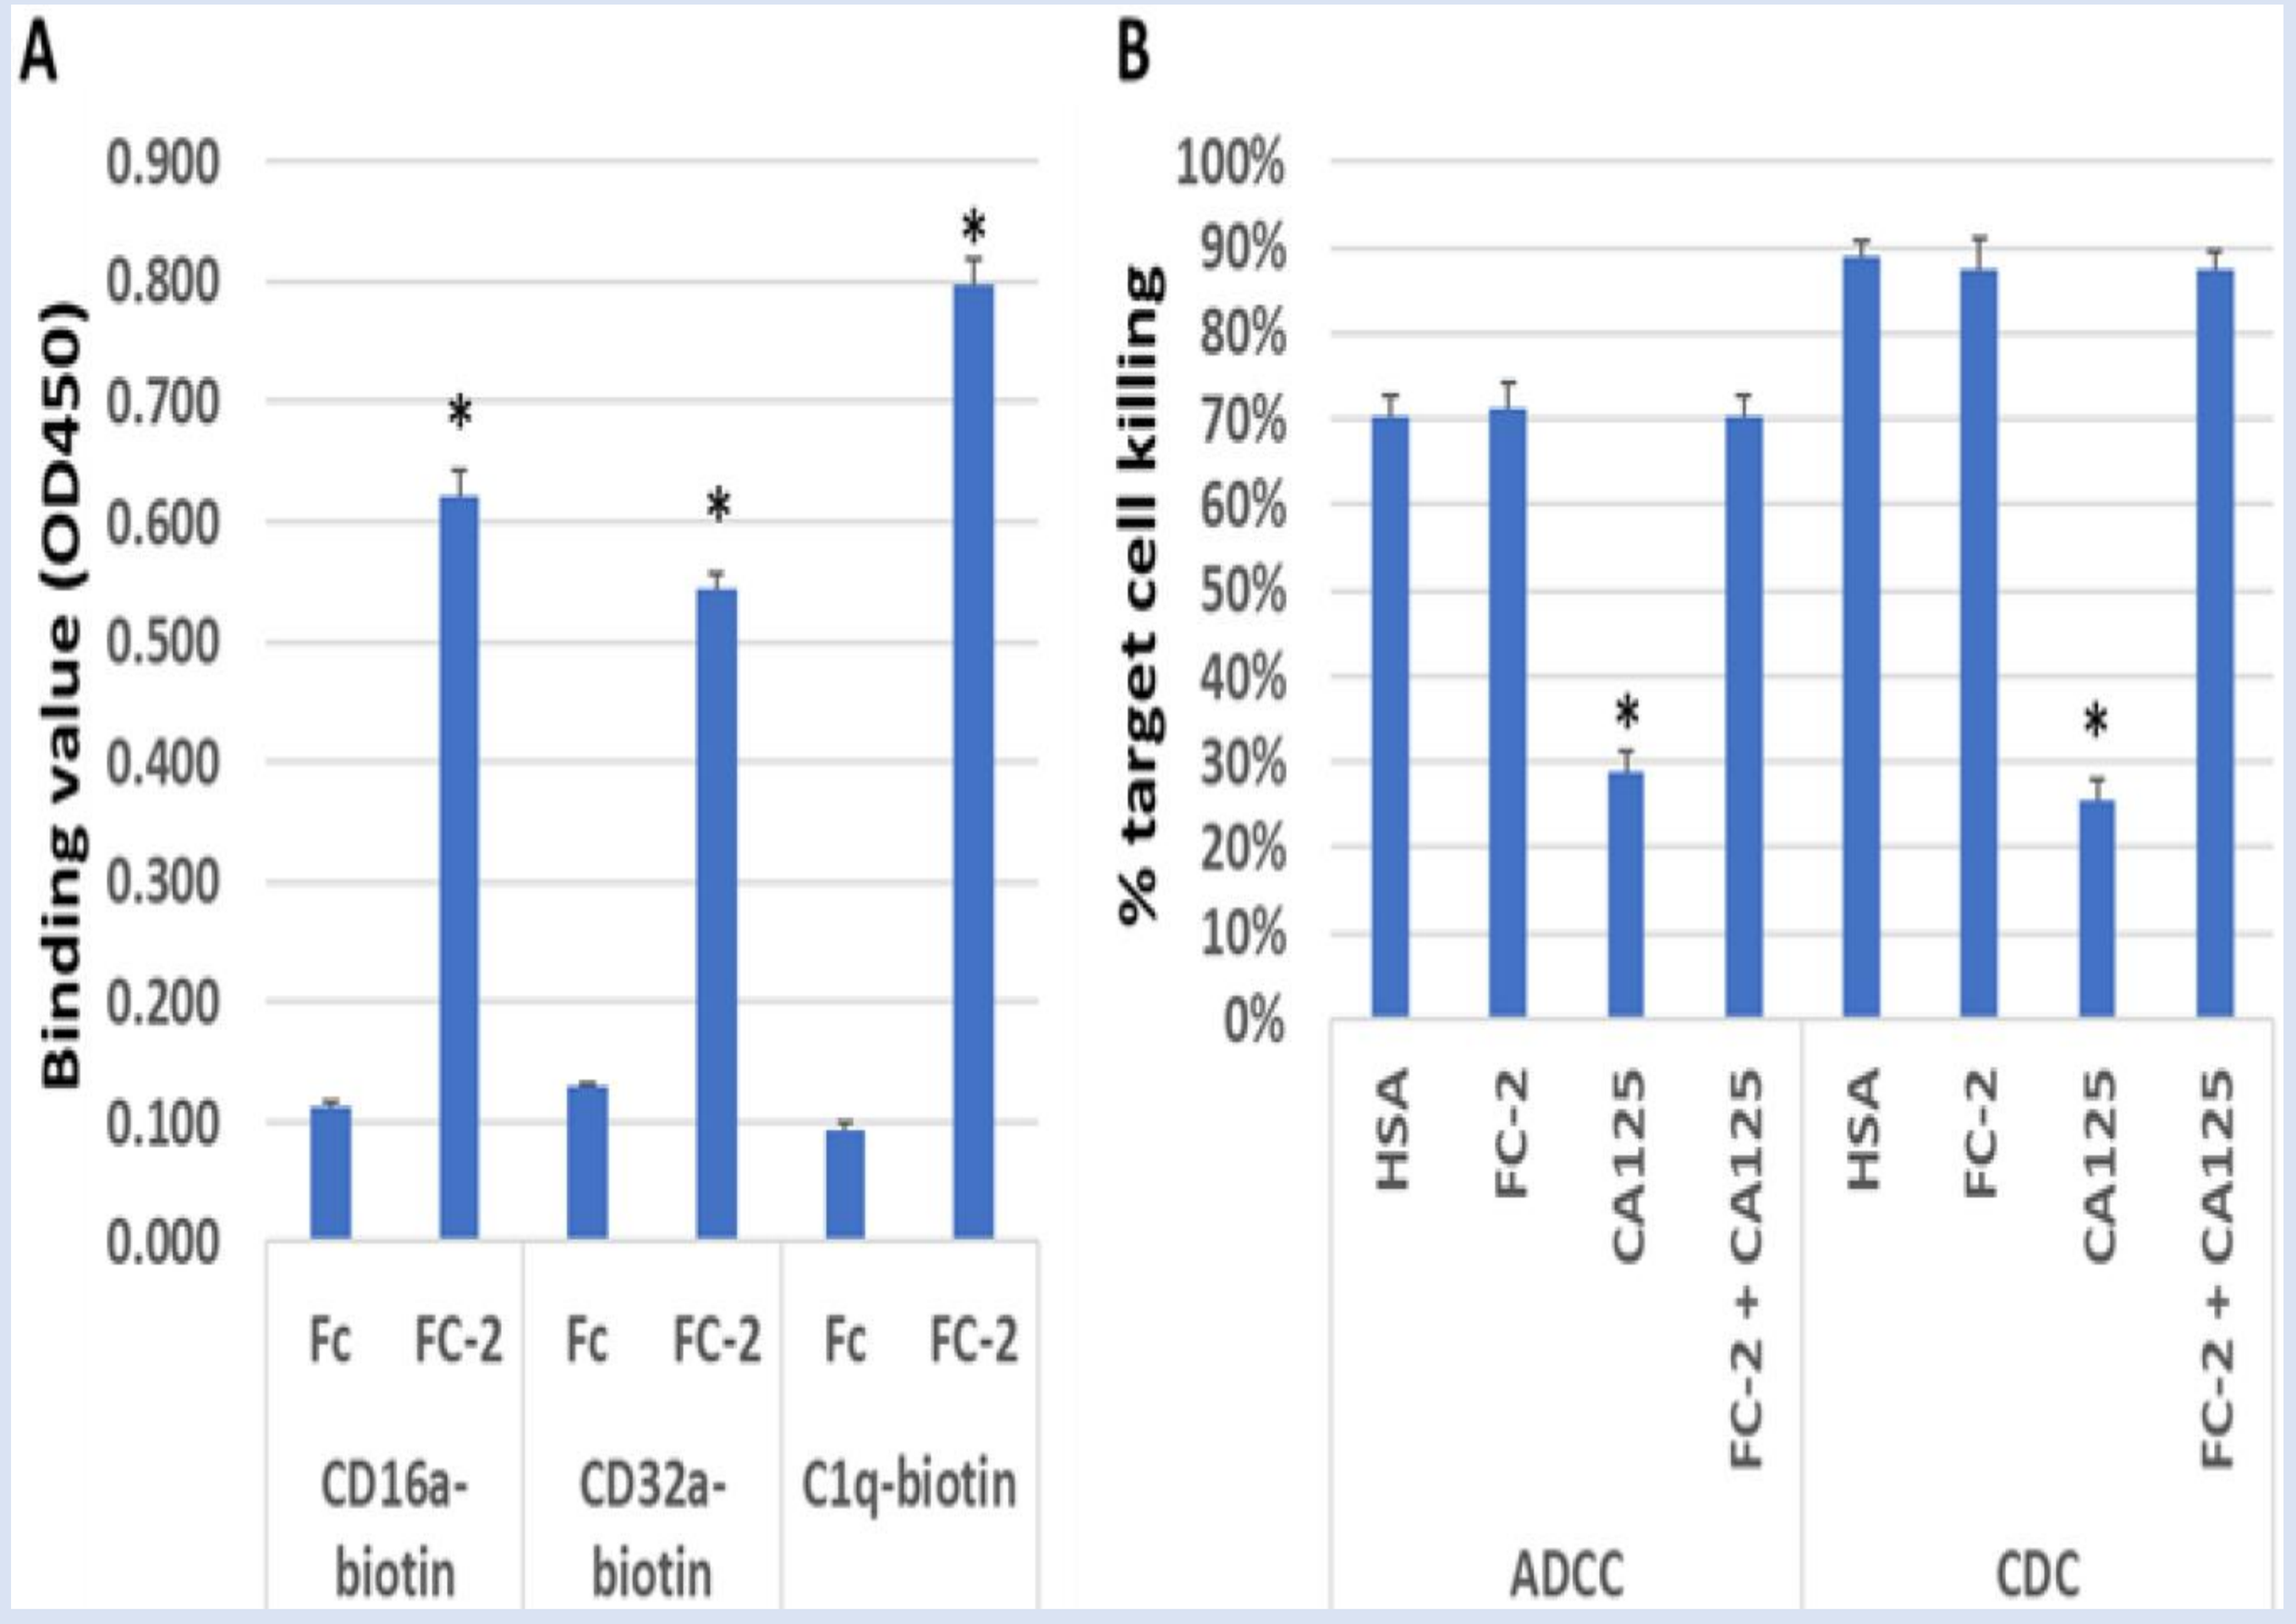

A
Binding value (OD450)
Fc
FC-2
CD16a-biotin
CD32a-biotin
C1q-biotin
B
% target cell killing
HSA
FC-2
CA125
FC-2 + CA125
ADCC
CDC

Fig.6. NAV-005 overcomes CA125 suppressive effects on RTX IE activity. A, ELISA plates were coated with 15 KU/mL CA125 and 5 μg/mL RTX and probed with biotinylted-CD16a, -CD32a, or -C1q proteins in the presence of FC or FC-2 (NAV-005). As shown, NAV-005 was able to restore binding of both Fc receptors and C1q to RTX. B, CHO-CD20 target cells were treated with RTX and CA125, NAV-005 (FC-2) or control proteins in the presence of PBMCs (ADCC) or complement (CDC). As shown, NAV-005 could rescue the immune-effector suppression elicited by CA125 on RTX. Values are from at least triplicate. P < 0.001 using T-test.

Figure 7. NAV-005 blocks the interaction of CA125 with Siglec-9

While NAV-005 has utility in rescuing the CDC and ADCC activities of antibodies that are directly bound by the immunosuppressive CA125 protein, another mechanism of immune suppression is the direct impact of CA125 on suppressing NK cell activation via binding to negative regulatory receptors (4,5). We have been studying the impact of NAV-005 on rescuing ADCC activity of antibodies using the Jurkat-CD16a reporter activation assay (Promega Corp), human peripheral blood mononuclear cells (PBMCs) and primary NK cells. As shown in Fig. 7, CA125 binds to the SIGLEC-9 receptor, which is expressed on NK cells and shown to suppress NK cell killing via ADCC (as reported by others, ref 4), but does not bind other family members such as SIGLEC-1. Moreover, NAV-005 (FC-2) is able to block the binding of CA125 to SIGLEC-9 and potentially overcome the inhibitory effects on NK/effector cell activation and ADCC activity.

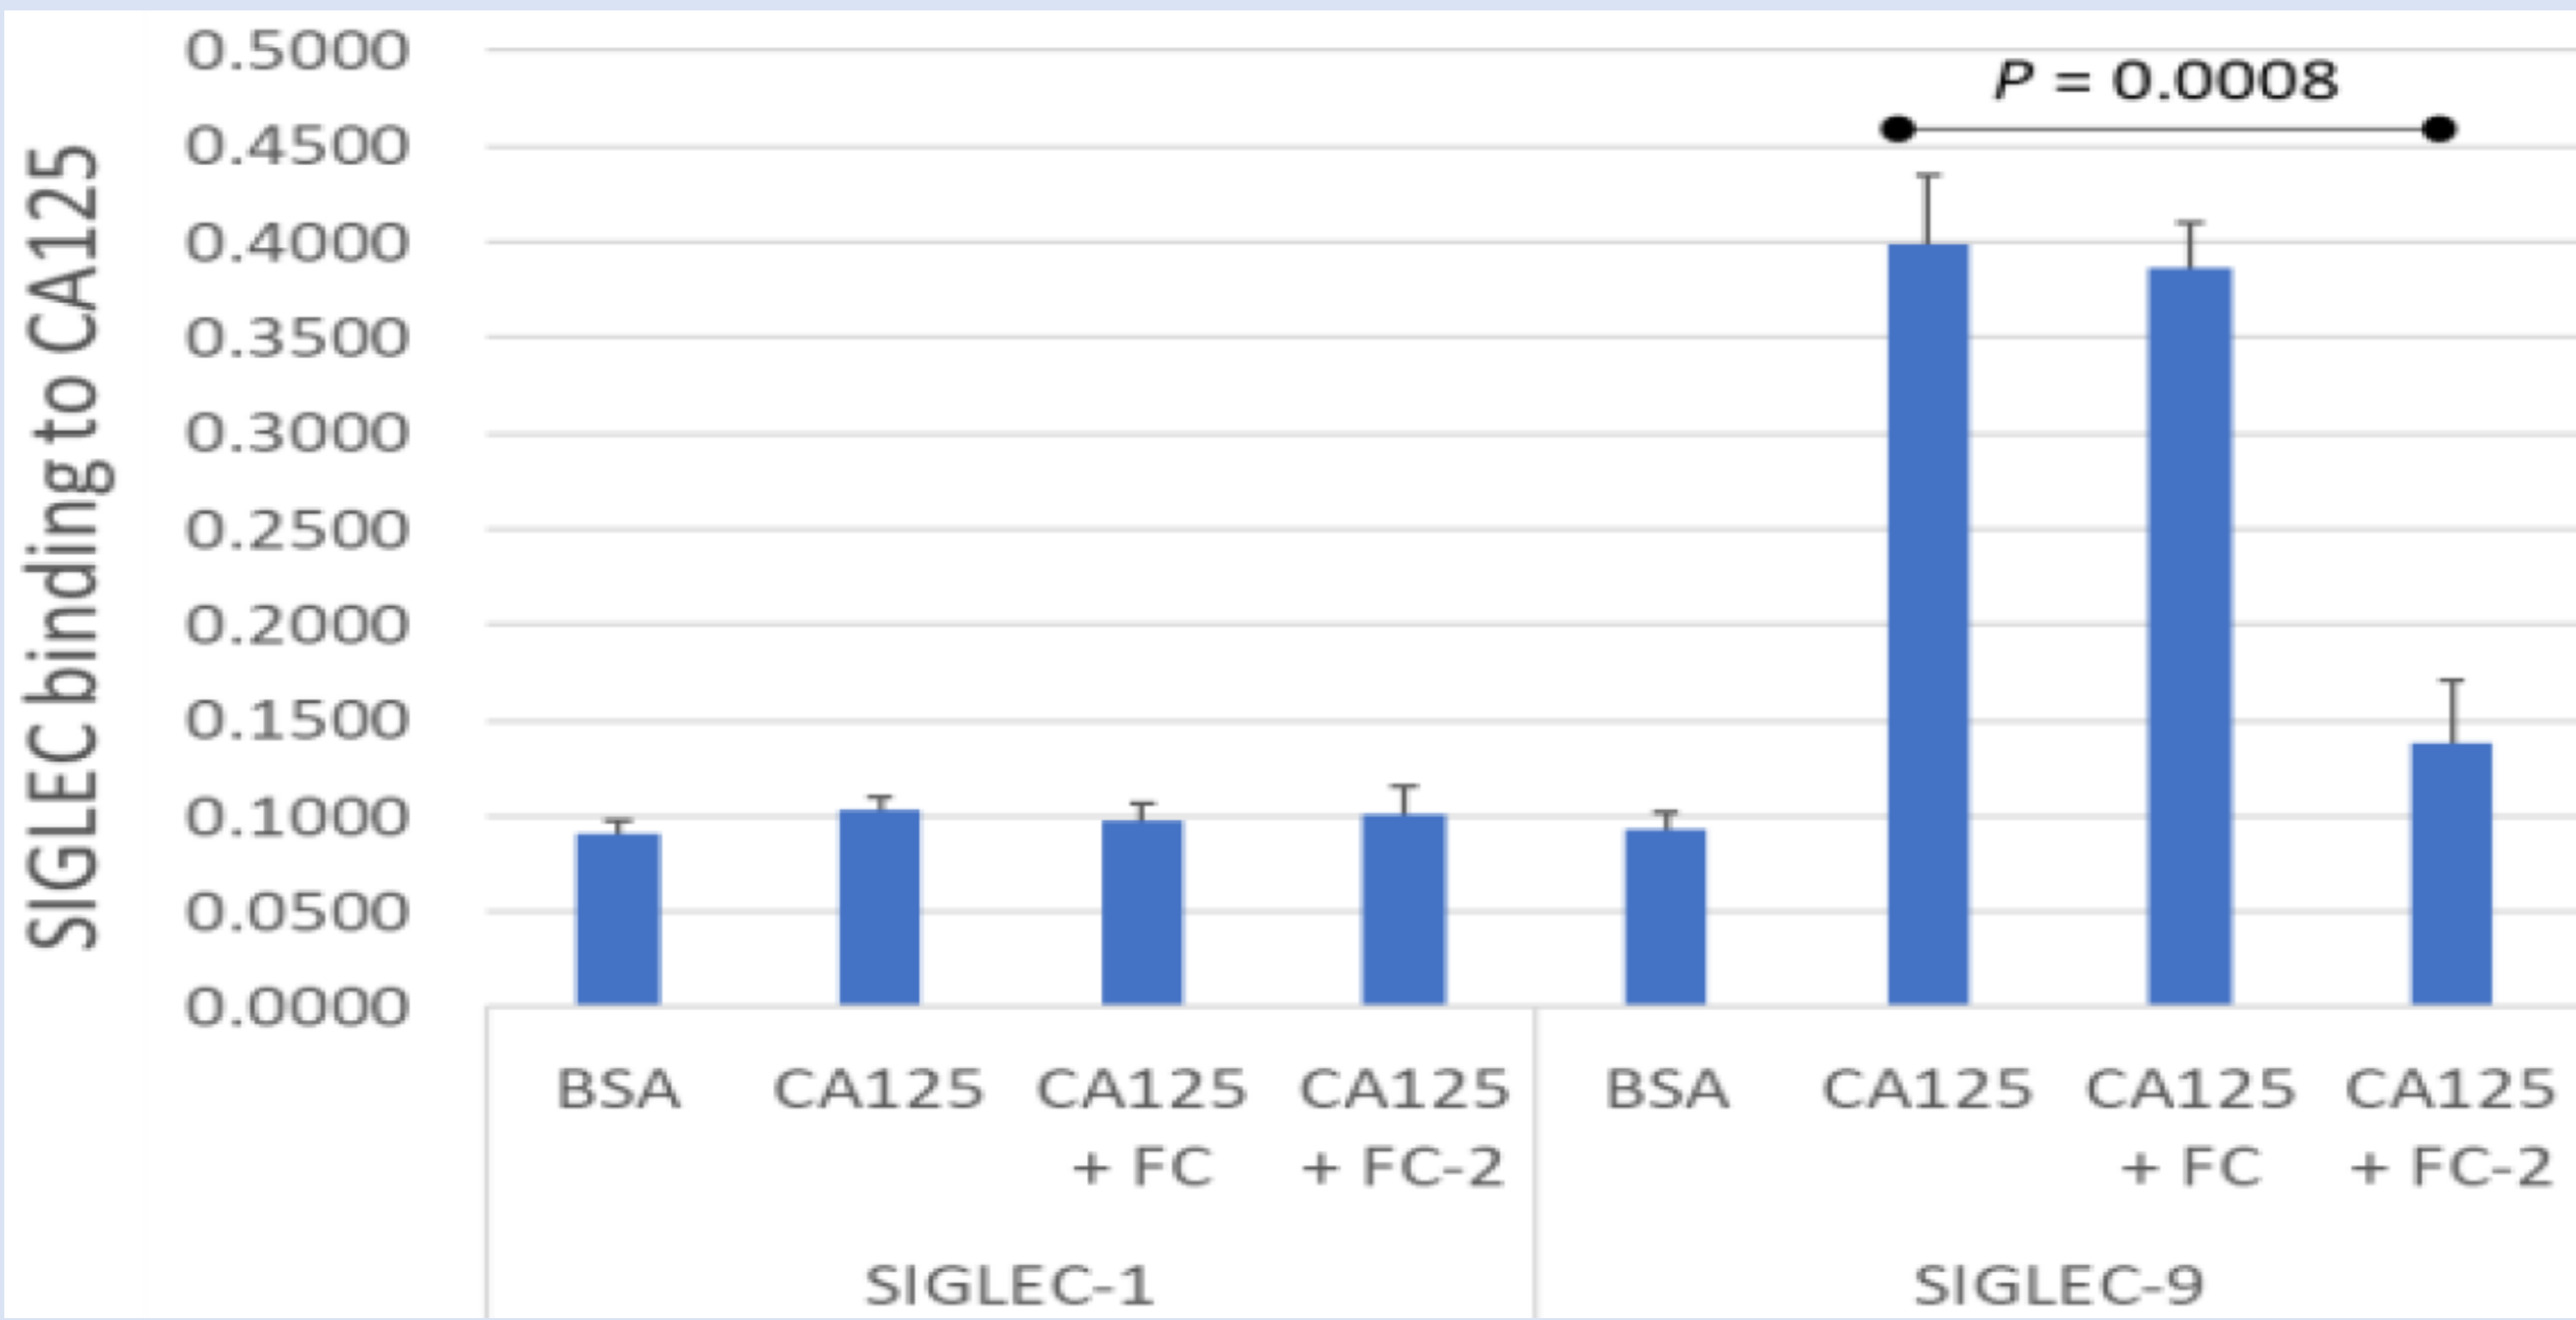

SIGLEC binding to CA125
P = 0.0008
BSA
CA125
CA125 + FC
CA125 + FC-2
SIGLEC-1
SIGLEC-9

Fig. 7 Siglec binding to CA125. ELISA plates coated with BSA or 15 KU/mL of CA125 were probed with 1-5 μg/mL of Siglec-1-His or Siglec-9-His +/- 5 μg/mL NAV-005 (FC-2) or Fc fragment. Binding was detected using anti-His-HRP antibody. As shown, CA125 directly binds CA125 and is blocked by NAV-005 (FC-2). All experiments are done in triplicate.

Figure 8. NAV-005 enhances humoral immune activities of RTX via blockade of CA125 binding as well as immune-effector cell activation

The Jurkat-CD16a reporter assay monitors antibody-CD16a Fc receptor activation using a luciferase readout. If antibody binds target cells and engages with Jurkat-CD16a, luciferase is activated and is quantified by luminescent signal intensity. Importantly, Jurkat-CD16a cells do not express SIGLEC-9 inhibitory receptor. Primary PBMCs and NK cells both express SIGLEC-9 (as well as other regulatory receptors) and are CD16a-postive effector cells. These features enable the screening and monitoring of the effect(s) that CA125 has on antibodies that it directly binds (such as RTX) as well as on IE activity of antibodies that it does not directly bind (such as anti-HER2 pertuzumab, PTZ). We have generated a target cell line (OV-CD20) that naturally produces high levels of CA125 and also expresses the CD20 and HER2 antigens, which allows for comparative analysis of RTX and PTZ on effector cell systems. In both systems, RTX and PTZ had activity against the OV-CD20 target cells. However, when used in combination with NAV-005 (FC-2) in Jurkat-CD16a assays, significant enhancement of activity was observed with RTX, as CA125 directly binds to RTX but no enhanced effect was observed for PTZ, which is not bound by CA125 (Fig. 8). However, both RTX and PTZ ADCC activity was enhanced when either was used in combination with NAV-005 (FC-2) in PBMC assays. This effect was reversed if >10 fold molar amounts of soluble SIGLEC-9 were added to the assay, suggesting the effect is SIGLEC-9 mediated. These data support a process where PBMC ADCC activation is perturbed in part by CA125/SIGLEC-9 interaction.

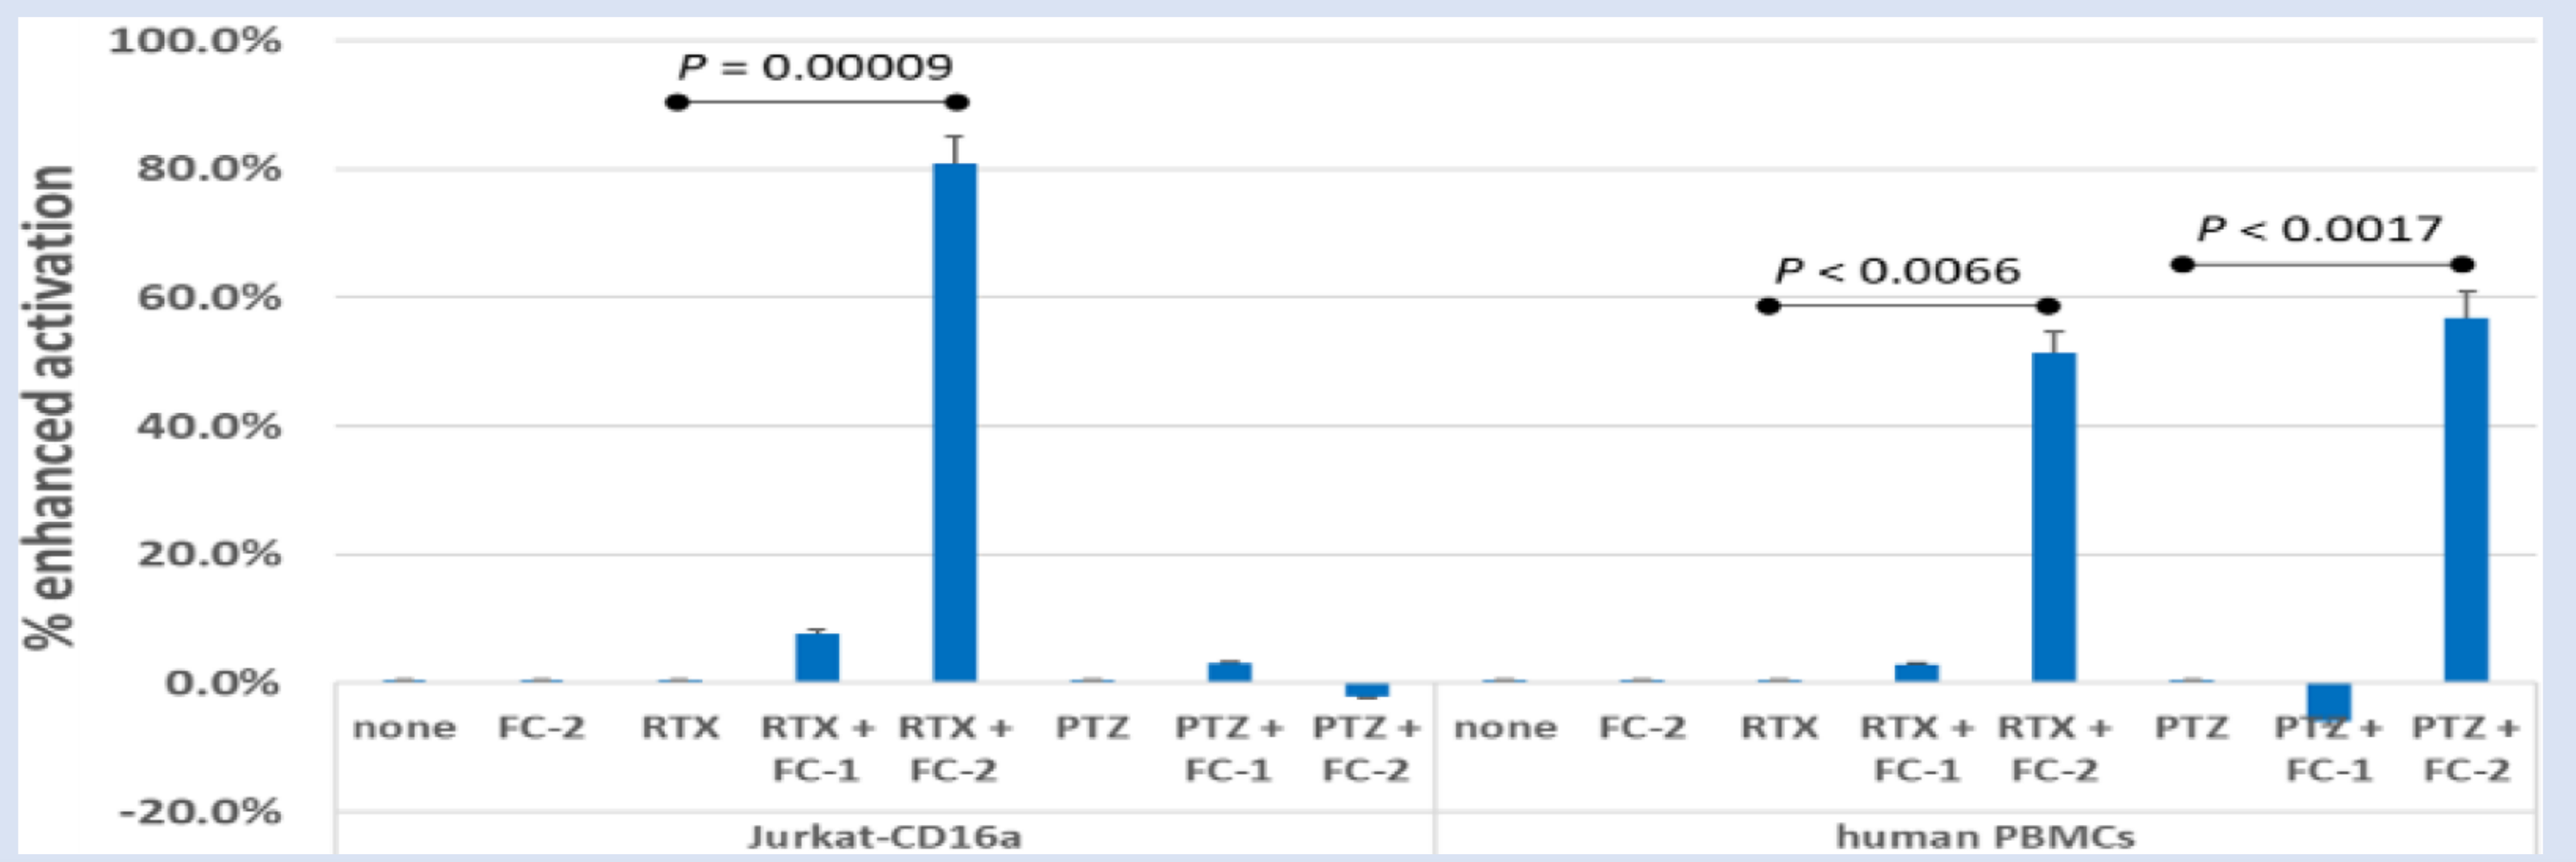

% enhanced activation
P = 0.00009
P < 0.0066
P < 0.0017
none
FC-2
RTX
RTX + FC-1
RTX + FC-2
PTZ
PTZ + FC-1
PTZ + FC-2
Jurkat-CD16a
human PBMCs

Fig.8. NAV-005 (FC-2) enhances CD16a activation of RTX and ADCC activity of RTX and PTZ on CA125-producing target cells. Left side shows enhanced activation of Jurkat-CD16a reporter cells in the presence of FC-2 using RTX. Note PTZ does not bind CA125 and therefore no enhanced activation is observed +/- FC-2. Right side shows enhanced ADCC killing of OV-CD20 target cells by PBMC effectors using FC-2 in the presence of RTX or PTZ as compared to OV-CD20 cultures treated with Ab alone.

REFERENCES

1. Kline JB, Kennedy RP, Albone E, Chao Q, Fernando S, McDonough JM, Rybinski K, Wang W, Somers EB, Schweizer C, Grasso L, Nicolaides NC. Tumor antigen CA125 suppresses antibody-dependent cellular cytotoxicity (ADCC) via direct antibody binding and suppressed Fc-γ receptor engagement. Oncotarget 8:52045-52060, 2017.
2. Kline JB, Fernando S, Ross EN, Grasso L, Nicolaides NC. Tumor shed antigen CA125 blocks complement-mediated killing via suppression of C1q-antibody binding. Eur J Immunol 48:1872-1882, 2018.
3. Vergote I, Armstrong D, Scambia G, Teneriello M, Sehouli J, Schweizer C, Weil SC, Bamias A, Fujiwara K, Ochiai K, Poole C, Gorbunova V, Wang W, O'Shannessy D, Herzog TJ. A randomized, double-blind, placebo-controlled, Phase III study to assess efficacy and safety of weekly farletuzumab in combination with carboplatin and taxane in patients with ovarian cancer in first platinum-sensitive relapse. J Clin Oncol 34:2271-8227, 2016.
4. Belisle JA, Horibata S, Gubbels, JA, et al. Identification of Siglec-9 as the receptor for MUC16 on human NK cells, B cells, and monocytes. Mol Cancer 9:118-132, 2010.
5. Jandus C, Boligan KF, Chijioke O, Liu H, et al. Interactions between Siglec-7/9 receptors and ligands influence NK cell–dependent tumor immunosurveillance. J Clin Invest 124:1810–1820, 2014.